Daily Versus Post-Dialysis Administration of Calcimimetics for the Treatment of Secondary Hyperparathyroidism in Hemodialysis Patients: An Interventional, Multi-Center Study

Tarek M. Demerdash,[1,2] Mostafa A. Abdelkhalek,[1,3] Mohammed M. Assem,[1,2] Mahmoud M. Shaheen,[1,2] Farida Elsafty,[1] Ibrahim A. Jubran,[1] Saad S. Alobaili,[1] Abdullah Kashgary,[1,4] Wisam H. Al-Badr,[1] Abdulkareem Alsuwaida[1]

[1]DaVita Saudi Arabia, Riyadh, Saudi Arabia; [2]Cairo University, Cairo, Egypt; [3]Mansoura Faculty of Medicine - Mansoura University, Mansoura, Egypt; [4]King Abdulaziz University, Jeddah, Saudi Arabia

Introduction

- Secondary hyperparathyroidism (SHPT) is a common and important complication of end-stage renal disease (ESRD), with impacts on bone health and associations with cardiovascular morbidity and mortality.[1,2]
- Use of the calcimimetic drug cinacalcet can lower serum levels of intact parathyroid hormone (iPTH), and decrease risk of bone fractures.[3,4]
- Cinacalcet is typically administered as a daily oral medication. However, adherence to cinacalcet among HD patients is imperfect, particularly because the medication may lead to gastrointestinal side effects.
- Because cinacalcet has a half-life of over 30 hours,[5] administration of the drug at thrice-weekly in-center treatments may be an effective strategy to achieve SHPT control among hemodialysis patients.

Objective

To assess the effectiveness of supervised, thrice-weekly administration of cinacalcet in controlling SHPT and mineral bone disorder (MBD) among patients with ESRD treated with hemodialysis.

Methods

- This was a multi-center study in adult hemodialysis patients treated at centers operated by a large dialysis organization in Saudi Arabia. All participants gave informed consent prior to starting the study. The work was approved by the King Fahd Medical City Institutional Review Board, and was carried out in accordance with the Declaration of Helsinki.
- Included patients were adults treated with hemodialysis for at least 6 months with proven SHPT and treated with daily cinacalcet.
 - Patients with previous parathyroidectomy, baseline corrected calcium < 8.4 mg/dL, diagnosis of tertiary hyperparathyroidism, and patients having other causes of hyper- and hypo-calcemia were excluded.
- All patients in the study were shifted to thrice-weekly administration of cinacalcet at the conclusion of each hemodialysis session.
 - Cinacalcet dose was titrated every 4 weeks in 30-mg increments to achieve desired iPTH level.
 - Dose was held if maximum dose was reached; if corrected serum calcium decreased below 8 mg/dL; or if any adverse effects were noted.
- Pre-dialysis iPTH, serum calcium, phosphorus, and alkaline phosphatase; doses of phosphate binders; and dose of vitamin D analogs were compared at baseline and at 6 months after switching to thrice-weekly cinacalcet administration.
- Comparisons were made using t-tests, Mann-Whitney tests, Chi-square tests, or Fisher exact tests as appropriate. All analysis was performed with SPSS (Chicago, IL, USA).

Results

Patient Characteristics and Medication Use

- A total of 201 patients who met the inclusion/exclusion criteria consented to participate in the study (Table 1).
- The utilization of relevant medications other than cinacalcet was recorded at baseline and at 6 months after switching to thrice-weekly cinacalcet (Table 2).
 - Use of calcium supplements and activated vitamin D did not change between baseline and 6-months post-switch.
 - Use of sevelamer was incrementally higher at 6-months post-switch.

Table 1: Patient Characteristics at Baseline

	Enrolled Patients N = 201
Sex, n (%)	
Male	99 (49.3)
Female	102 (50.7)
Age, years, mean ± SD	49.3 ± 14.4
Duration of pre-trial cinacalcet treatment, months, mean [min - max]	16.3 [6 - 41]

Abbreviations: max, maximum; min, minimum; SD, standard deivation

Table 2: Use of Other Relevant Medications During the Study Period

	Baseline	6 Months Post-Switch	*P*
Calcium, mg/day			
mean	2214	2324	0.50
[min - max]	[600 - 5400]	[600 - 5400]	
Sevelamer, mg/day			
mean	4257	4480	0.05
[min - max]	[800 - 7200]	[800 - 7200]	
Paricalcitol, mcg/week			
mean	16	16	0.59
[min - max]	[5 - 45]	[5 - 45]	

Abbreviations: max, maximum; min, minimum

SHPT Parameters

- The dose of cinacalcet declined over the study period, from a mean of 130 mg/day at baseline to a mean of 87 mg/day 6 months after switching to the thrice-weekly dosing schedule (Figure 1).
- During this same period, iPTH levels declined from a mean of 1079 pg/mL at baseline to a mean of 987 pg/mL (Figure 1).
 - No differences were observed in either serum calcium or serum phosphate.
 - Alkaline phosphatase levels declined from a mean of 190 U/L at baseline to a mean of 171 U/L at 6 months.
- The number of patients with iPTH ≤ 600 pg/mL increased from 36 at baseline to 63 after 6 months (Figure 2).
 - The numbers of patients with iPTH > 600 - ≤ 1200pg/mL declined from 86 to 77, and the number with iPTH > 1200 declined from 79 to 61.

Figure 1: Mineral Bone Disease Parameters Before and After Switching to Thrice-Weekly Cinacalcet Dosing

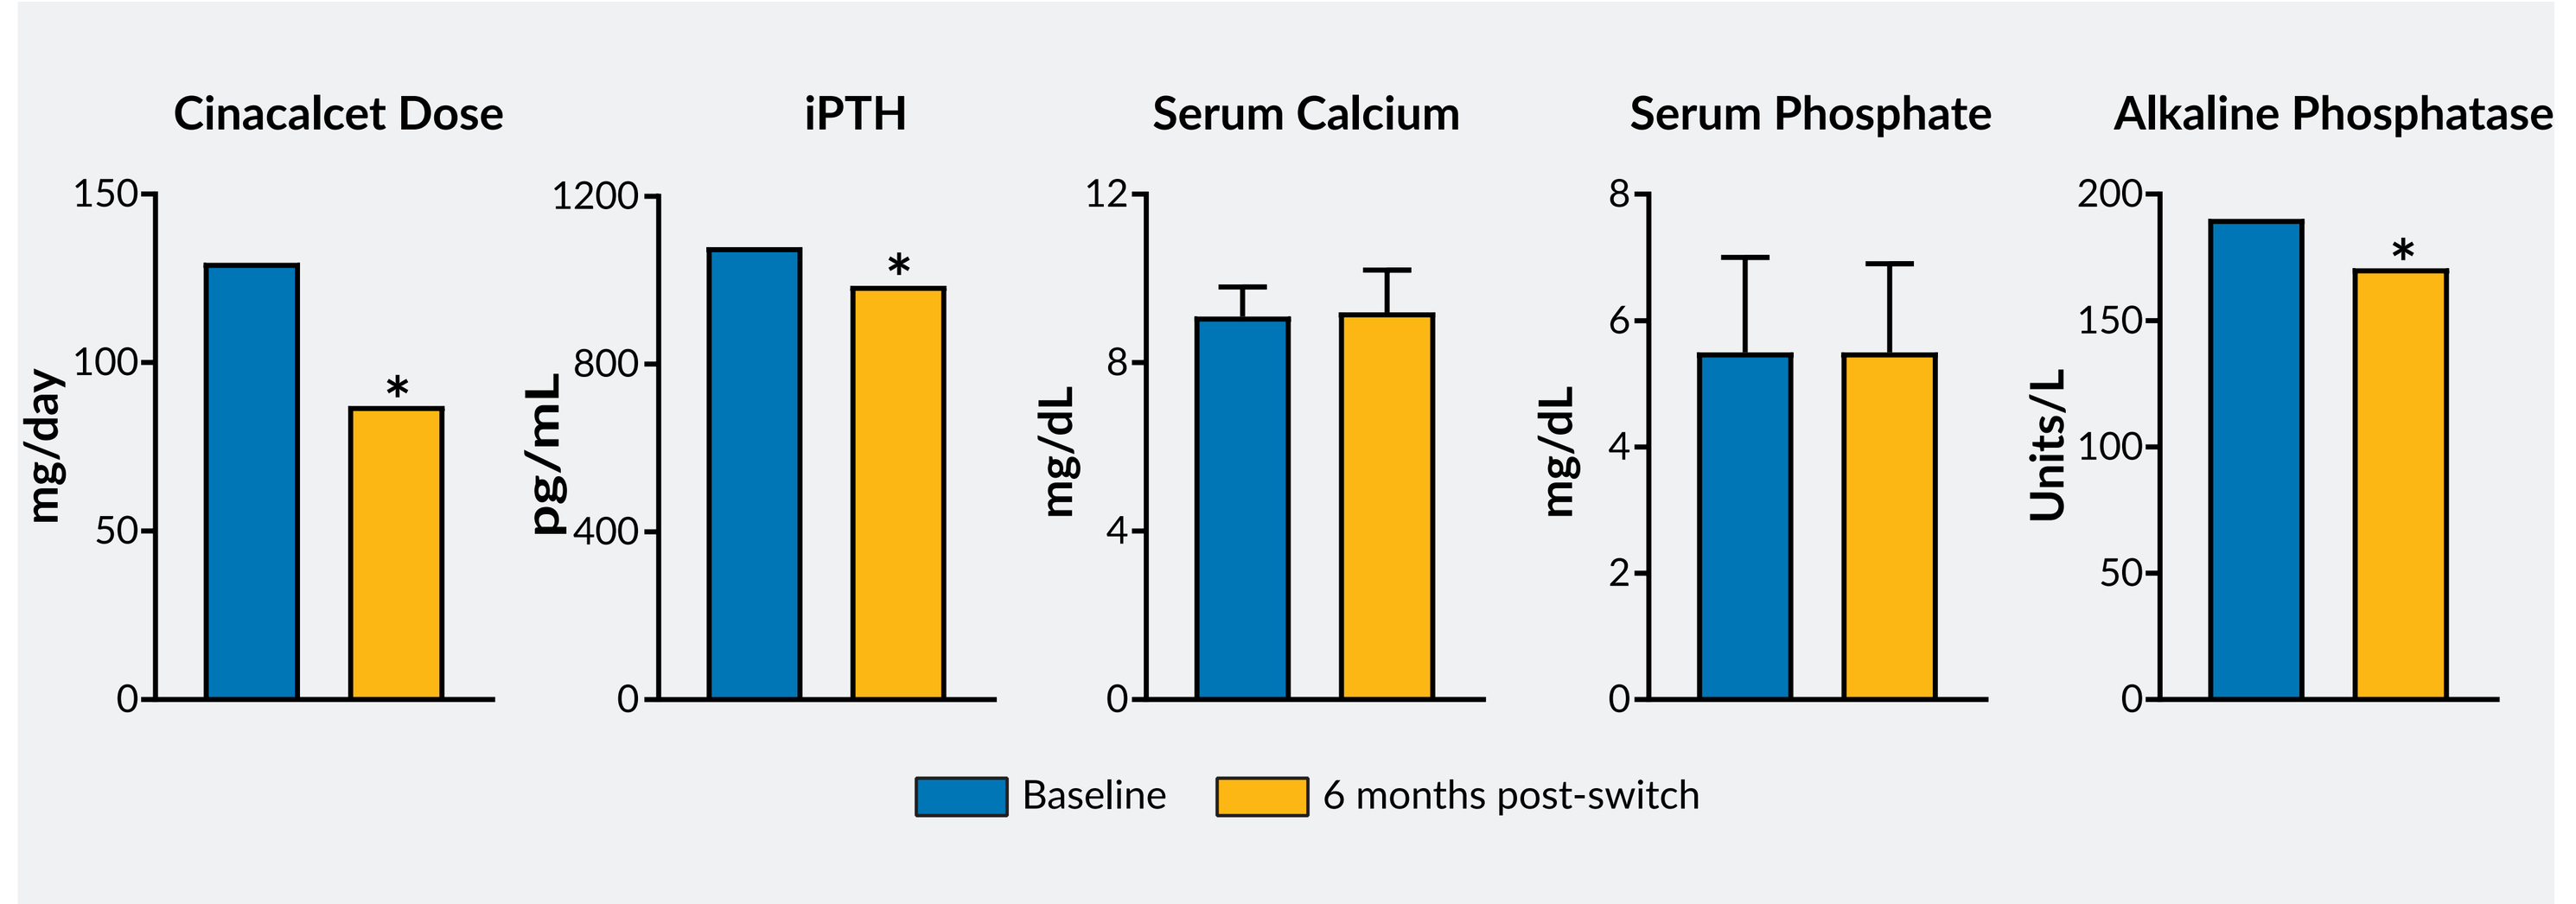
Cinacalcet Dose
iPTH
Serum Calcium
Serum Phosphate
Alkaline Phosphatase
mg/day
pg/mL
mg/dL
mg/dL
Units/L
Baseline
6 months post-switch

Plotted values are means and standard deviations. For variables with no standard deviations shown, the minimum and maximum values are as follows: cinacalcet, baseline 60 - 360; cinacalcet, 6 months 30 - 240; iPTH, baseline 148 - 2500; iPTH, 6 months 99 - 2500; alkaline phosphatase, baseline 44 - 1241; alkaline phosphatase, 6 months 44 - 909. * *P* <0.05.

Figure 2: Change in iPTH Levels After Switching to Thrice-Weekly Cinacalcet

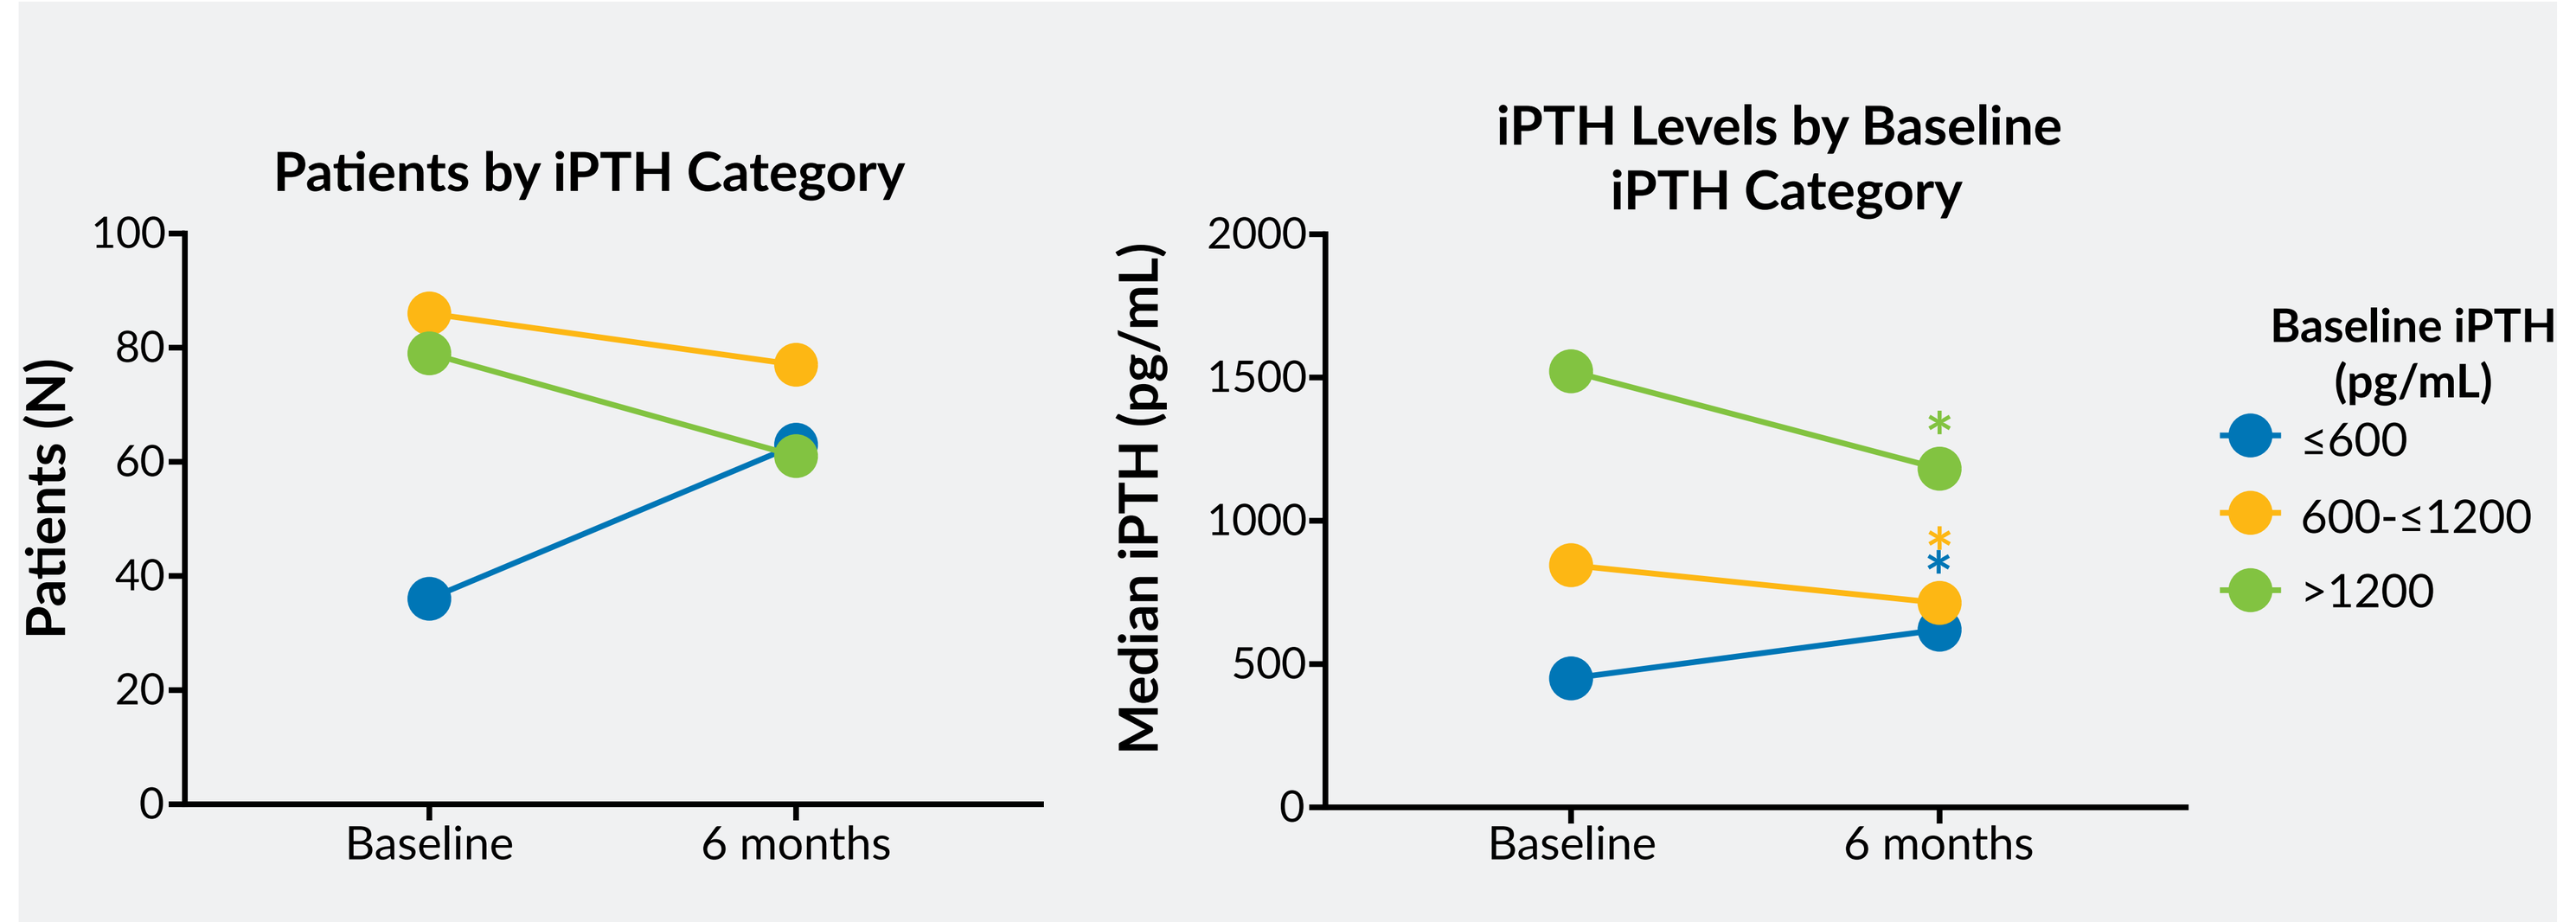
Patients by iPTH Category
Patients (N)
Baseline
6 months
iPTH Levels by Baseline iPTH Category
Median iPTH (pg/mL)
Baseline iPTH (pg/mL)
≤600
600-≤1200
>1200

SHPT Parameters at Baseline and After 6 months of Thrice-Weekly Cinacalcet

- The median (interquartile range) iPTH concentration among patients with a baseline iPTH of ≤ 600 pg/mL increased from 450 (180.5) to 621 (577.5) pg/mL over the study period (Figure 2).
- Among patients with starting iPTH of > 600 - ≤ 1200 and > 1200 pg/mL, median iPTH declined over the study period.
 - Among patients with starting iPTH of > 600 - ≤ 1200 pg/mL, median iPTH declined from 845 (301) to 714 (577.8) pg/mL.
 - Among patients with starting iPTH of > 1200 pg/mL, median iPTH declined from 1523 (416) to 1183 (658) pg/mL.

Conclusions

- Supervised, thrice-weekly in-center administration of cinacalcet was effective in controlling SHPT among patients with ESRD treated with hemodialysis.
- Thrice-weekly administration of cinacalcet may represent an economically advantageous strategy for managing SHPT.
- Additional, larger-scale clinical trials are needed to verify this finding.

References

1. Tentori, F., et al., Mortality risk for dialysis patients with different levels of serum calcium, phosphorus, and PTH: the Dialysis Outcomes and Practice Patterns Study (DOPPS). *Am J Kidney Dis*, 2008. 52(3): p. 519-30.
2. Melamed, M.L., et al., Changes in serum calcium, phosphate, and PTH and the risk of death in incident dialysis patients: a longitudinal study. *Kidney Int*, 2006. 70(2): p. 351-7.
3. Urena, P., et al., Cinacalcet and achievement of the NKF/K-DOQI recommended target values for bone and mineral metabolism in real-world clinical practice--the ECHO observational study. *Nephrol Dial Transplant*, 2009. 24(9): p. 2852-9.
4. Moe, S.M., et al., Effects of Cinacalcet on Fracture Events in Patients Receiving Hemodialysis: The EVOLVE Trial. *J Am Soc Nephrol*, 2015. 26(6): p. 1466-75
5. Padhi, D. and R. Harris, Clinical pharmacokinetic and pharmacodynamic profile of cinacalcet hydrochloride. *Clin Pharmacokinet*, 2009. 48(5): p. 303-11.

Acknowledgments

The authors wish to thank Dena E. Cohen, PhD, of DaVita Clinical Research for editorial contributions in preparing this poster.

Correspondence: tarek.demerdash@davita.com

Poster available at www.davitaclinicalresearch.com

American Society of Nephrology Kidney Week, October 23 - 28, 2018; San Diego, CA

© 2018 DaVita Inc. All rights reserved. Proprietary. May not be copied, reprinted, or distributed without the permission of DaVita Inc.