Impact of PEAK Methodology on Interdialytic Weight Gain: A Multi-Center Experience

Atef Z. Altaher, Marta C. Bento, Janet Cowperthwaite

DaVita Saudi Arabia, Riyadh, Saudi Arabia

Introduction

- Hemodialysis (HD) patients who experience larger interdialytic weight gain (IDWG) are at higher risk of cardiovascular and all-cause mortality.[1,2]
- Diastolic dysfunction associated with fluid overload occurs frequently among dialysis patients, and may directly reduce survival.[3]
- In particular, IDWG of ≥ 4% of dry weight may be associated with a substantially higher risk of death.

Objective

To reduce the percent of patients experiencing IDWG ≥4% through the application of "Pursuing Excellence by Advancing Knowledge" (PEAK) methodology.

Methods

Clinics and Patients

- This study considered 15 DaVita Saudi Arabia clinics that were selected to participate in the quality improvement program targeting IDWG.
- All prevalent and incident patients treated with HD at those clinics and who had valid data reported in the electronic health records system were included (n=1472).

PEAK Methodology

- PEAK methodology is based upon six sigma and lean processes.
- The methodology was introduced incrementally at each participating clinic.
- During the implementation period, teams were provided with education about PEAK and supported to hold daily reviews of current status, followed by discussion of ideas and action plans to implement patient-centered strategies to promote compliance.

Analysis

- The percent of patients with IDWG ≥ 4% on each treatment day during the 9-month study period was determined from electronic health records.
- Average values were calculated for each treatment week and month.
- Results were compared at 3, 6, and 9 months using the Chi^2 test.

Results

Summary of PEAK Methodology

- The PEAK method aims to instill continuous improvement principles at the clinic level.
- PEAK is a frontline problem-solving approach that focuses on patient care and teammate experience.
- A key component of the PEAK method is the daily team huddle, focusing on the key performance indicator (in this case, IDWG; Figure 1).

Figure 1:The PEAK Method

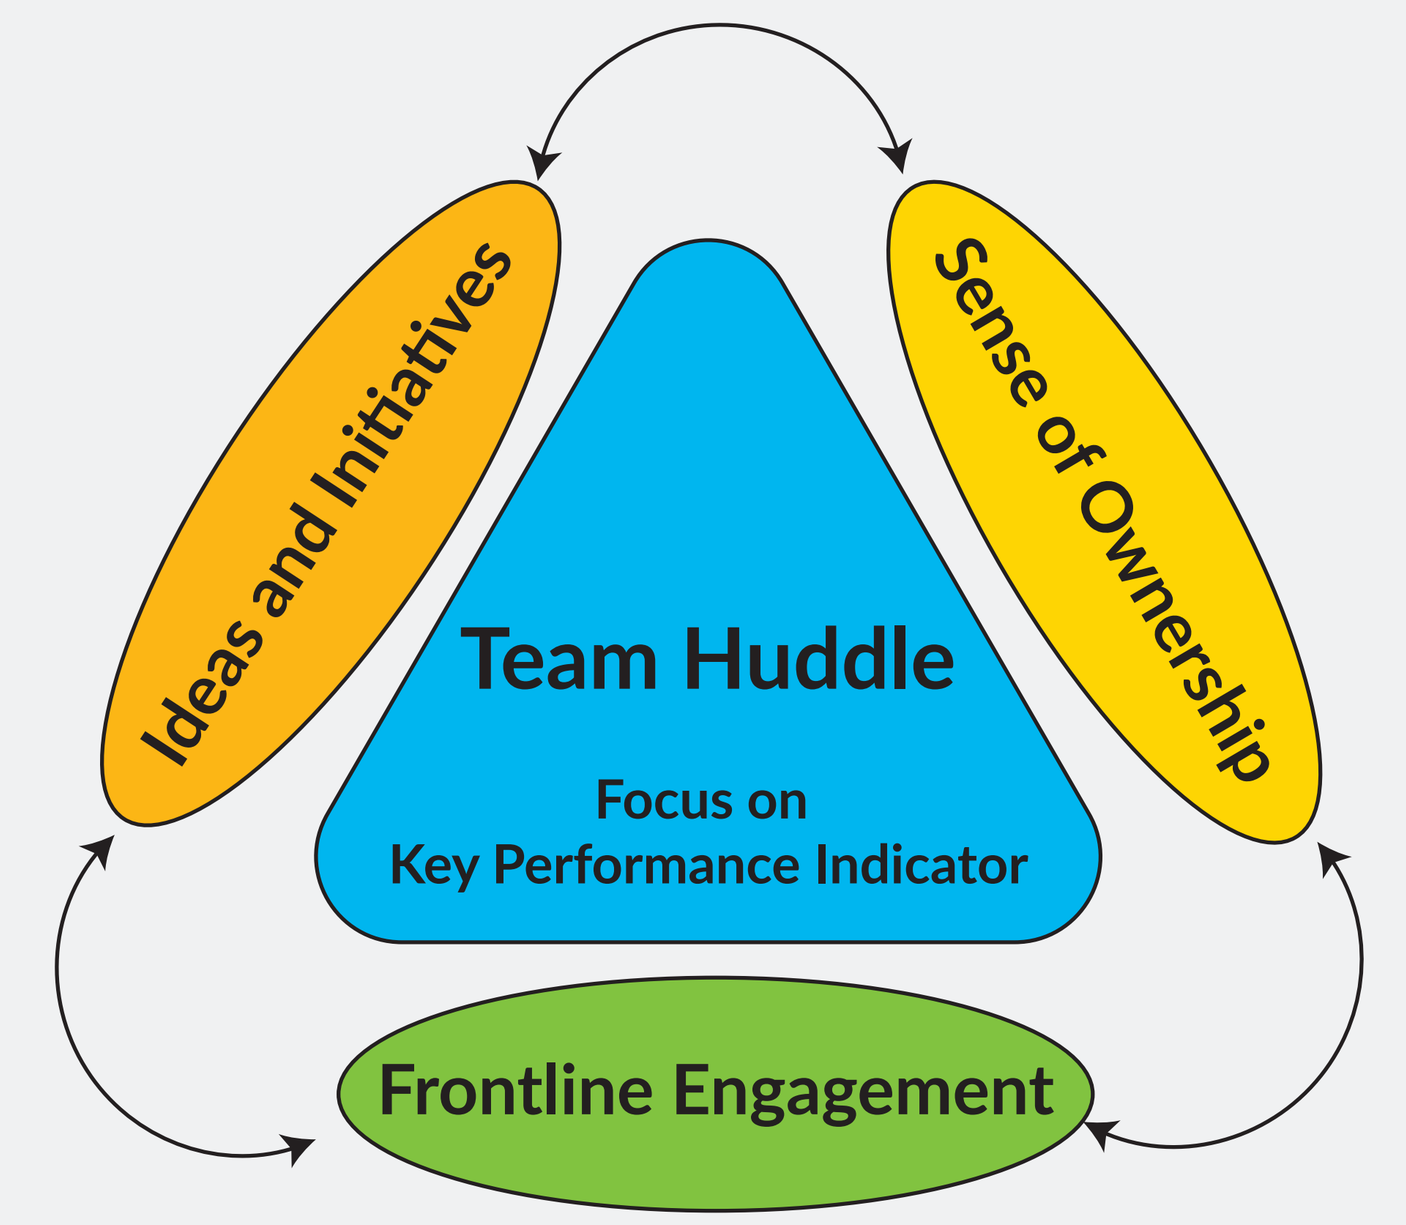
Ideas and Initiatives
Sense of Ownership
Team Huddle
Focus on
Key Performance Indicator
Frontline Engagement

Table 1: Clinic-Level Census and Percent of Patients with IDWG ≥4% at Baseline and Following Introduction of PEAK Program

	Baseline Period		3 Months		6 Months		9 Months	
Clinic ID	Clinic Census	IDWG ≥ 4% (%)	Clinic Census	IDWG ≥ 4% (%)	Clinic Census	IDWG ≥ 4% (%)	Clinic Census	IDWG ≥ 4% (%)
1	87	36.6	87	44.5	75	44.9	70	35.0
2	74	44.2	68	38.9	72	35.7	68	27.1
3	61	30.5	72	41.3	75	36.0	78	28.4
4	52	49.1	70	38	69	32.0	80	38.1
5	40	59.8	45	66.8	48	70.1	36	63.7
6	104	29	101	27.5	104	25.0	89	17.9
7	104	66.1	105	26.7	111	32.6	123	29.8
8	126	20.5	114	21.6	106	14.1	100	15.6
9	70	38.1	69	24.6	55	25.3	46	27.1
10	60	36.9	62	26.7	63	26.5	57	24.8
11	96	36.2	96	32.3	91	26.0	90	23.6
12	168	30.3	186	40.9	186	39.5	192	31.3
13	120	38.8	127	37.2	137	41.3	135	32.0
14	85	44.9	89	32.2	93	29.0	93	33.0
15	181	34.8	180	32.0	187	33.7	189	26.8

Impact of PEAK Program on IDWG

- The number of patients treated at each clinic during the study period is summarized in Table 1.
 - At baseline, the percentage of patients with IDWG ≥ 4% ranged from 20.5% to 66.1% across clinics.
 - At 9 months after initiation of the PEAK program, the percentage of patients with IDWG ≥ 4% ranged from 15.6% to 63.7%.
 - The proportion of patients with IDWG ≥ 4% declined from baseline to 9 months after PEAK initiation in participating clinics.
- The mean percentage of patients with IDWG ≥ 4% of dry weight over the study period is shown in Figure 2.
- The proportion of patients with IDWG ≥ 4% was significantly lower at 9 months after initiation of the PEAK program compared to the baseline period (Table 2).

Figure 2: Percent of Patients with IDWG ≥4% Over 9-Month Observation Period

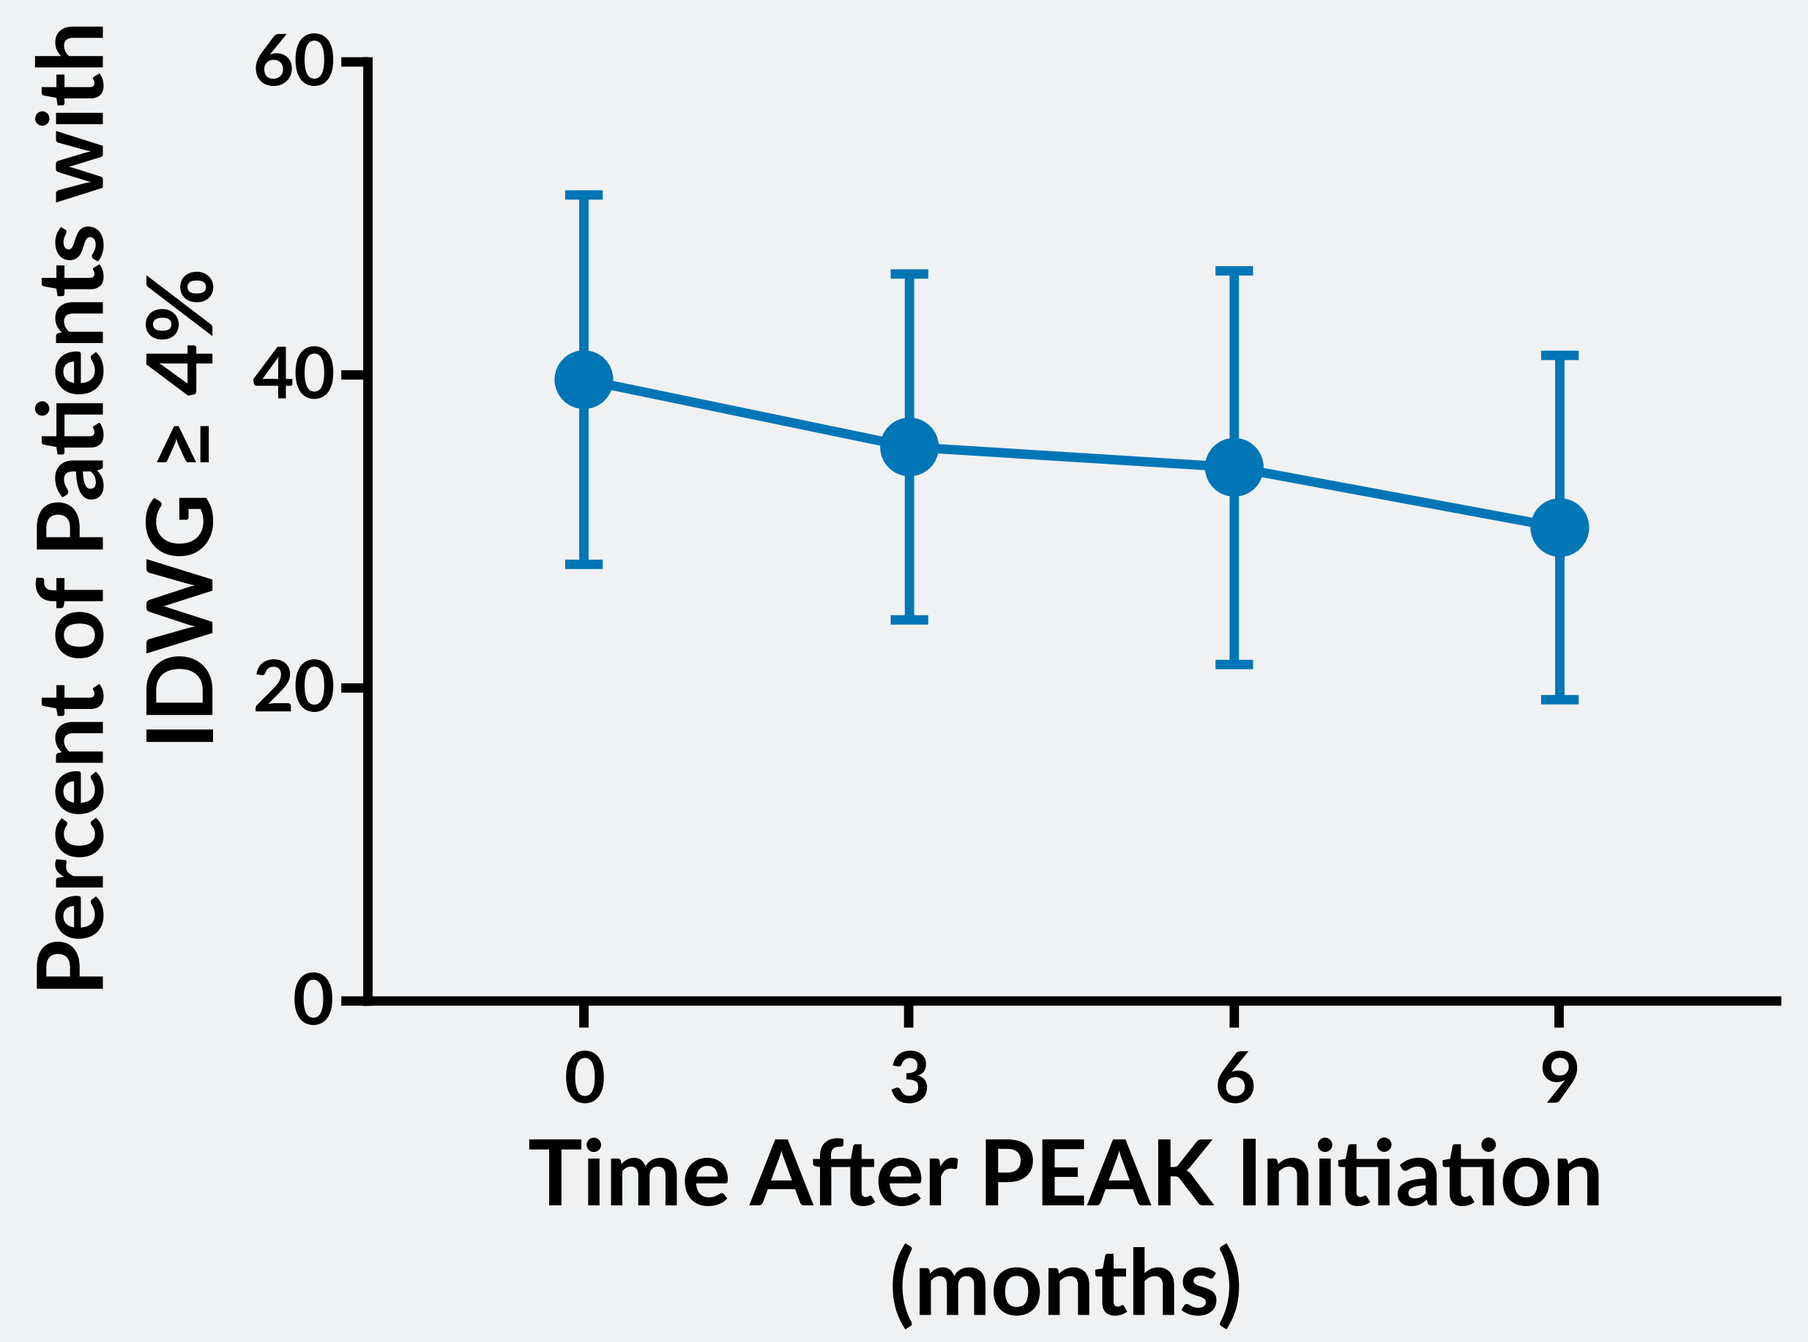
Percent of Patients with IDWG ≥ 4%
60
40
20
0
0
3
6
9
Time After PEAK Initiation (months)

The mean (standard deviation) percent of patients with IDWG ≥4% across the 15 participating clinics in each study period is shown. Month 0 represents the baseline, prior to initiation of the PEAK program.

Table 2: Percent of Patients with IDWG ≥ 4% Over 9-Month Observation Period

	% Patients with IDWG ≥ 4%	*P*-Value vs. Baseline Period
Baseline Period	37.7	N/A
3 months	34.4	N.S.
6 months	33.5	N.S.
9 months	28.9	<0.001

Abbreviations: N/A, not applicable; N.S., not significant

Conclusions

By implementing the PEAK methodology and encouraging team focus on the defined outcome of IDWG ≥ 4%, all of the clinics that implemented the program achieved a decrease in the frequency of IDWG in their patient populations.

References

1. Foley, R.N., Herzog, C.A., and Collins, A.J. Blood pressure and long-term mortality in United States hemodialysis patients: USRDS Waves 3 and 4 Study. Kidney Int. 2002; 62: 1784–1790.
2. Kalantar-Zadeh, K., Regidor, D.L., Kovesdy, C.P. et al. Fluid retention is associated with cardiovascular mortality in patients undergoing long-term hemodialysis. Circulation. 2009; 119: 671–679.
3. Wanner, C., Amann, K., Shoji, T. The heart and vascular system in dialysis. The Lancet. 2016; 388: 276-84.

Acknowledgments

The authors wish to thank Dena E. Cohen, PhD, of DaVita Clinical Research for editorial contributions in preparing this poster.

Correspondence: atef.altaher@davita.com

Poster available at www.davitaclinicalresearch.com

American Society of Nephrology Kidney Week, October 23 - 28, 2018; San Diego, CA

© 2018 DaVita Inc. All rights reserved. Proprietary. May not be copied, reprinted, or distributed without the permission of DaVita Inc.