Effect of Hemofiltration on Anemia, Adequacy, and Bone Mineral Disease in Hemodialysis Patients

Mostafa A. Abdelkhalek,[1,2] Tarek M. Demerdash,[1,3] Mohammed M. Assem,[1,3] Muhammad Awais,[1] Mahmoud M. Shaheen,[1,3] Ayman Sabri,[1] Hany A. Salama,[1] Ibrahim A. Jurbran,[1] Saad S. Alobaili,[1] Wisam H. Al-Badr,[1] Abdulkareem Alsuwaida,[1] Abdullah Kashgary[1,4]

[1]DaVita Saudi Arabia, Riyadh, Saudi Arabia; [2]Mansoura University, Mansoura, Egypt; [3]Cairo University, Cairo, Egypt; [4]King Abdulaziz University, Jeddah, Saudi Arabia

Introduction

- Anemia, hyperphosphatemia, and low Kt/V (< 1.2) are significant problems among patients treated with hemodialysis (HD) in Saudi Arabia.[1]
- Hemofiltration (HF) is a treatment option that combines the benefits of diffusion and convection.
 - Although it was first described in the 1970's,[2] it is not widely utilized.
- Previous studies did not observe a benefit of HF compared to HD with respect to outcomes such as all-cause mortality, cardiovascular mortality, or hypotension during dialysis.[3]
- However, the impact of HF on other clinically important outcomes has not been addressed.

Objective

To compare dialysis adequacy, anemia, and bone mineral disease parameters among contemporary patients treated with HD versus HF in Saudi Arabia

Methods

Patients and Timeframe

- This was a retrospective, observational study.
- Included patients were those who, between January 2015 and May 2018, were age > 18 years, were treated at a large dialysis organization (LDO) operating in Saudi Arabia, had dialysis vintage > 90 days, and who provided written consent that their medical records be analyzed in this study.
- Patients dialyzing using any vascular access other than an arteriovenous fistula (AVF) were excluded, as were those in whom HF treatment was interrupted, or who utilized HF as a treatment modality for < 90 days.
- The study adhered to the Declaration of Helsinki and was approved by the King Fahd Medical City Institutional Review Board.

Analysis

- Demographic variables were assessed as of the time of dialysis initiation with the LDO.
- Among patients who initiated HF, values for Kt/V, phosphate, calcium, parathyroid hormone, hemoglobin, ferritin, transferrin saturation, and erythropoietin dose were assessed immediately prior to the start of HF, and again after 90 days of HF treatment.
 - These values were assessed on an analogous timeframe for patients treated with HD.
- Continuous variables were compared across groups using t-tests and Mann-Whitney tests as appropriate based on data distribution. Chi-square or Fisher exact tests were used for comparisons of categorical variables. The cut-off for statistical significance was considered as $P < 0.05$ throughout.
- All analyses were conducted using SPSS version 17 (SPSS, Inc, Chicago, IL, USA).

Results

Patient Selection and Characteristics

- Of 1843 patients receiving dialysis at the LDO during the study period, 1115 patients met all study inclusion/exclusion criteria: 900 treated with HD and 215 treated with HF (Figure 1).
- Patients treated with HF were similar to those treated with HD with respect to gender and prevalence of diabetes. Prevalence of hypertension was somewhat lower in the HF group compared to the HD group (Table 1).

Mineral Bone Disease Outcomes

- Among patients treated with HF, serum phosphate levels were lower after 90 days of HF treatment compared to prior to HF treatment. However, even after HF treatment, phosphate levels remained higher than those observed in patients treated with HD (Figure 2A).
- Among patients treated with HF, serum calcium levels were lower after 90 days of HF treatment compared to prior to HF treatment. Levels after 90 days of HF treatment were comparable to those observed in patients treated with HD (Figure 2B).
- HF treatment did not significantly affect parathyroid hormone levels. Levels following 90 days of HF treatment were similar to those observed among patients treated with HD (Figure 2C).

Figure 1: Selection of Study Patients

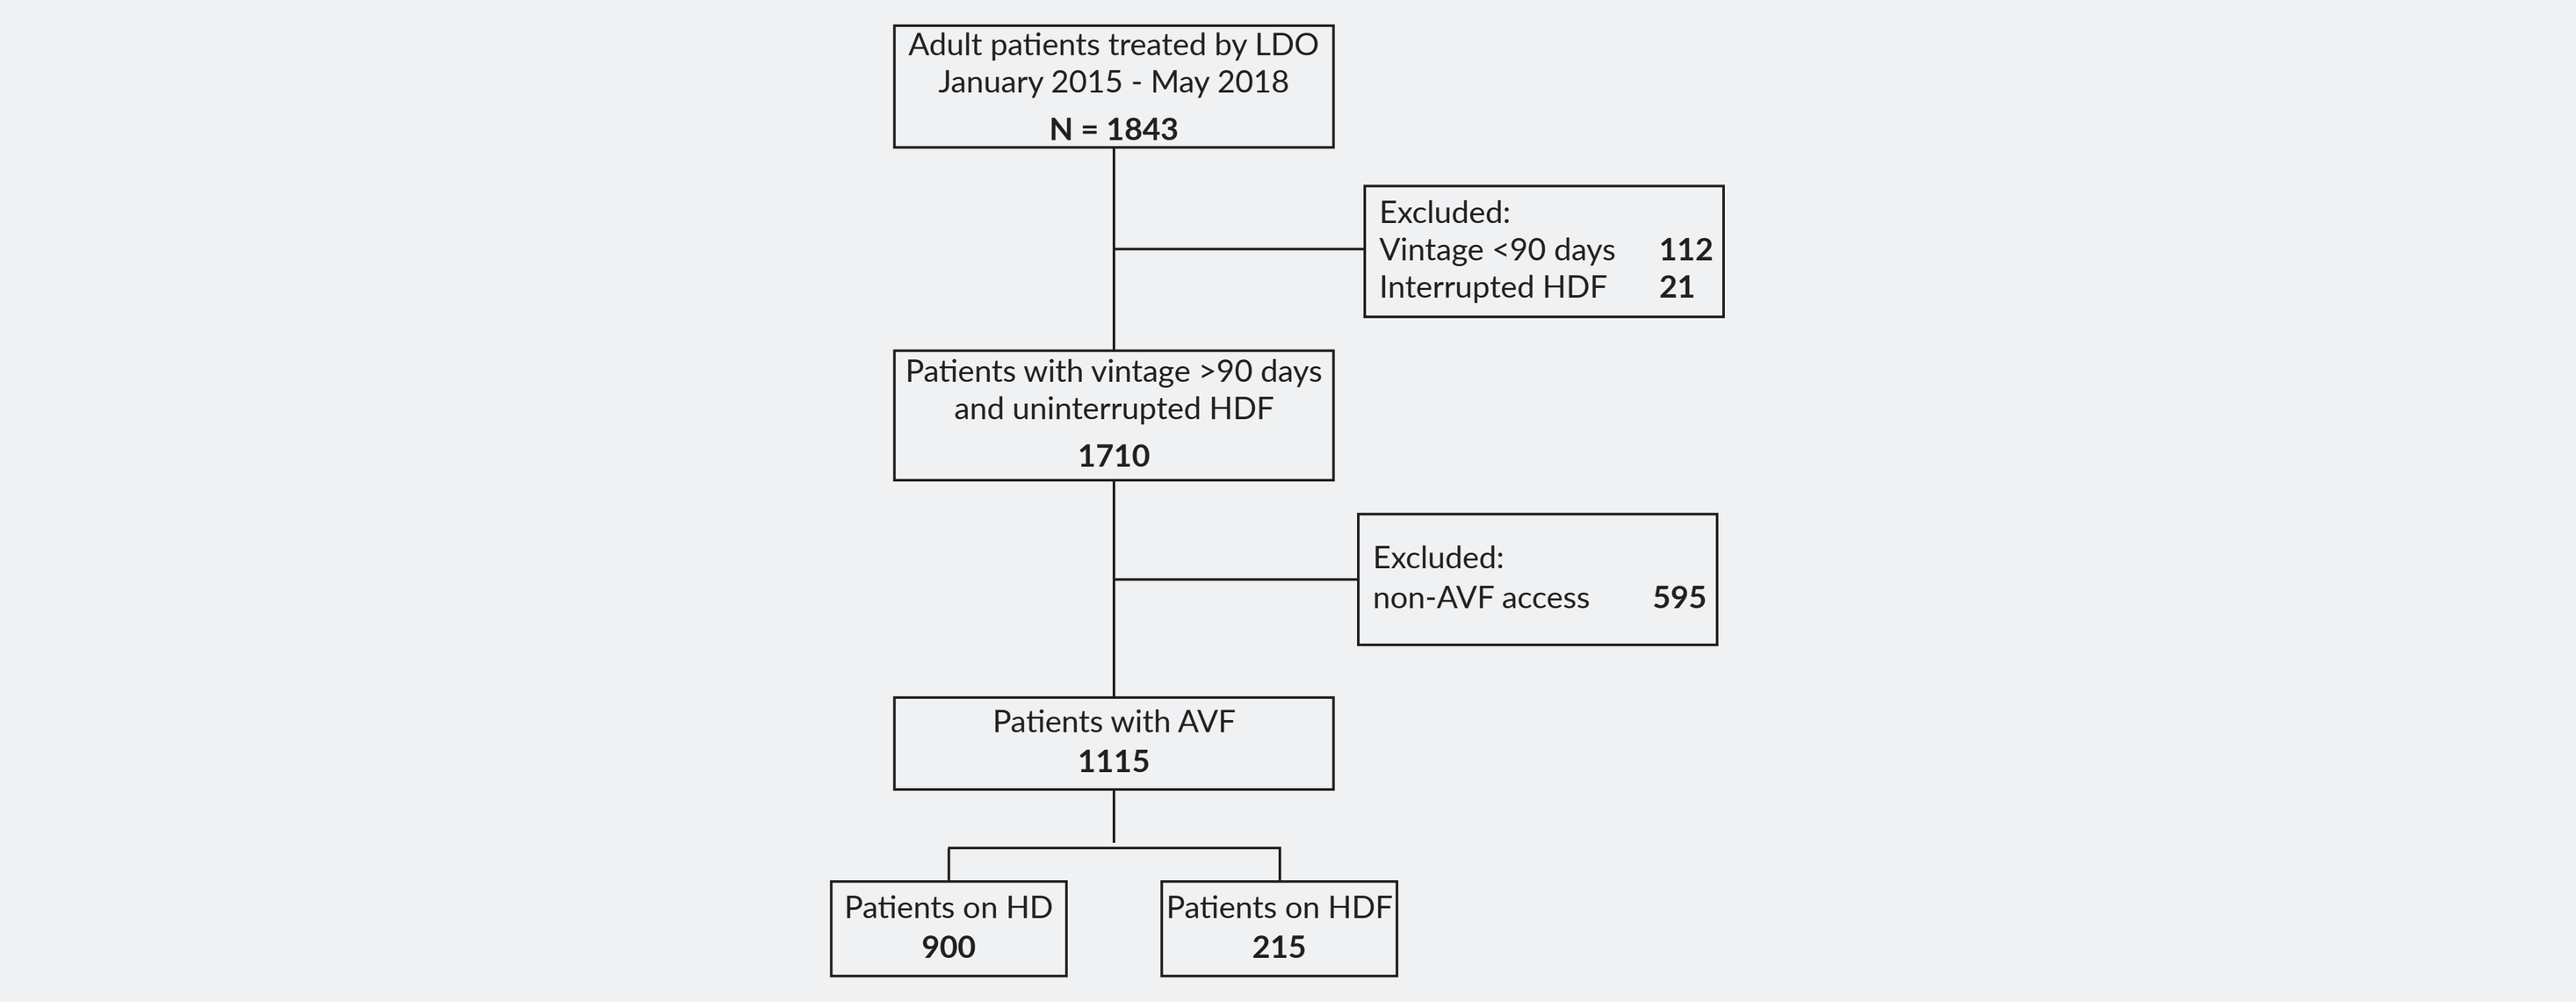
Adult patients treated by LDO
January 2015 - May 2018
N = 1843
Excluded:
Vintage <90 days 112
Interrupted HDF 21
Patients with vintage >90 days and uninterrupted HDF
1710
Excluded:
non-AVF access 595
Patients with AVF
1115
Patients on HD
900
Patients on HDF
215

Table 1: Patient Characteristics by Treatment Group

	Overall N = 1115	Hemodialysis N = 900	Hemofiltration N = 215
Sex, n (%)			
Male	594 (53.3)	482 (53.6)	112 (52.1)
Female	521 (46.7)	418 (46.4)	103 (47.9)
Diabetes, n (%)	387 (34.7)	307 (34.1)	80 (37.2)
Hypertension, n (%)	876 (78.6)	726 (80.7)	150 (69.8)

Anemia Outcomes

- Hemoglobin levels did not change significantly after 90 days of HF, and were somewhat higher in patients after 90 days of HF than patients treated with HD (Figure 3A).
- No significant differences were observed between any groups with respect to either ferritin or saturated transferrin (Figure 3B and C).
- Erythropoiesis-stimulating agent use was markedly lower after 90 days of HF treatment compared to prior to HF initiation; no difference was observed between patients treated with HD and patients after 90 days of HF (Figure 3D).

Figure 2: Mineral Bone Disease Outcomes

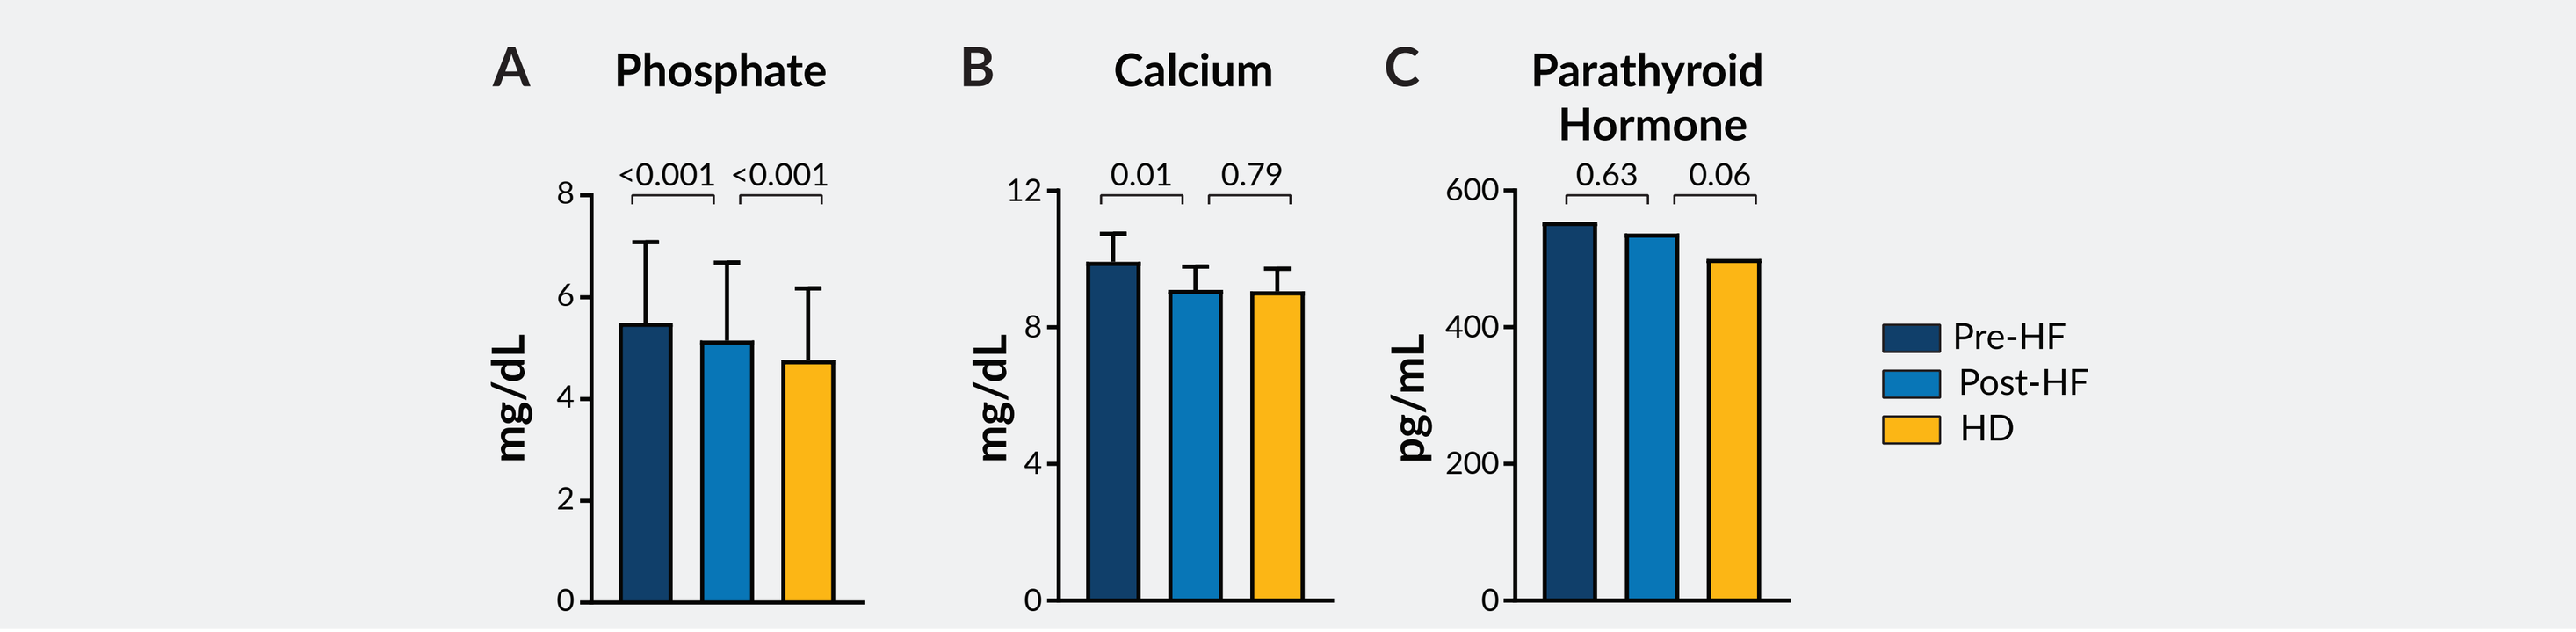
A Phosphate
<0.001 <0.001
mg/dL
B Calcium
0.01 0.79
mg/dL
C Parathyroid Hormone
0.63 0.06
pg/mL
Pre-HF
Post-HF
HD

A) mean ± standard deviation for serum phosphate are shown. B) mean ± standard deviation for serum calcium are shown. C) median values of parathyroid hormone are shown. Interquartile ranges are: Pre-HF, 557; Post-HF, 580; HD, 471.

Abbreviations: HD, hemodialysis; Pre-HF, prior to initiation of hemofiltration; Post-HF, 90 days after initiation of hemofiltration

Figure 3: Anemia Outcomes

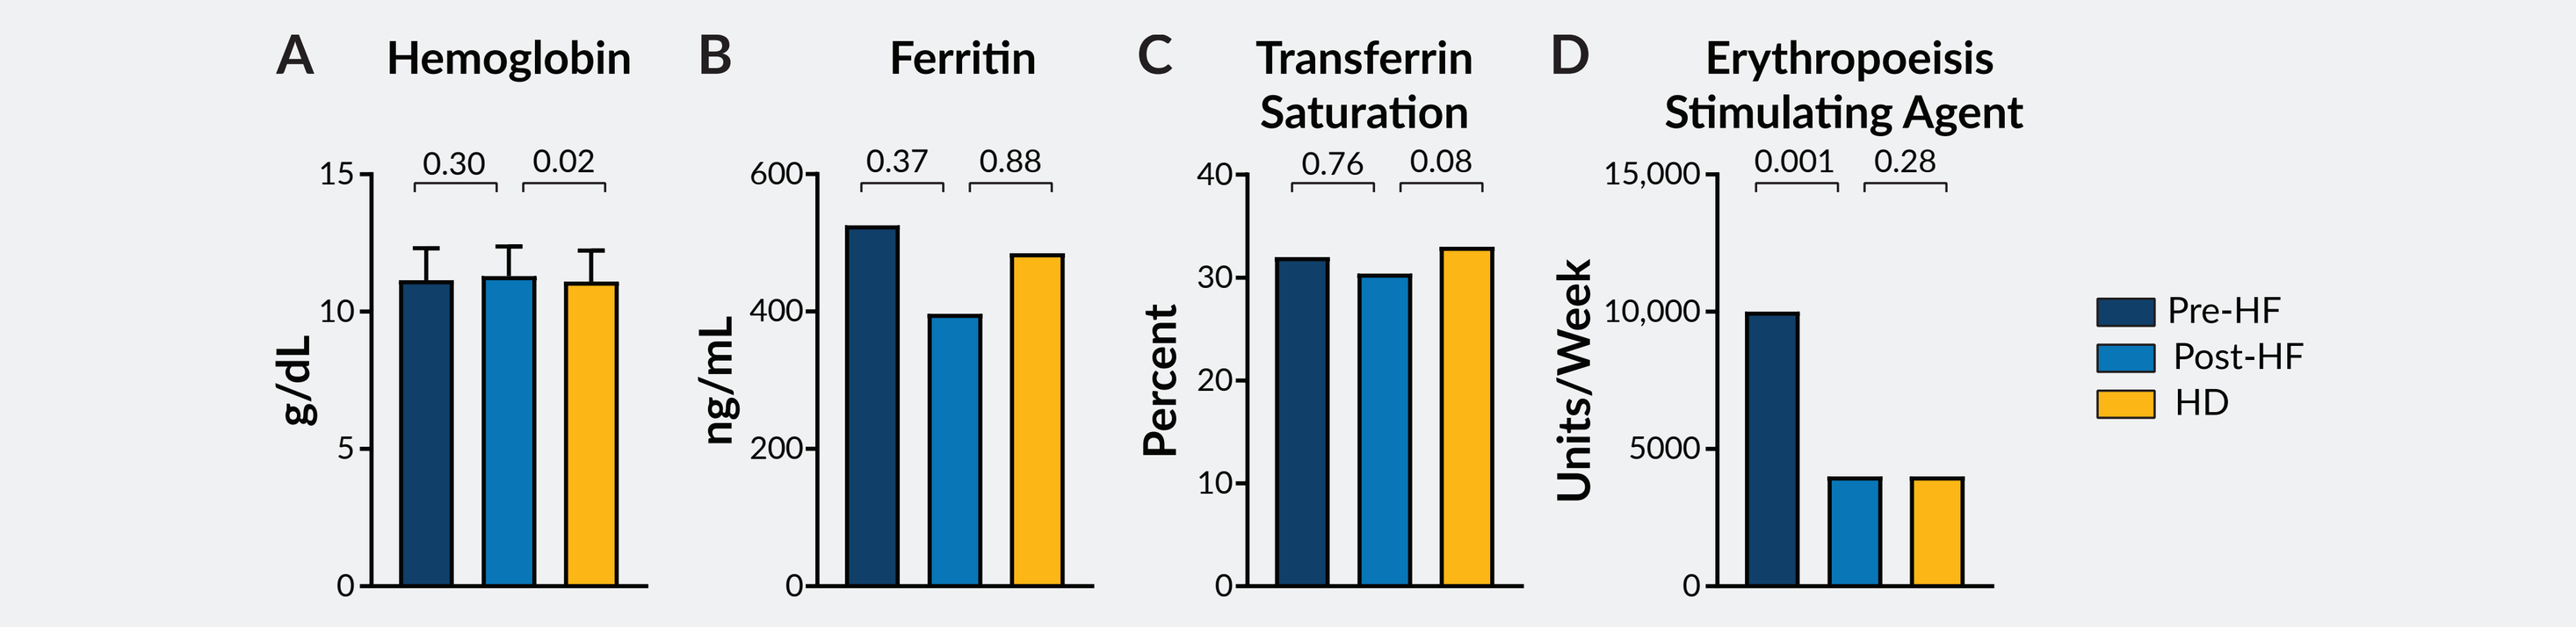
A Hemoglobin
0.30 0.02
g/dL
B Ferritin
0.37 0.88
ng/mL
C Transferrin Saturation
0.76 0.08
Percent
D Erythropoeisis Stimulating Agent
0.001 0.28
Units/Week
Pre-HF
Post-HF
HD

A) mean ± standard deviation for hemoglobin are shown. B) mean ± standard deviation for ferritin are shown. C) median values of transferrin saturation are shown. Interquartile ranges are: Pre HF, 26; Post-HF, 25; HD, 15. D) median values of erythropoiesis-stimulating agent dose per week are shown. Interquartile ranges are: Pre HF, 12,000; Post-HF, 6000; HD, 8000.

Abbreviations: HD, hemodialysis; Pre-HF, prior to initiation of hemofiltration; Post-HF, 90 days after initiation of hemofiltration

Figure 4: Nutrition and Dialysis Adequacy

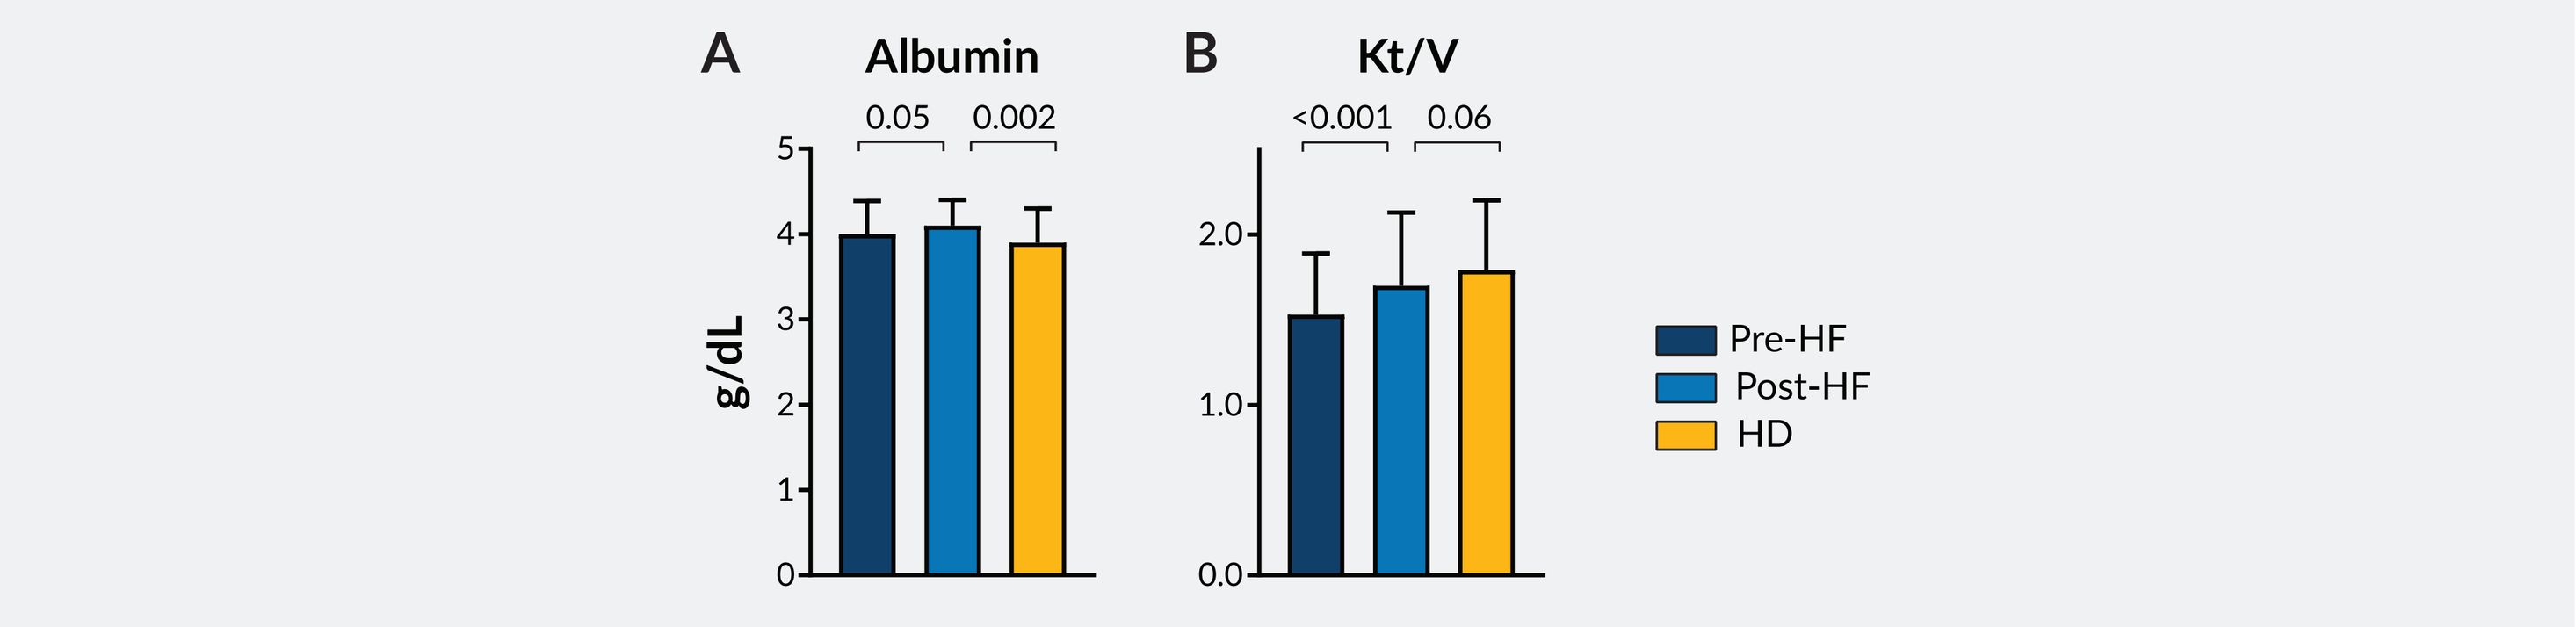
A Albumin
0.05 0.002
g/dL
B Kt/V
<0.001 0.06
Pre-HF
Post-HF
HD

A) mean ± standard deviation for serum albumin are shown. B) mean ± standard deviation for Kt/V are shown.

Abbreviations: HD, hemodialysis; Pre-HF, prior to initiation of hemofiltration; Post-HF, 90 days after initiation of hemofiltration

Nutrition and Dialysis Adequacy Outcomes

- Albumin levels were significantly higher after 90 days of HF treatment compared to prior to HF treatment initiation. Albumin levels after 90 days of HF treatment were higher than those in patients treated with HD (Figure 4A).
- Kt/V was significantly higher after 90 days of HF treatment compared to prior to HF treatment initiation. Kt/V after 90 days of HF treatment did not differ from that of patients treated with HD (Figure 4B).

Conclusions

- Among patients who do not achieve dialysis adequacy targets or adequate phosphorus control despite receiving standard of care treatment, HF represents a treatment option that may improve dialysis outcomes.
- In this study, 90 days of HF treatment was associated with higher Kt/V, lower erythropoiesis-stimulating agent dose, higher hemoglobin, and lower serum phosphate.
- Prospective, randomized-controlled trials are needed to validate these findings.

References

1. Dialysis Outcomes and Practice Patterns Study (DOPPS) 2018.
2. Henderson, LW, Colton, CK, and Ford, CA. Kinetics of hemodiafiltration II: Clinical characterization of a new blood cleansing modality. *J Lab Clin Med* 85(3):372-91, 1975.
3. Nistor, I, et. al. Haemodiafiltration, haemofiltration, and haemodialysis for end stage kidney disease. *Cochrane Database Syst Rev*, 5: CD006258, 2015.

Acknowledgments

The authors thank Dena E. Cohen, PhD, of DaVita Clinical Research for editorial contributions in preparing this poster.

Correspondence: abdullah.kashgary@davita.com

Poster available at www.davitaclinicalresearch.com

American Society of Nephrology Kidney Week, October 23 - 28, 2018; San Diego, CA

© 2018 DaVita Inc. All rights reserved. Proprietary. May not be copied, reprinted, or distributed without the permission of DaVita Inc.