Impact of Patient Awareness and Age on Serum Albumin in Chronic Hemodialysis Patients

Shehabaldeen M. Alanazi,[1] Ezzedine Abderrahim,[1] Wisam H. Al-Badr,[1] Abdulkareem Alsuwaida,[1] Suad Abdulaziz,[1] Amani M. Shams,[1] Riham A. Albalbisi,[1] Saad S. Alobaili,[1] Ibrahim A. Jubran,[1] Abdullah Kashgary,[1,2]

[1]DaVita Saudi Arabia, Riyadh, Saudi Arabia; [2]King Abdulaziz University, Jeddah, Saudi Arabia

Introduction

- Hypoalbuminemia is associated with cardiovascular (CV) death in patients on maintenance hemodialysis (HD) and is a marker of malnutrition-inflammation complex syndrome (MICS).
- In addition to dialysis adequacy, serum albumin is influenced by socioeconomic status, level of education, and awareness regarding dietary requirements.
- However, the influence of socioeconomic and educational factors on hypoalbuminemia may vary by national origin of the patient population.

Objective

To determine the associations between a variety of factors and hypoalbuminemia among a population of Saudi Arabian patients treated with maintenance HD

Methods

- This study considered patients, who, as of February 2018, were age > 18 and treated with maintenance HD at a large dialysis organization operating in Saudi Arabia.
- On the basis of serum albumin measurements made during February 2018, patients were allocated to one of two groups: low (albumin ≤ 3.5 g/dL), and high (albumin > 3.5g/dL).
- Socioeconomic data, as well as data about food preparation and intake, were assessed in each group.
- Comparisons between groups were performed using Student's unpaired t-test (continuous variables) or Chi-squared tests (categorical variables) as appropriate.
- Logistic regression was used to identify parameters that were independently associated with serum albumin level.
- Differences with a *P*-value of < 0.05 were accepted as significant.

Results

Table 1: Demographic and Social Characteristics, by Albumin Level

	Albumin ≤ 3.5 g/dL N = 97	Albumin > 3.5 g/dL N = 140	Total N = 237	*P*-Value
Age, years, n (%)				< 0.001
< 40	12 (12.4)	40 (28.6)	52 (22.2)	
40 - < 60	20 (20.6)	64 (45.7)	84 (35.7)	
60 - < 75	43 (44.3)	29 (20.7)	72 (30.6)	
≥ 75	21 (21.6)	6 (4.3)	27 (11.5)	
Sex, n (%)				> 0.99
Male	40 (41.2)	57 (40.7)	97 (40.9)	
Female	57 (58.8)	83 (59.3)	140 (59.1)	
Education Level, n (%)				< 0.001
Illiterate	61 (62.9)	47 (33.6)	108 (45.6)	
Primary	14 (14.4)	24 (17.1)	38 (16)	
Middle & secondary	14 (14.4)	43 (30.7)	57 (24)	
Post Secondary	8 (8.2)	26 (18.6)	34 (14.4)	
Monthly Income, dollars, n (%)				0.31
< 3000	47 (48.5)	55 (39.3)	102 (43.1)	
3000-5000	28 (28.9)	43 (30.7)	71 (29.9)	
> 5000	22 (22.7)	42 (30)	64 (27)	
Accommodation, n (%)				0.32
Ownership	77 (79.4)	102 (72.9)	179 (75.5)	
Other	20 (20.6)	38 (27.1)	59 (24.5)	
Living with, n (%)				0.005
Alone	3 (3.1)	3 (2.1)	6 (2.5)	
Parents	10 (10.3)	38 (27.1)	48 (20.3)	
Married-Family	84 (86.6)	99 (70.7)	183 (77.2)	
Number of family members				
mean ± SD	6.28 ± 3.29	6.60 ± 3.07	6.47 ± 3.16	0.42
range	[0 – 20]	[1 – 17]	[0 – 20]	

Abbreviations: SD, standard deviation.

Table 2: History of ESRD Treatment, Comorbidities, and Related Symptoms, by Albumin Level

	Albumin ≤ 3.5 g/dL N = 97	Albumin > 3.5 g/dL N = 140	Total N = 237	*P*-Value
ESRD duration, years, n (%)				0.08
< 1 Year	32 (33.0)	27 (19.3)	59 (24.9)	
1 - 2 Years	19 (19.6)	26 (18.6)	45 (19.0)	
2 - 5 Years	26 (26.8)	45 (32.1)	71 (29.9)	
> 5 Years	20 (20.6)	42 (30.0)	62 (26.2)	
Diabetes mellitus, n (%)				< 0.001
Yes	54 (55.7)	41 (29.3)	95 (40.1)	
No	43 (44.3)	99 (70.7)	142 (59.1)	
Vascular access, n (%)				0.16
Fistula	73 (75.3)	117 (83.6)	190 (80.2)	
Catheter	24 (24.7)	23 (16.4)	47 (19.8)	
Body mass index, kg/m^2, n (%)				0.03
< 18.5 (underweight)	14 (14.4)	21 (15.0)	35 (14.7)	
18.5 - 25 (normal)	46 (47.4)	41 (29.3)	87 (36.8)	
25 - 30 (overweight)	14 (14.4)	37 (26.4)	51 (21.5)	
≥ 35 (obese)	23 (23.7)	41 (29.3)	64 (27.0)	
Symptoms, n (%)				0.15
No symptoms	35 (36.1)	65 (46.4)	100 (42.2)	
≥ 1 Symptom	62 (63.9)	75 (53.6)	137 (57.8)	
Weight variation (admission/ current), %				
median	0	0	0	0.14
range	[-12.5; 24.8]	[-18.0; 10.9]	[-18.0, 10.9]	

Table 3: Food Procurement and Intake, by Albumin Level

	Albumin ≤ 3.5 g/dL N = 97	Albumin > 3.5 g/dL N = 140	Total N = 237	*P*-Value
Procurement of food ingredients, n (%)				0.03
Myself	19 (19.6)	47 (33.6)	66 (27.8)	
Family member	78 (80.4)	93 (66.4)	171 (72.2)	
Food cooking, n (%)				0.04
Myself	8 (8.2)	26 (18.6)	34 (14.3)	
Other	89 (91.8)	114 (81.4)	203 (85.7)	
Regular food intake on HD days, n (%)				< 0.001
0 - 2 meals/day	62 (63.9)	76 (54.3)	138 (58.2)	
3 - 4 meals/day	35 (36.1)	64 (45.7)	99 (41.8)	
Regular food intake on other days, n (%)				0.18
0 - 2 meals/day	35 (36.1)	44 (31.4)	79 (33.3)	
3 - 4 meals/day	62 (63.9)	96 (68.6)	158 (66.7)	
Diet knowledge, n (%)				< 0.001
Level 1	13 (13.4)	3 (2.1)	16 (6.8)	
Level 2 - 3	48 (49.5)	52 (37.1)	100 (42.2)	
Level 4 - 5	36 (37.1)	85 (60.7)	121 (51)	
Factors with negative impact on diet compliance, n (%)				0.002
No Factors	40 (41.2)	88 (62.9)	128 (54)	
≥ 1 Factor	57 (58.8)	52 (37.1)	109 (46)	
Protein intake, g/kg/day				
mean ± SD	1.10 ± 0.55	1.08 ± 0.32	1.09 ± 0.43	0.74
range	[0.33–5.03]	[0.24–2.03]	[0.24–5.03]	
Energy intake, kcal/day				
mean ± SD	1685 ± 416	1722 ± 495	1707 ± 463	0.55
range	[600–2500]	[300–3000]	[300–3000]	

Abbreviations: SD, standard deviation.

Table 4: Biological Parameters, by Albumin Level

	Albumin ≤ 3.5 g/dL N = 97	Albumin > 3.5 g/dL N = 140	Total N = 237	*P*-Value
Serum creatinine, mg/dL				
mean ± SD	6.99 ± 2.18	9.39 ± 2.73	8.4 ± 2.8	< 0.001
range	[2.72 – 16.46]	[2.98 – 21]	[2.72 – 21]	
Cholesterol, mg/dL				
mean ± SD	149 ± 39	165 ± 40	158.3 ± 40.3	< 0.004
range	[29 – 298]	[86 – 272]	[29 – 298]	
Phosphorus, mg/dL				
mean ± SD	4.17 ± 1.28	4.89 ± 1.31	4.59 ± 1.34	< 0.001
range	[0.9 – 9.5]	[1.66 – 8.9]	[0.9 – 9.52]	
Calcium, mg/dL				
mean ± SD	9.09 ± 0.73	9.12 ± 0.84	9.1 ± 0.8	0.85
range	[7.3 – 12.1]	[6.7 – 12.1]	[6.7 – 12.1]	
nPCR, g/kg/day				
mean ± SD	0.99 ± 0.28	1.10 ± 0.26	1.05 ± 0.29	0.02
range	[0.39 – 2.20]	[0.32 – 1.99]	[0.32 – 2.20]	
Hemoglobin, g/dL				
mean ± SD	10.6 ± 1.2	11.1 ± 1.1	10.89 ± 1.18	0.002
range	[6.8 – 13.3]	[7.7 – 14.7]	[6.8 – 14.7]	
Kt/V				
mean ± SD	1.75 ± 0.45	1.78 ± 0.49	1.77 ± 0.47	0.62
range	[0.59 – 3.13]	[0.96 – 3.90]	[0.59 – 3.90]	
Potassium, mmol/L				
mean ± SD	4.8 ± 0.6	4.99 ± 0.65	4.9 ± 0.63	0.01
range	[3.2 – 6.1]	[3.0 – 6.8]	[3.0 – 6.8]	

Abbreviations: SD, standard deviation.

Figure 1: Independent Factors Associated with Low Serum Albumin

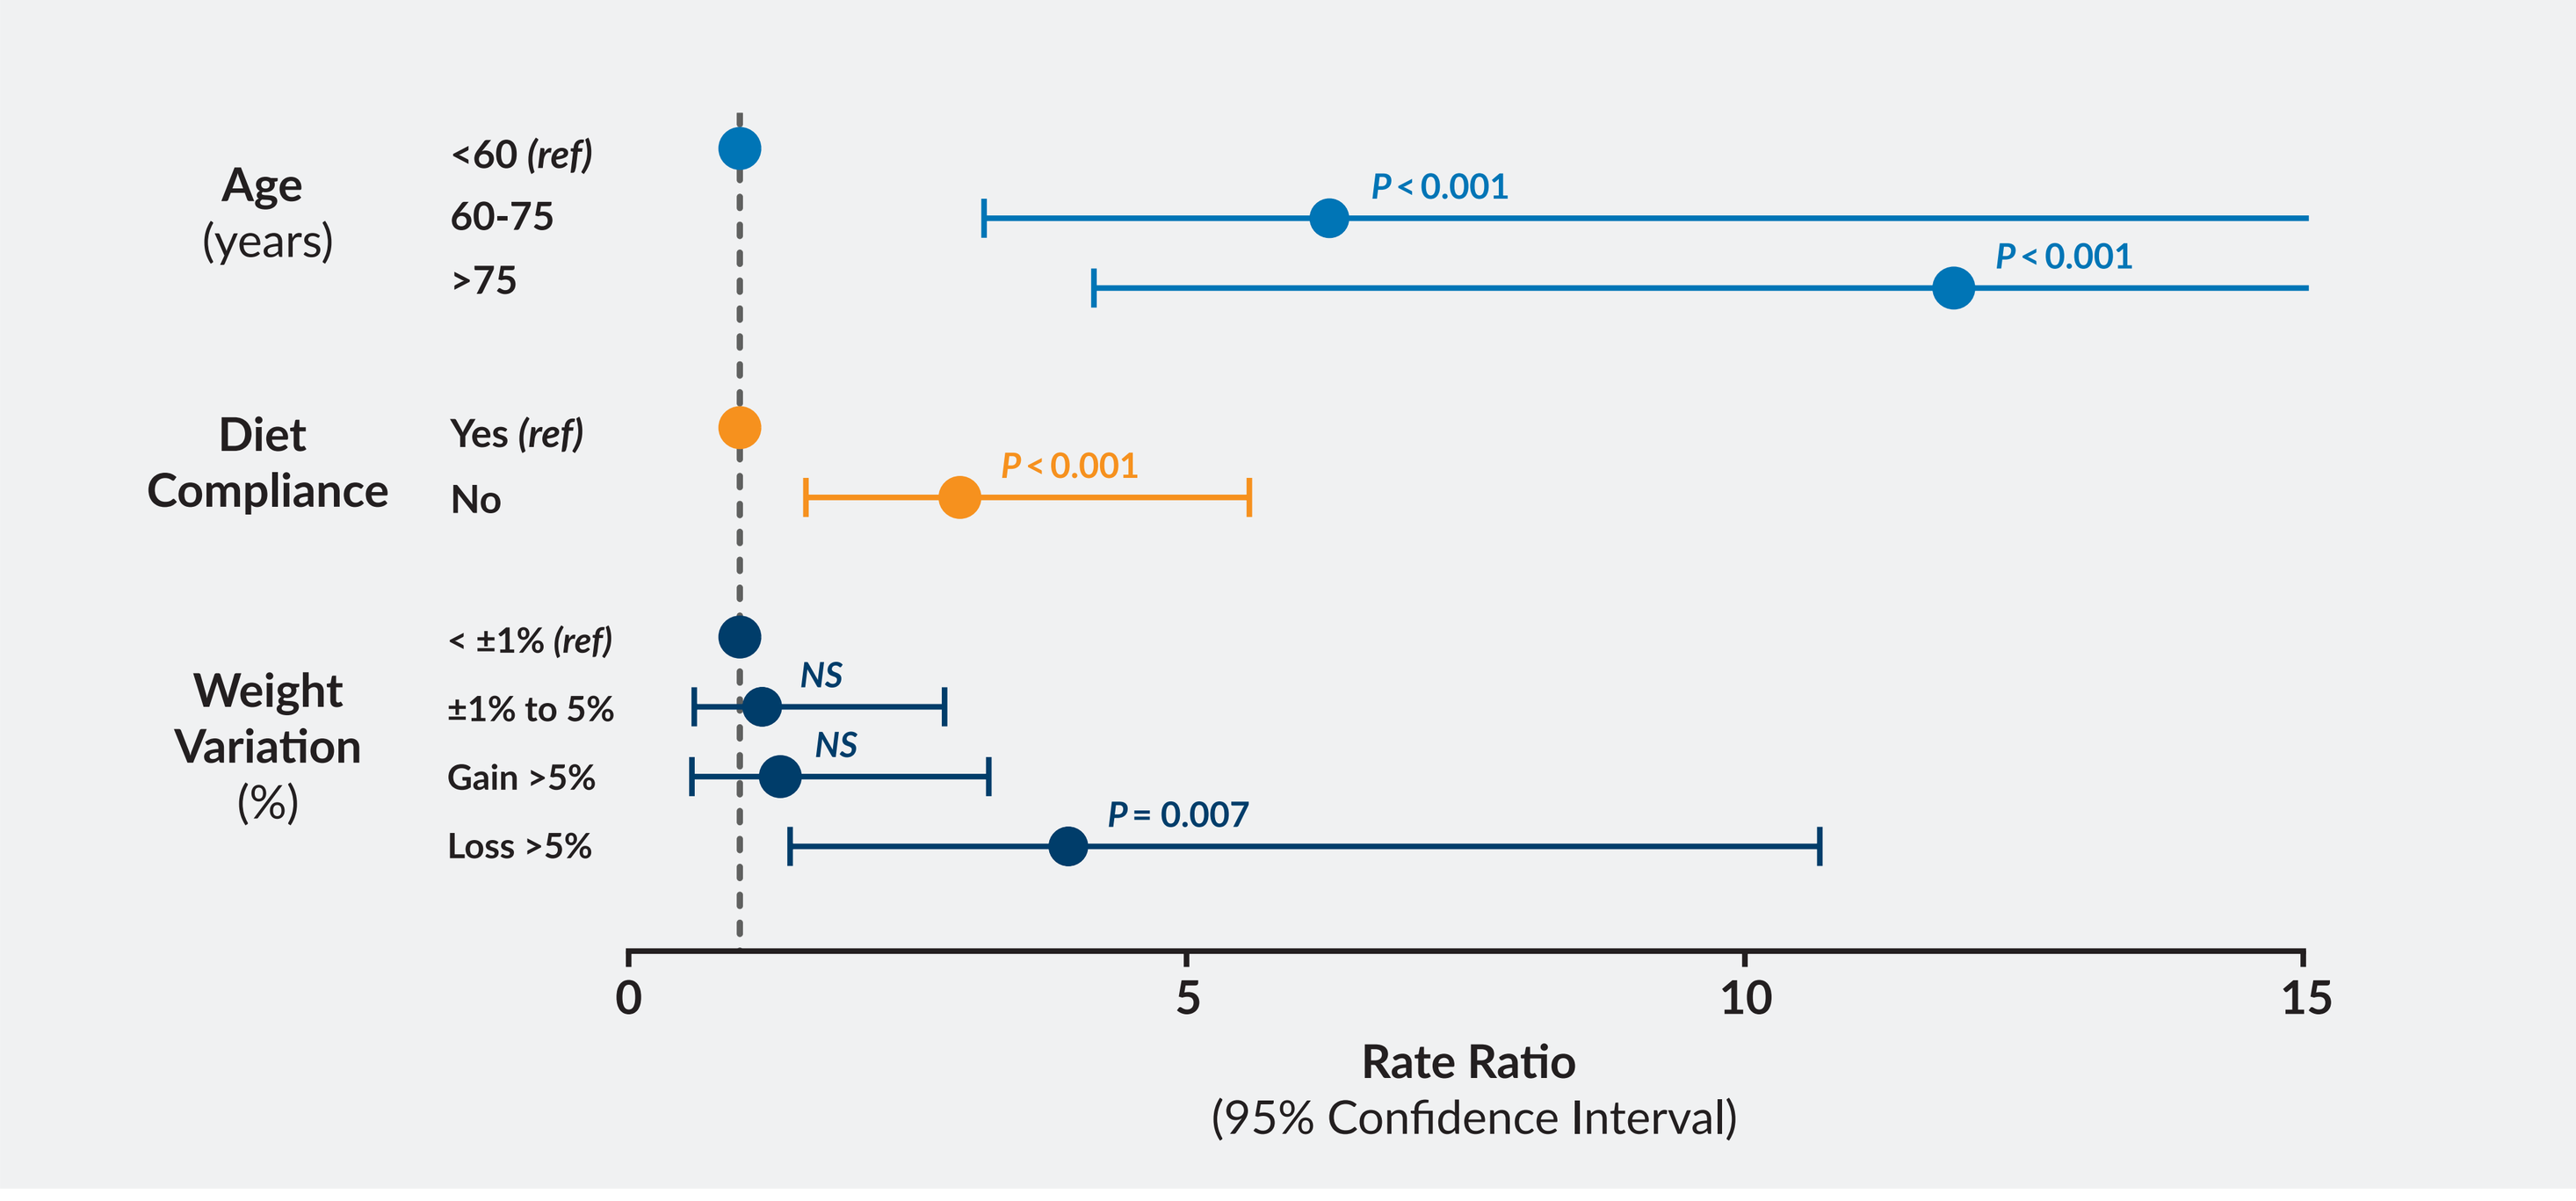
Age (years)
<60 (ref)
60-75
>75
P < 0.001
P < 0.001
Diet Compliance
Yes (ref)
No
P < 0.001
Weight Variation (%)
< ±1% (ref)
±1% to 5%
Gain >5%
Loss >5%
NS
NS
P = 0.007
0
5
10
15
Rate Ratio
(95% Confidence Interval)

Conclusions

- In this study we found that hypoalbuminemia was correlated with patient age and level of education.
- Lower serum albumin is associated with weight loss and poor dietary compliance.
 - No correlation with monthly income or gender was observed.
- Efforts to reduce the number of patients with low albumin through nutritional education and interventions examining the benefits of increasing serum albumin are needed.

References

Kalantar-Zadeh K, Kilpatrick RD, Kuwae N, McAllister CJ, Alcorn H, Kopple JD, Greenland S. Revisiting mortality predictability of serum albumin in the dialysis population: time dependency, longitudinal changes and population-attributable fraction. Neph Dial Transplant 20(9):1880-1888, 2005.

Acknowledgments

Data used in this study form part of the Quality Improvement and Risk Management database, with no funding from external contributers outside DaVita KSA. The authors thank Dena E. Cohen, PhD, and Abigail E. Hunt, PhD, of DaVita Clinical Research for editorial contributions in preparing this poster.

Correspondence: Shehabaldeen.Alanazi@davita.com

Poster available at www.davitaclinicalresearch.com

American Society of Nephrology Kidney Week, October 23 - 28, 2018; San Diego, CA

© 2018 DaVita Inc. All rights reserved. Proprietary. May not be copied, reprinted, or distributed without the permission of DaVita Inc.