A Peritonitis Surveillance System Developed by a Large Dialysis Organization to Improve the Accuracy and Consistency of Peritonitis Rate Reporting in the United States

Martin J. Schreiber, Levi Njord, Meredith L. Zywno, Bram van Hout, Julie A. Rodriguez, Michelle Cassin, James R. Tetley
DaVita Inc, Denver, CO, USA

Introduction

- Peritonitis (PTN) remains a major reason for hospitalization of peritoneal dialysis (PD) patients and may result in modality failure and transfer to in-center hemodialysis.[1-4]
- The ability to accurately and consistently identify PTN events is necessary to make comparisons across and between PD programs and to allow monitoring of efforts to reduce PTN rates.
- In August 2015, a standardized approach to PTN surveillance was introduced by an LDO to improve the consistency and accuracy of PTN event identification across affiliated programs in the US.

Objective

The goal of this analysis was to examine the impact of introduction of the standardized surveillance system on the reporting of PTN events.

Methods

- Under the standardized surveillance system, patients with PTN are identified based on the following 6 hierarchical business rules:
 - 4 standard diagnostic rules for PTN (based on positive culture, positive cell count, abdominal pain)
 - hospitalization for infection
 - receipt of intraperitoneal (IP) antibiotics (> 8 days, > 2 vancomycin doses within 8 days, or > 5 doses of non-vancomycin within 8 days).
 - An exemption process exists for PTN events identified based on the IP antibiotic dosing rule alone. PTN events identified under the IP antibiotic dosing rule during each month are reviewed during the last 7 days of the month and may be reclassified.
- Episodes occurring within 28 days of identification of a prior episode with the same causal organism were considered relapse episodes and were not included in calculations of PTN rates.

Results

Table 1: Summary of Peritonitis Episodes, 2015–2017

Diagnostic Category	PTN Episodes n (%) 2015[a]	2016	2017	Total
Standard diagnostic rules	**1144 (56.8)**	**2650 (58.4)**	**2486 (58.3)**	**6280 (58.1)**
Positive culture + fluids	145 (7.2)	345 (7.6)	389 (9.1)	879 (8.1)
Positive culture + abdominal pain	245 (12.2)	526 (11.6)	453 (10.6)	1224 (11.3)
Positive culture + fluids + abdominal pain	610 (30.3)	1466 (32.3)	1343 (31.5)	3419 (31.6)
Fluids + abdominal pain (sterile episode)	144 (7.2)	313 (6.9)	301 (7.1)	758 (7.0)
Hospitalization for infection	**467 (23.2)**	**1141 (25.2)**	**1185 (27.8)**	**2793 (25.8)**
Receipt of IP antibiotic	**404 (20.1)**	**743 (16.4)**	**594 (13.9)**	**1741 (16.1)**

[a] 2015 data are for August to December only

Abbreviations: IP, intraperitoneal; PTN, peritonitis

Figure 1: Peritonitis Episodes Identified Under the Surveillance System, Aug 2015 to Dec 2017

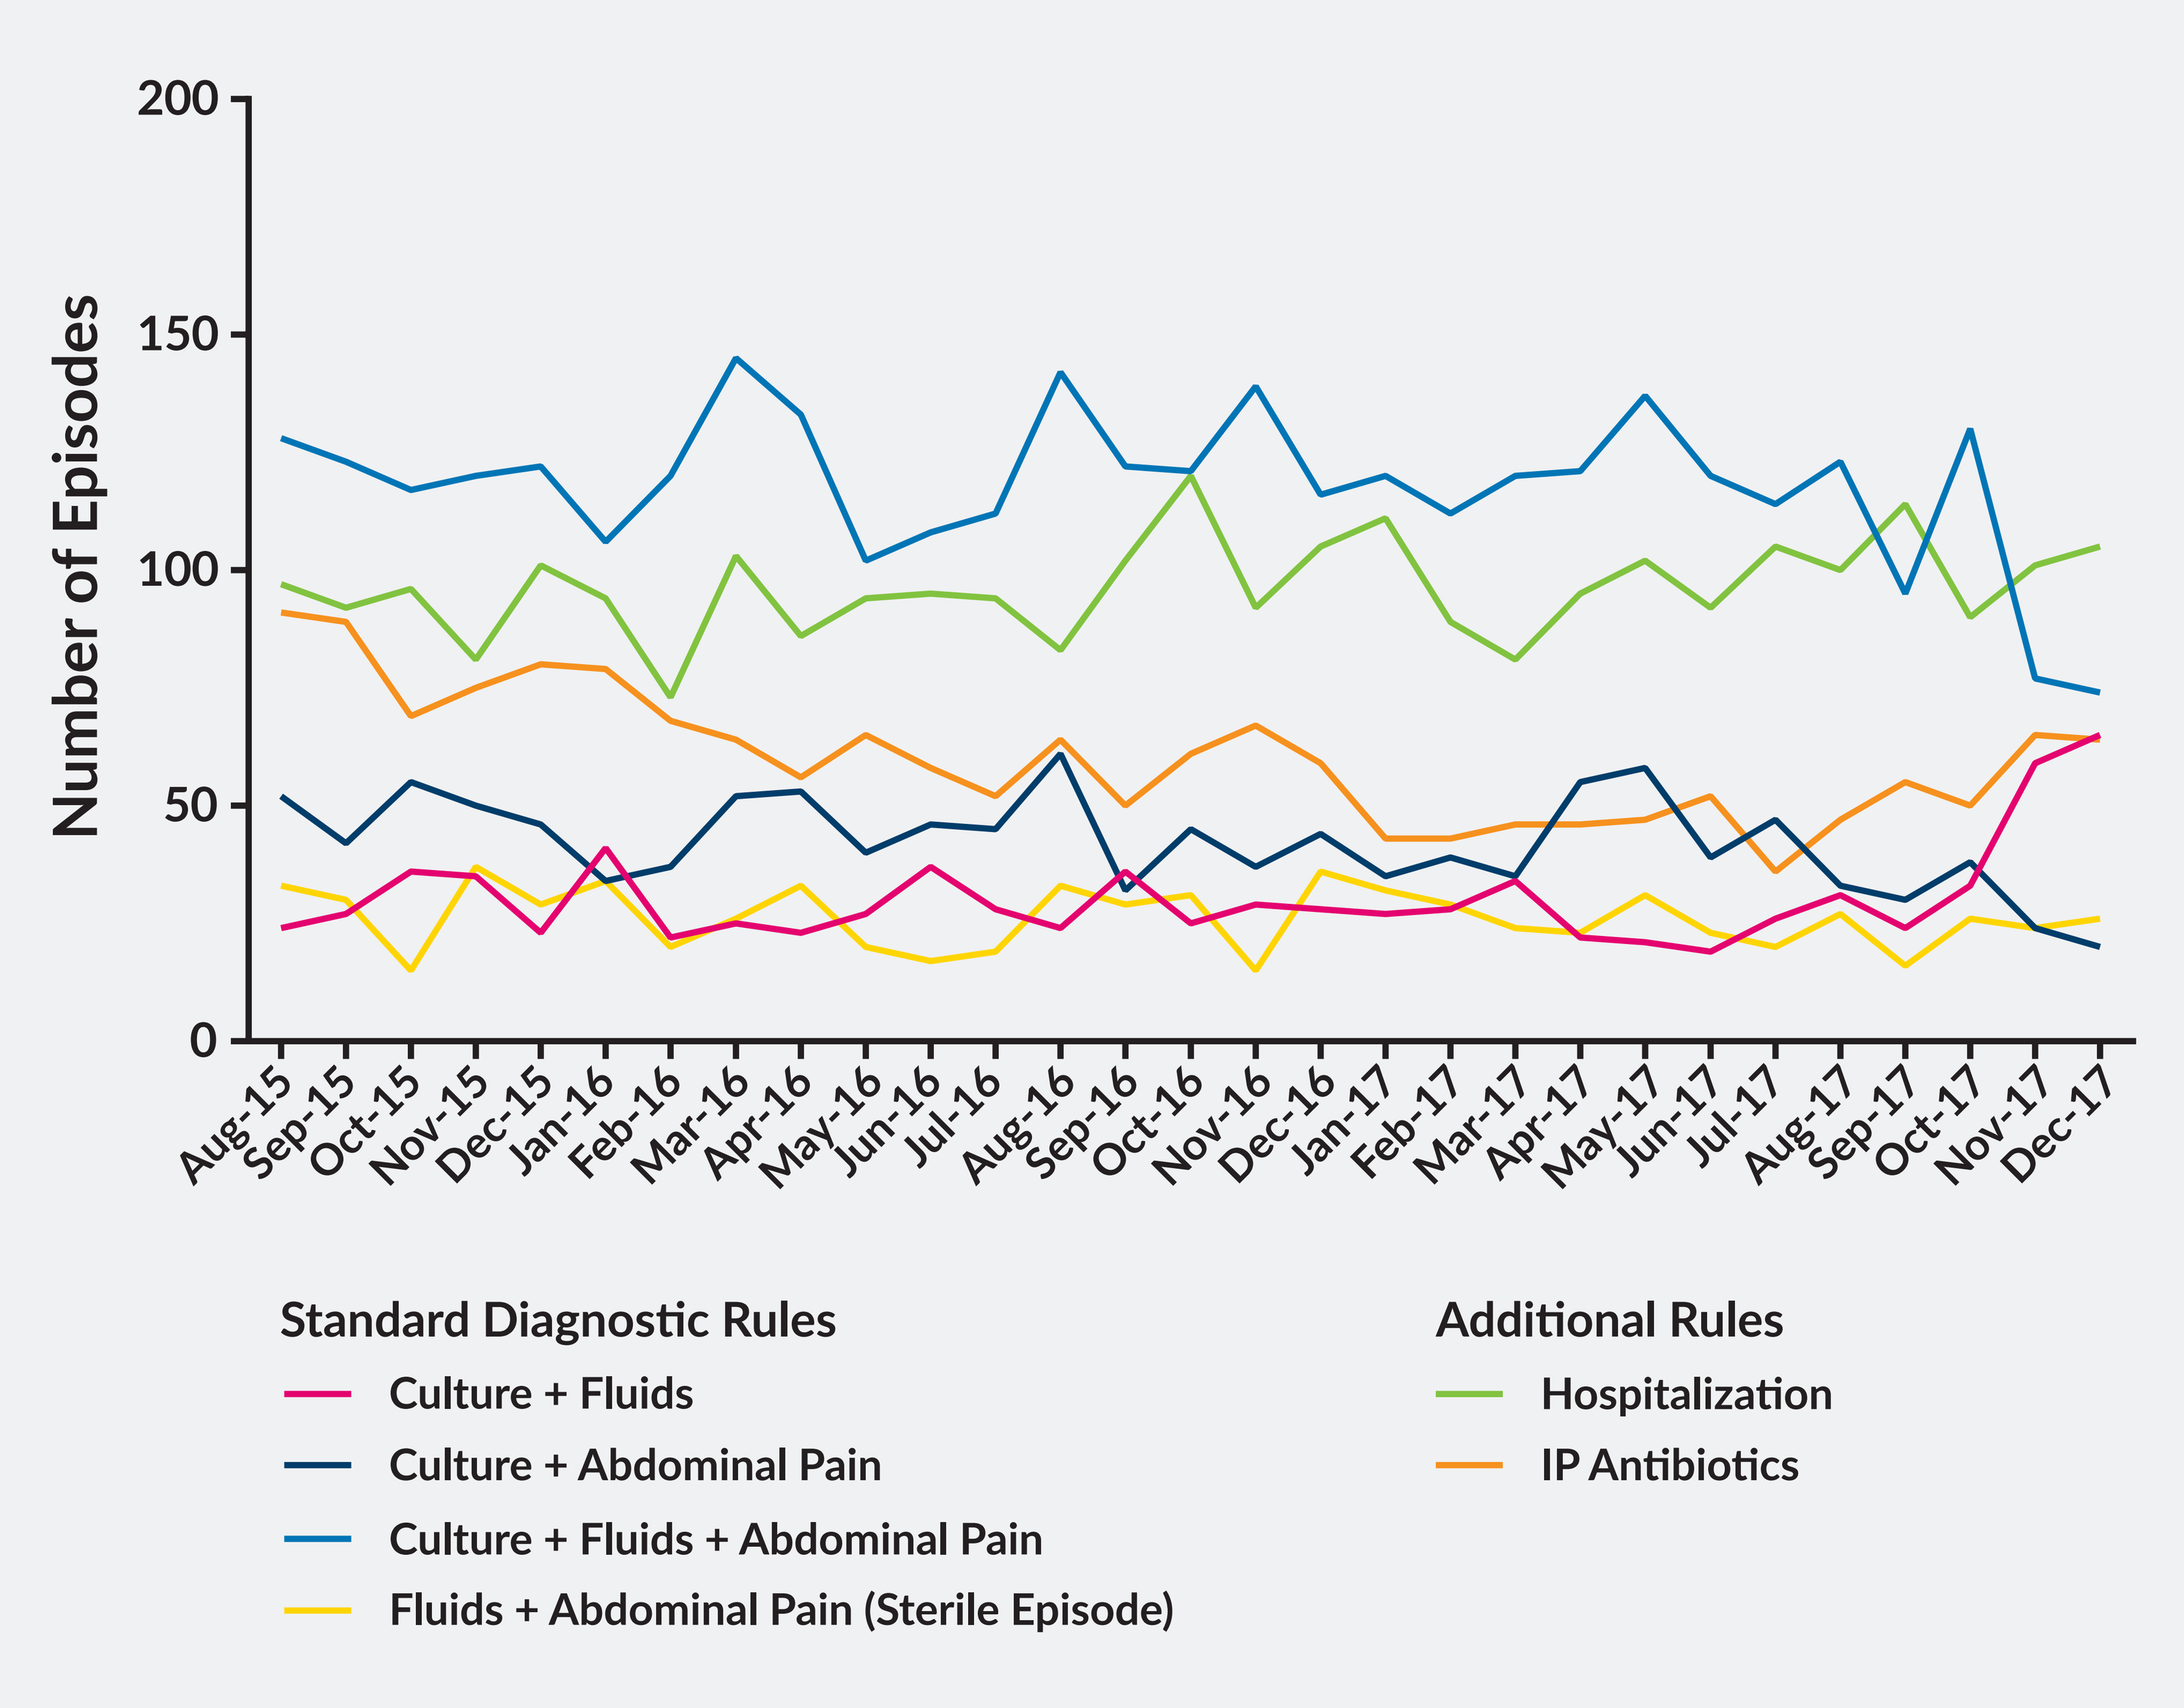
Number of Episodes
200
150
100
50
0
Aug-15 Sep-15 Oct-15 Nov-15 Dec-15 Jan-16 Feb-16 Mar-16 Apr-16 May-16 Jun-16 Jul-16 Aug-16 Sep-16 Oct-16 Nov-16 Dec-16 Jan-17 Feb-17 Mar-17 Apr-17 May-17 Jun-17 Jul-17 Aug-17 Sep-17 Oct-17 Nov-17 Dec-17
Standard Diagnostic Rules
Culture + Fluids
Culture + Abdominal Pain
Culture + Fluids + Abdominal Pain
Fluids + Abdominal Pain (Sterile Episode)
Additional Rules
Hospitalization
IP Antibiotics

Figure 2: PTN Episodes Identified Based on Receipt of IP Antibiotic, Aug 2015 to Dec 2017

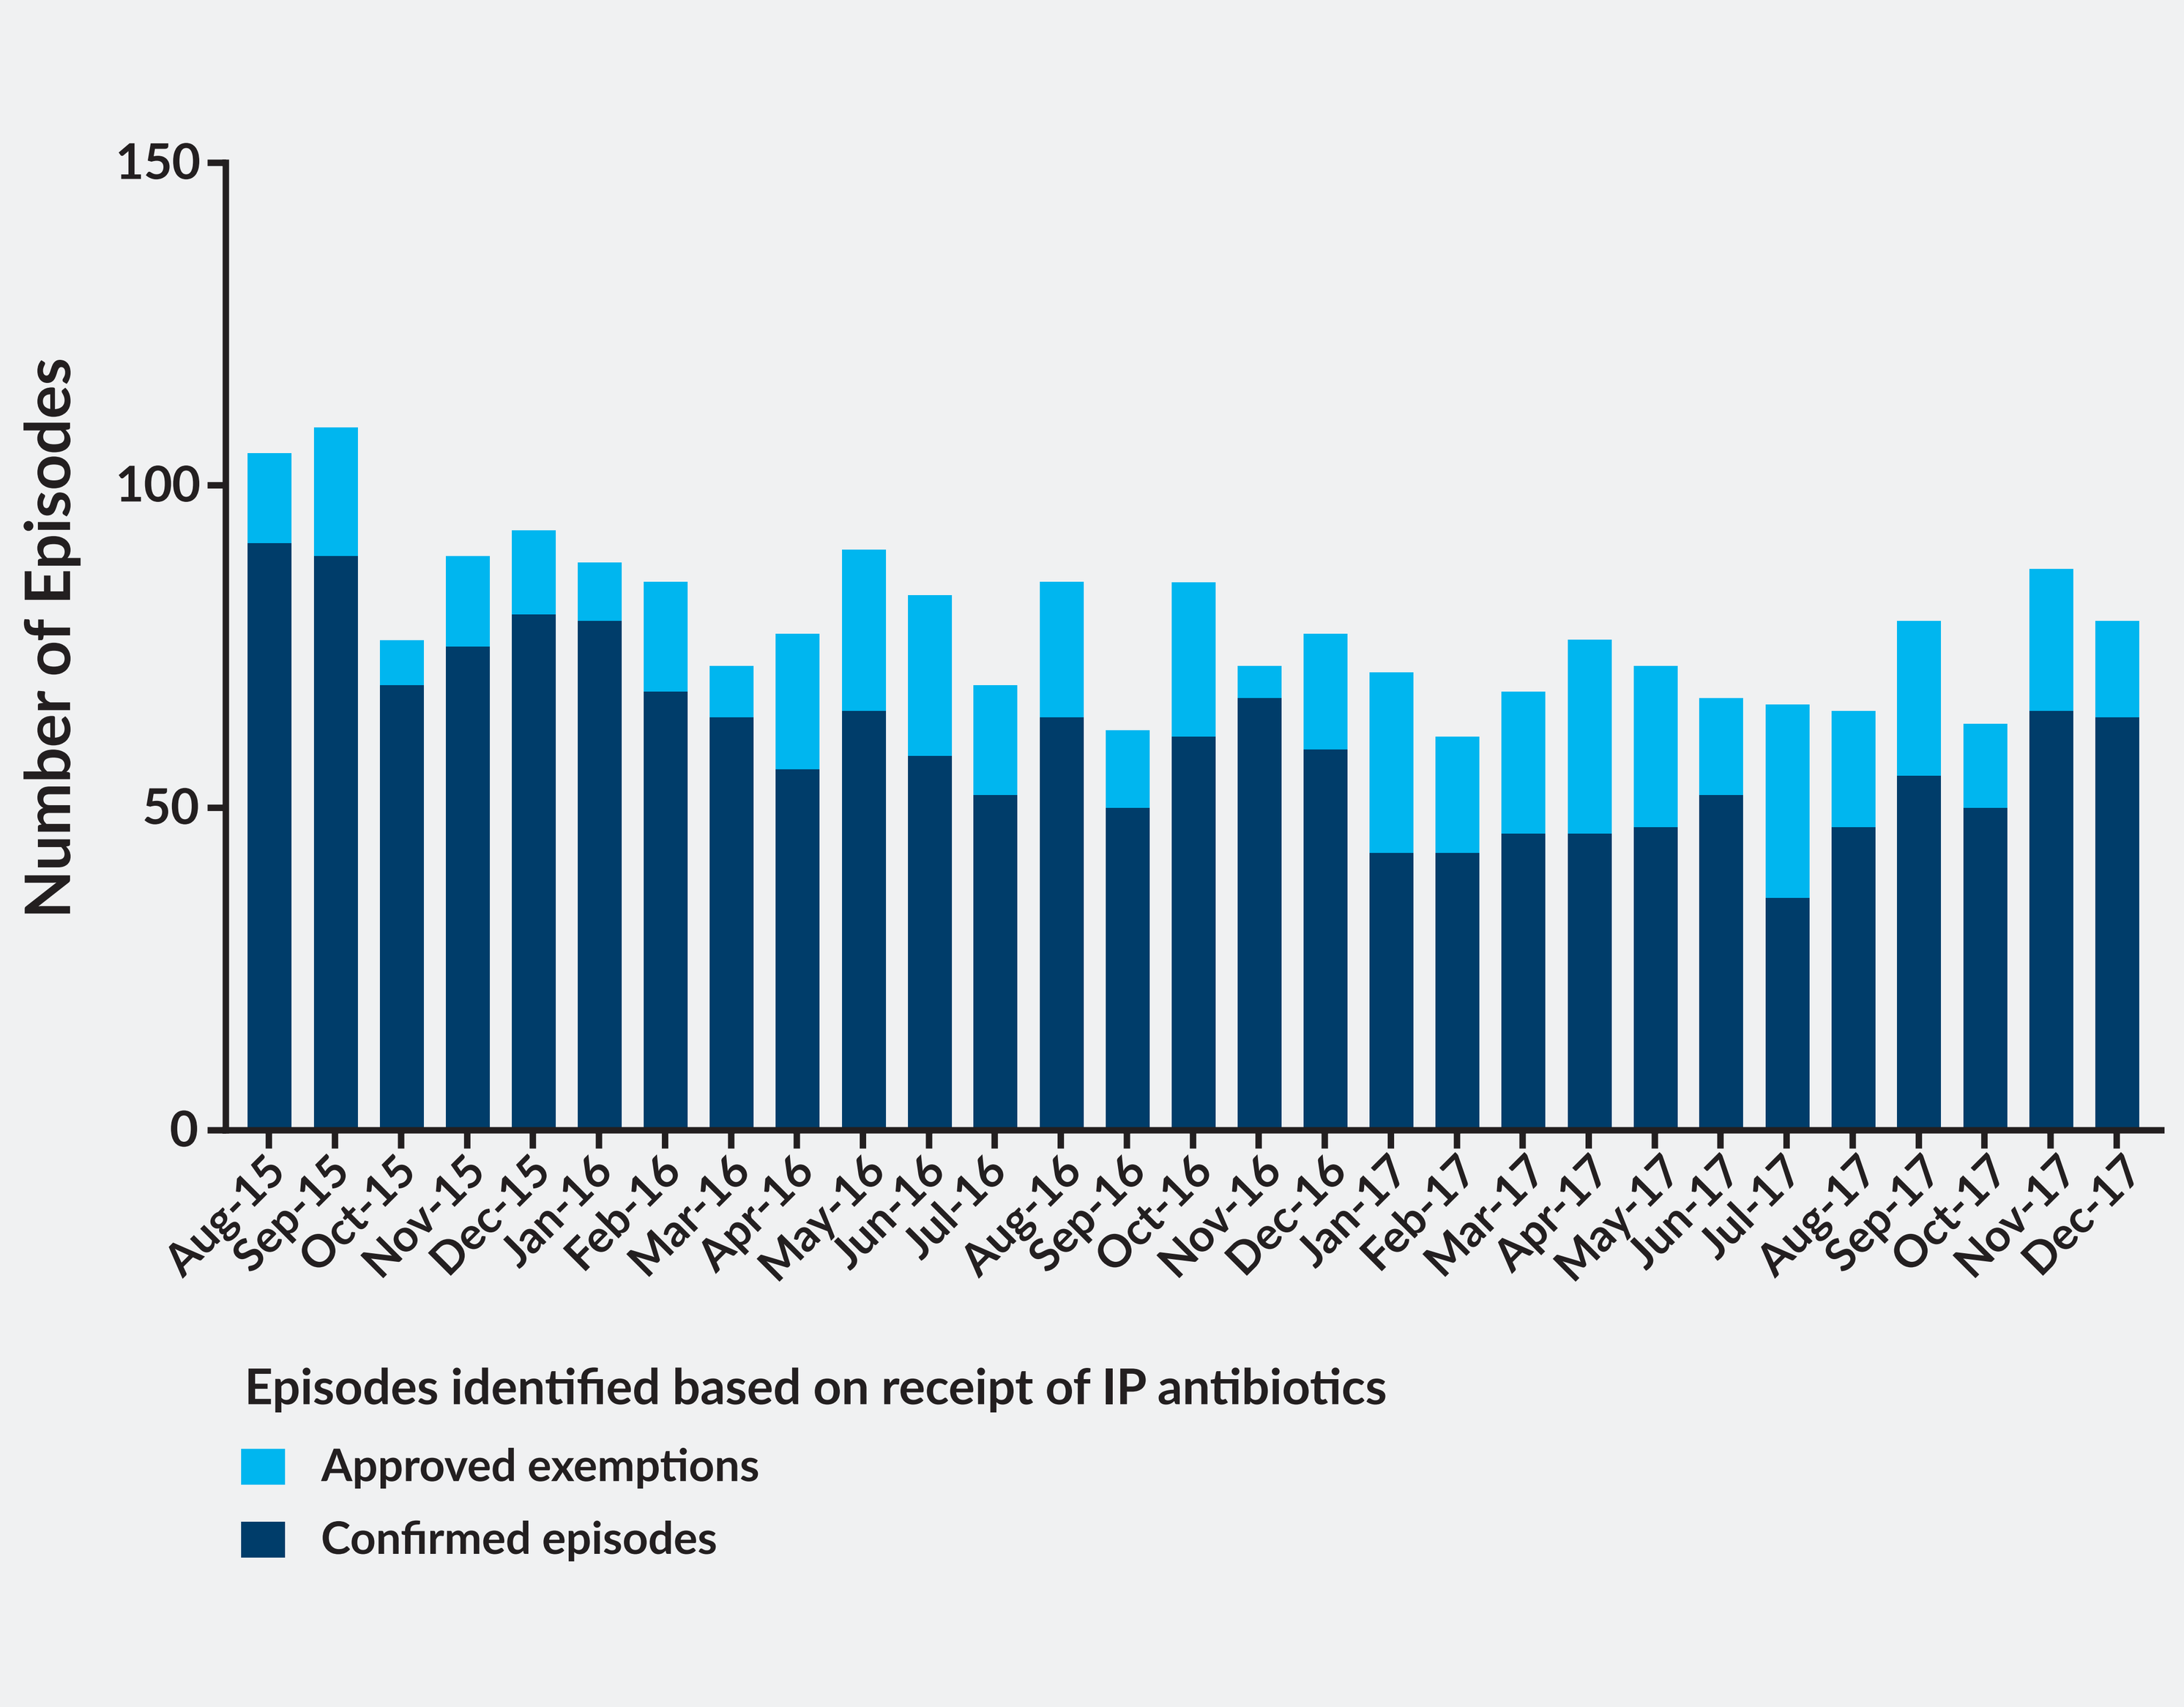
Number of Episodes
150
100
50
0
Aug-15 Sep-15 Oct-15 Nov-15 Dec-15 Jan-16 Feb-16 Mar-16 Apr-16 May-16 Jun-16 Jul-16 Aug-16 Sep-16 Oct-16 Nov-16 Dec-16 Jan-17 Feb-17 Mar-17 Apr-17 May-17 Jun-17 Jul-17 Aug-17 Sep-17 Oct-17 Nov-17 Dec-17
Episodes identified based on receipt of IP antibiotics
Approved exemptions
Confirmed episodes

- During the period August 2015 to December 2017, a total of 10,814 cases of PTN were identified.
 - Among PD patients overall, 88% experienced no episodes of PTN, 10% developed 1 episode/year, and 2% were observed to have 2 or more PTN events/year.
 - The average time to first PTN event was 19.4 months, with 8.6% of events occurring in the first 90 days on PD.
- Over the entire study period, 6280 episodes (58.1%) were identified by standard diagnostic rules, 2793 (25.8%) were identified based on hospitalization, and 1741 (16.1%) were identified based on receipt of IP antibiotics (Table 1 and Figure 1).
 - The proportion of PTN episodes identified by standard diagnostic rules was constant over time.
 - There was a slight increase in the proportion of events identified based on hospitalization and a decrease in the proportion of events identified based on receipt of an IP antibiotic.
 - Exemption filings have remained relatively constant over time (Figure 2).

Conclusions

- The surveillance system provides a consistent approach for the identification of PTN events, combining standard criteria with infection-related hospitalizations and IP antibiotic dosing.
- Use of the PTN surveillance system will allow comparisons of rates between practices in the US and across LDOs and different countries.

References

1. Mehrotra R, Devuyst O, Davies SJ, Johnson DW. J Am Soc Nephrol. 2016;27(11):3238-52.
2. Cho Y, Johnson DW. Am J Kidney Dis. 2014;64(2):278-89.
3. Johnson DW, Dent H, Hawley CM, McDonald SP, Rosman JB, Brown FG, et al. Clin J Am Soc Nephrol. 2009;4(10):1620-8.
4. Davenport A. Perit Dial Int. 2009;29(3):297-302.

Acknowledgments

We extend our sincere appreciation to the teammates in more than 2,000 DaVita clinics who work every day to take care of patients and also to ensure the extensive data collection on which our work is based. The authors wish to thank Abigail E. Hunt, PhD, of DaVita Clinical Research for assistance in preparing this poster.

Correspondence: martin.schreiber@davita.com

Poster available at www.davitaclinicalresearch.com

American Society of Nephrology Kidney Week, October 23 - 28, 2018; San Diego, CA

© 2018 DaVita Inc. All rights reserved. Proprietary. May not be copied, reprinted, or distributed without the permission of DaVita Inc.