Analysis of After-Hours Oral Antibiotic Protocol for the Treatment of Suspected Peritonitis in Peritoneal Dialysis Patients

Martin J. Schreiber,[1] David A. Pegues,[2] Bram van Hout,[1] Julie A. Rodriguez,[1] Michelle Cassin,[1] Aaron Huwe,[1] Levi Njord,[1] George R. Aronoff[1]

[1] DaVita Inc, Denver, CO, USA; [2] Hospital of the University of Pennsylvania, Philadelphia, PA, USA

Introduction

- Delays in initiating antibacterial therapy for the treatment of peritonitis may lead to increased risk of peritoneal dialysis (PD) failure.[1]
- Because intraperitoneal (IP) antibiotic treatment is not available after business hours at most dialysis centers, it is important to have a protocol in place for oral "bridge" therapy until a patient can receive IP treatment.
- Historically, ciprofloxacin has been the medication provided for oral administration in the case of suspected peritonitis. However, fluoroquinolone antibiotics have been associated with disabling and potentially permanent serious side effects when used systemically.[2,3]
 - In 2016 the FDA issued a safety communication recommending that fluoroquinolone antibiotic use should be restricted in the case of uncomplicated infections and reserved only for patients who have no other treatment options.[4]

Objective

The current analysis was conducted to evaluate the probability of activity of selected oral antibiotic agents against the spectrum of PD infections observed at a large dialysis organization (LDO).

Methods

- Causative organisms and oral antibiotic susceptibility were analyzed for all peritonitis cases identified among PD patients treated by an LDO during 2016.
- Based on these findings, we determined the probability of activity of selected oral antibiotic agents (cefazolin, cefdinir, ciprofloxacin, trimethoprim/sulfamethoxazole [TMP/SMX]) against PD infections.
 - Probability of activity of each agent was assessed as the proportion of infections that would be expected to be susceptible and was considered for PD infections overall and separately for both gram-positive and gram-negative infections.

Results

Figure 1: Causal Organisms for Peritonitis Cases Identified During 2016

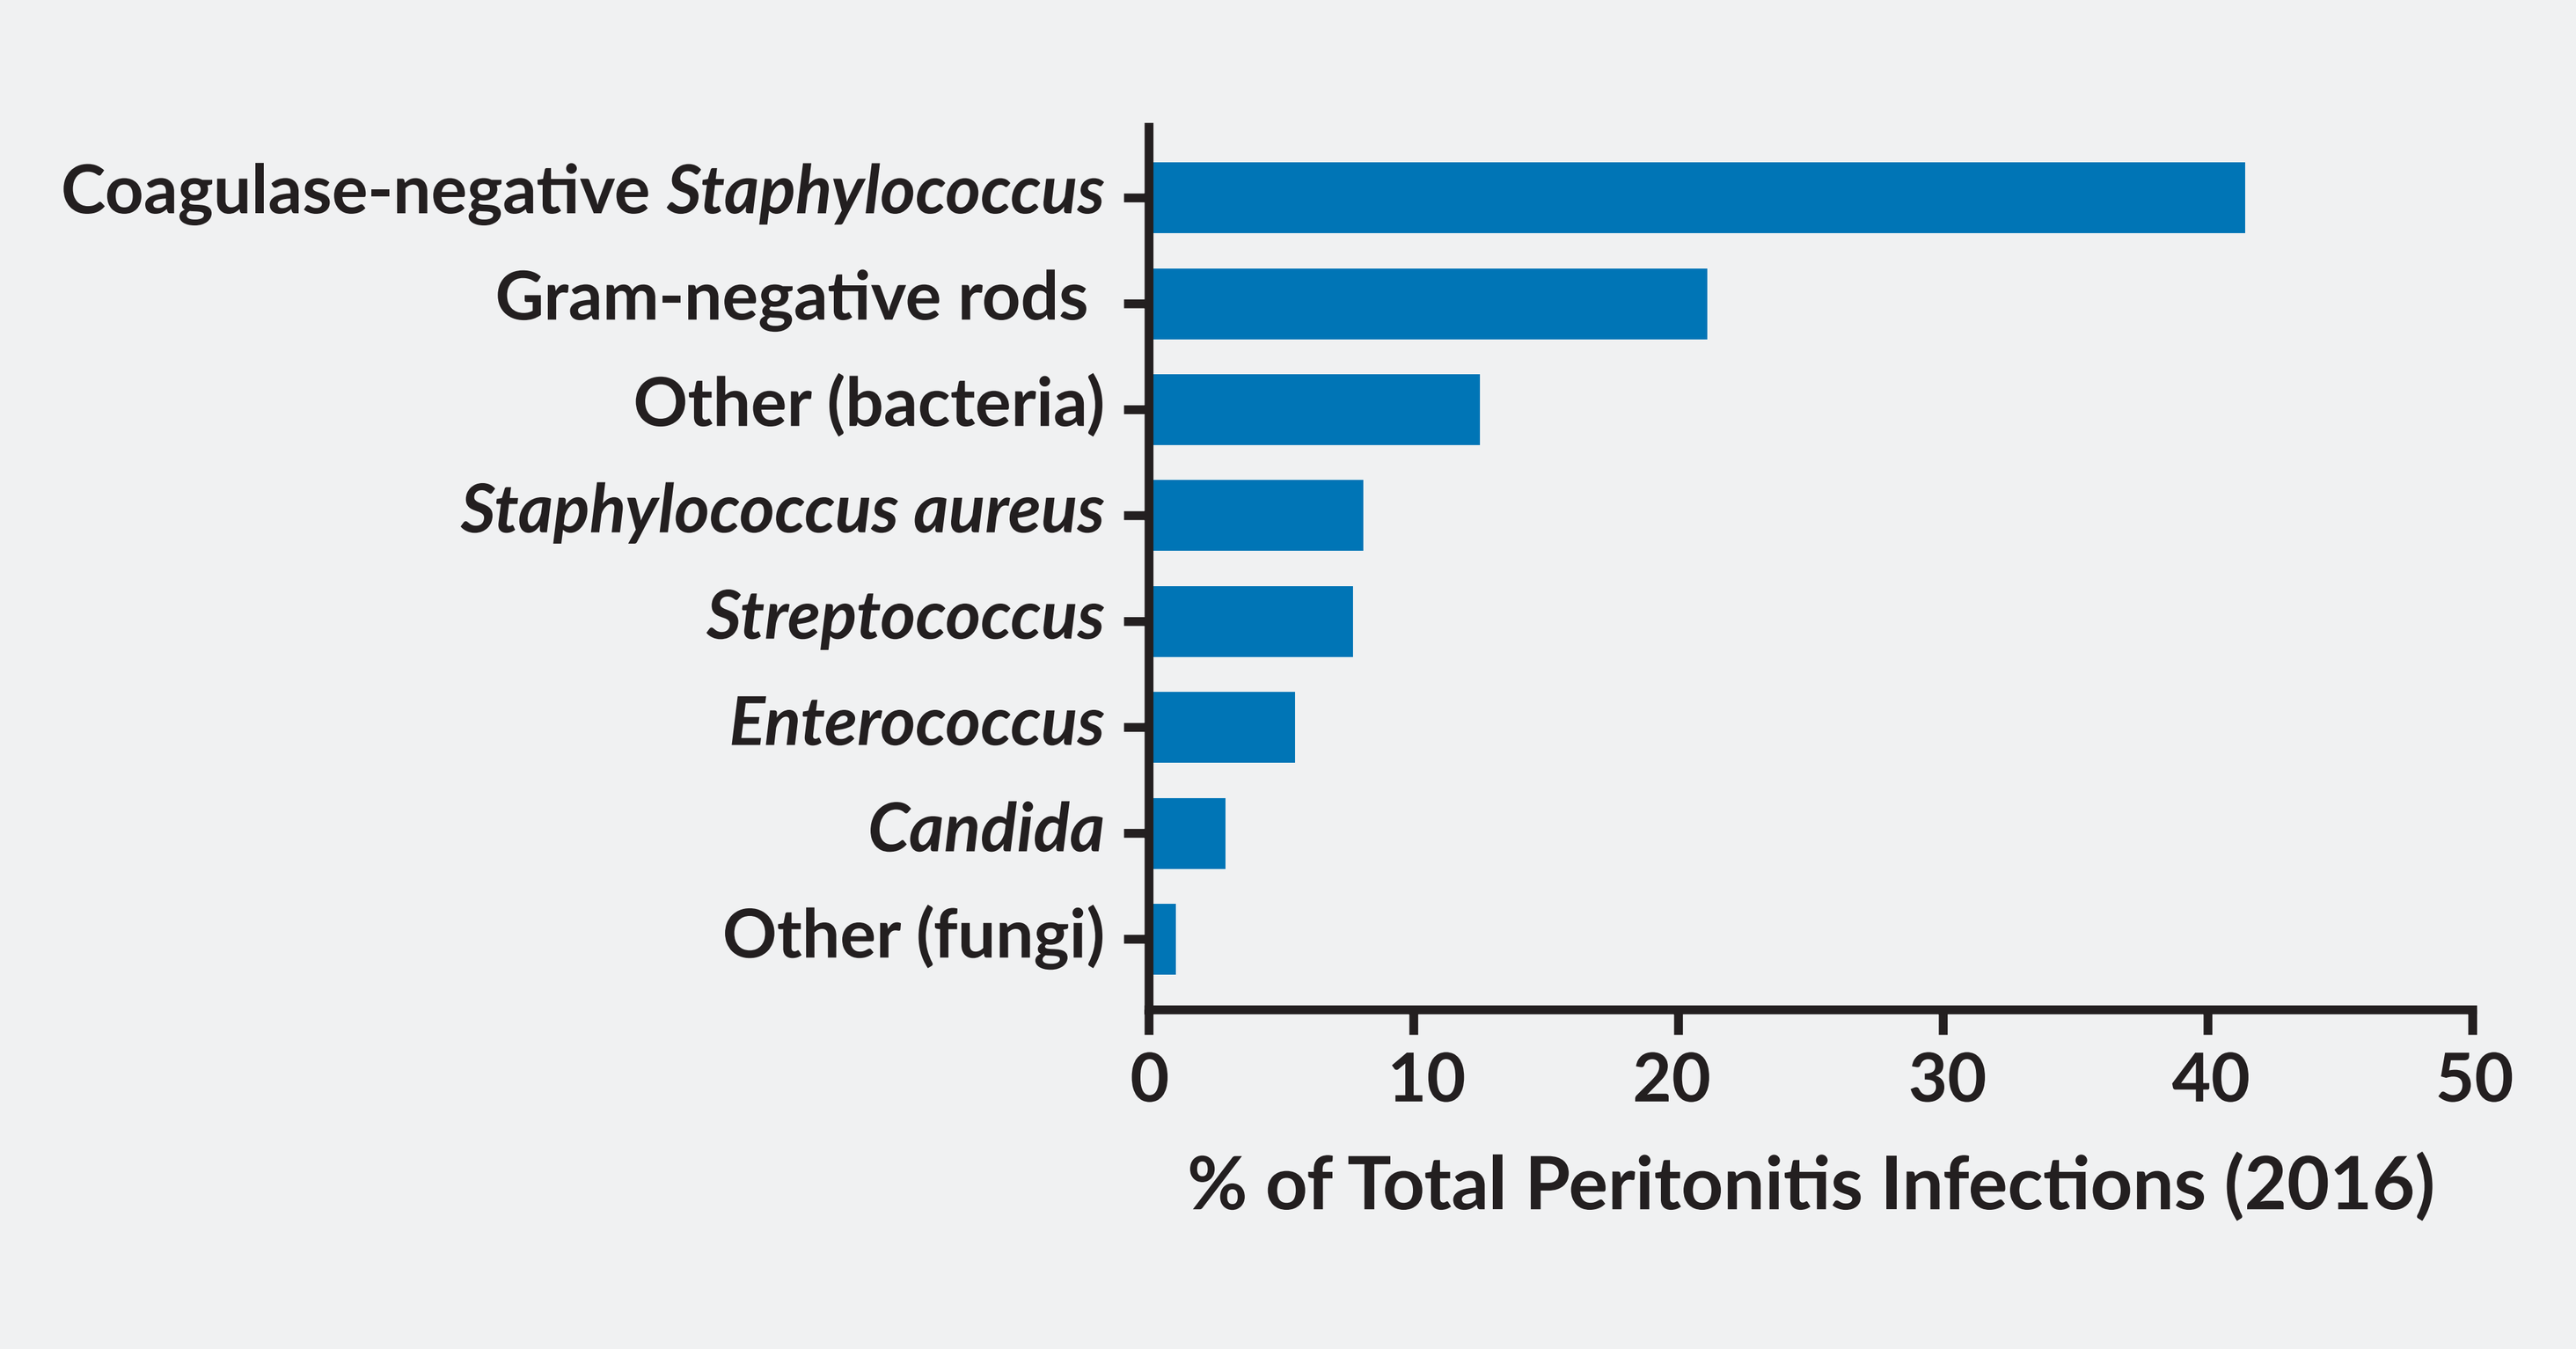
Coagulase-negative *Staphylococcus*
Gram-negative rods
Other (bacteria)
Staphylococcus aureus
Streptococcus
Enterococcus
Candida
Other (fungi)
0
10
20
30
40
50
% of Total Peritonitis Infections (2016)

Gram-negative rods category included: *Escherichia coli, Klebsiella pneumoniae, Psdeudomonas aeruginosa, Klebsiella oxytoca, Serratia marcescens, Stenotrophomonas maltophilia, Citrobacter freundii, Sphingomonas paucimmobilis, Pseudomonas putida, Bacteroides fragilis, Enterobacter aerogenes, Morganella mrganii, Neisseria elongata, Raoultella planticola*

Figure 2: Predicted Susceptibility of Peritonitis Infections to Oral Antibiotic Agents

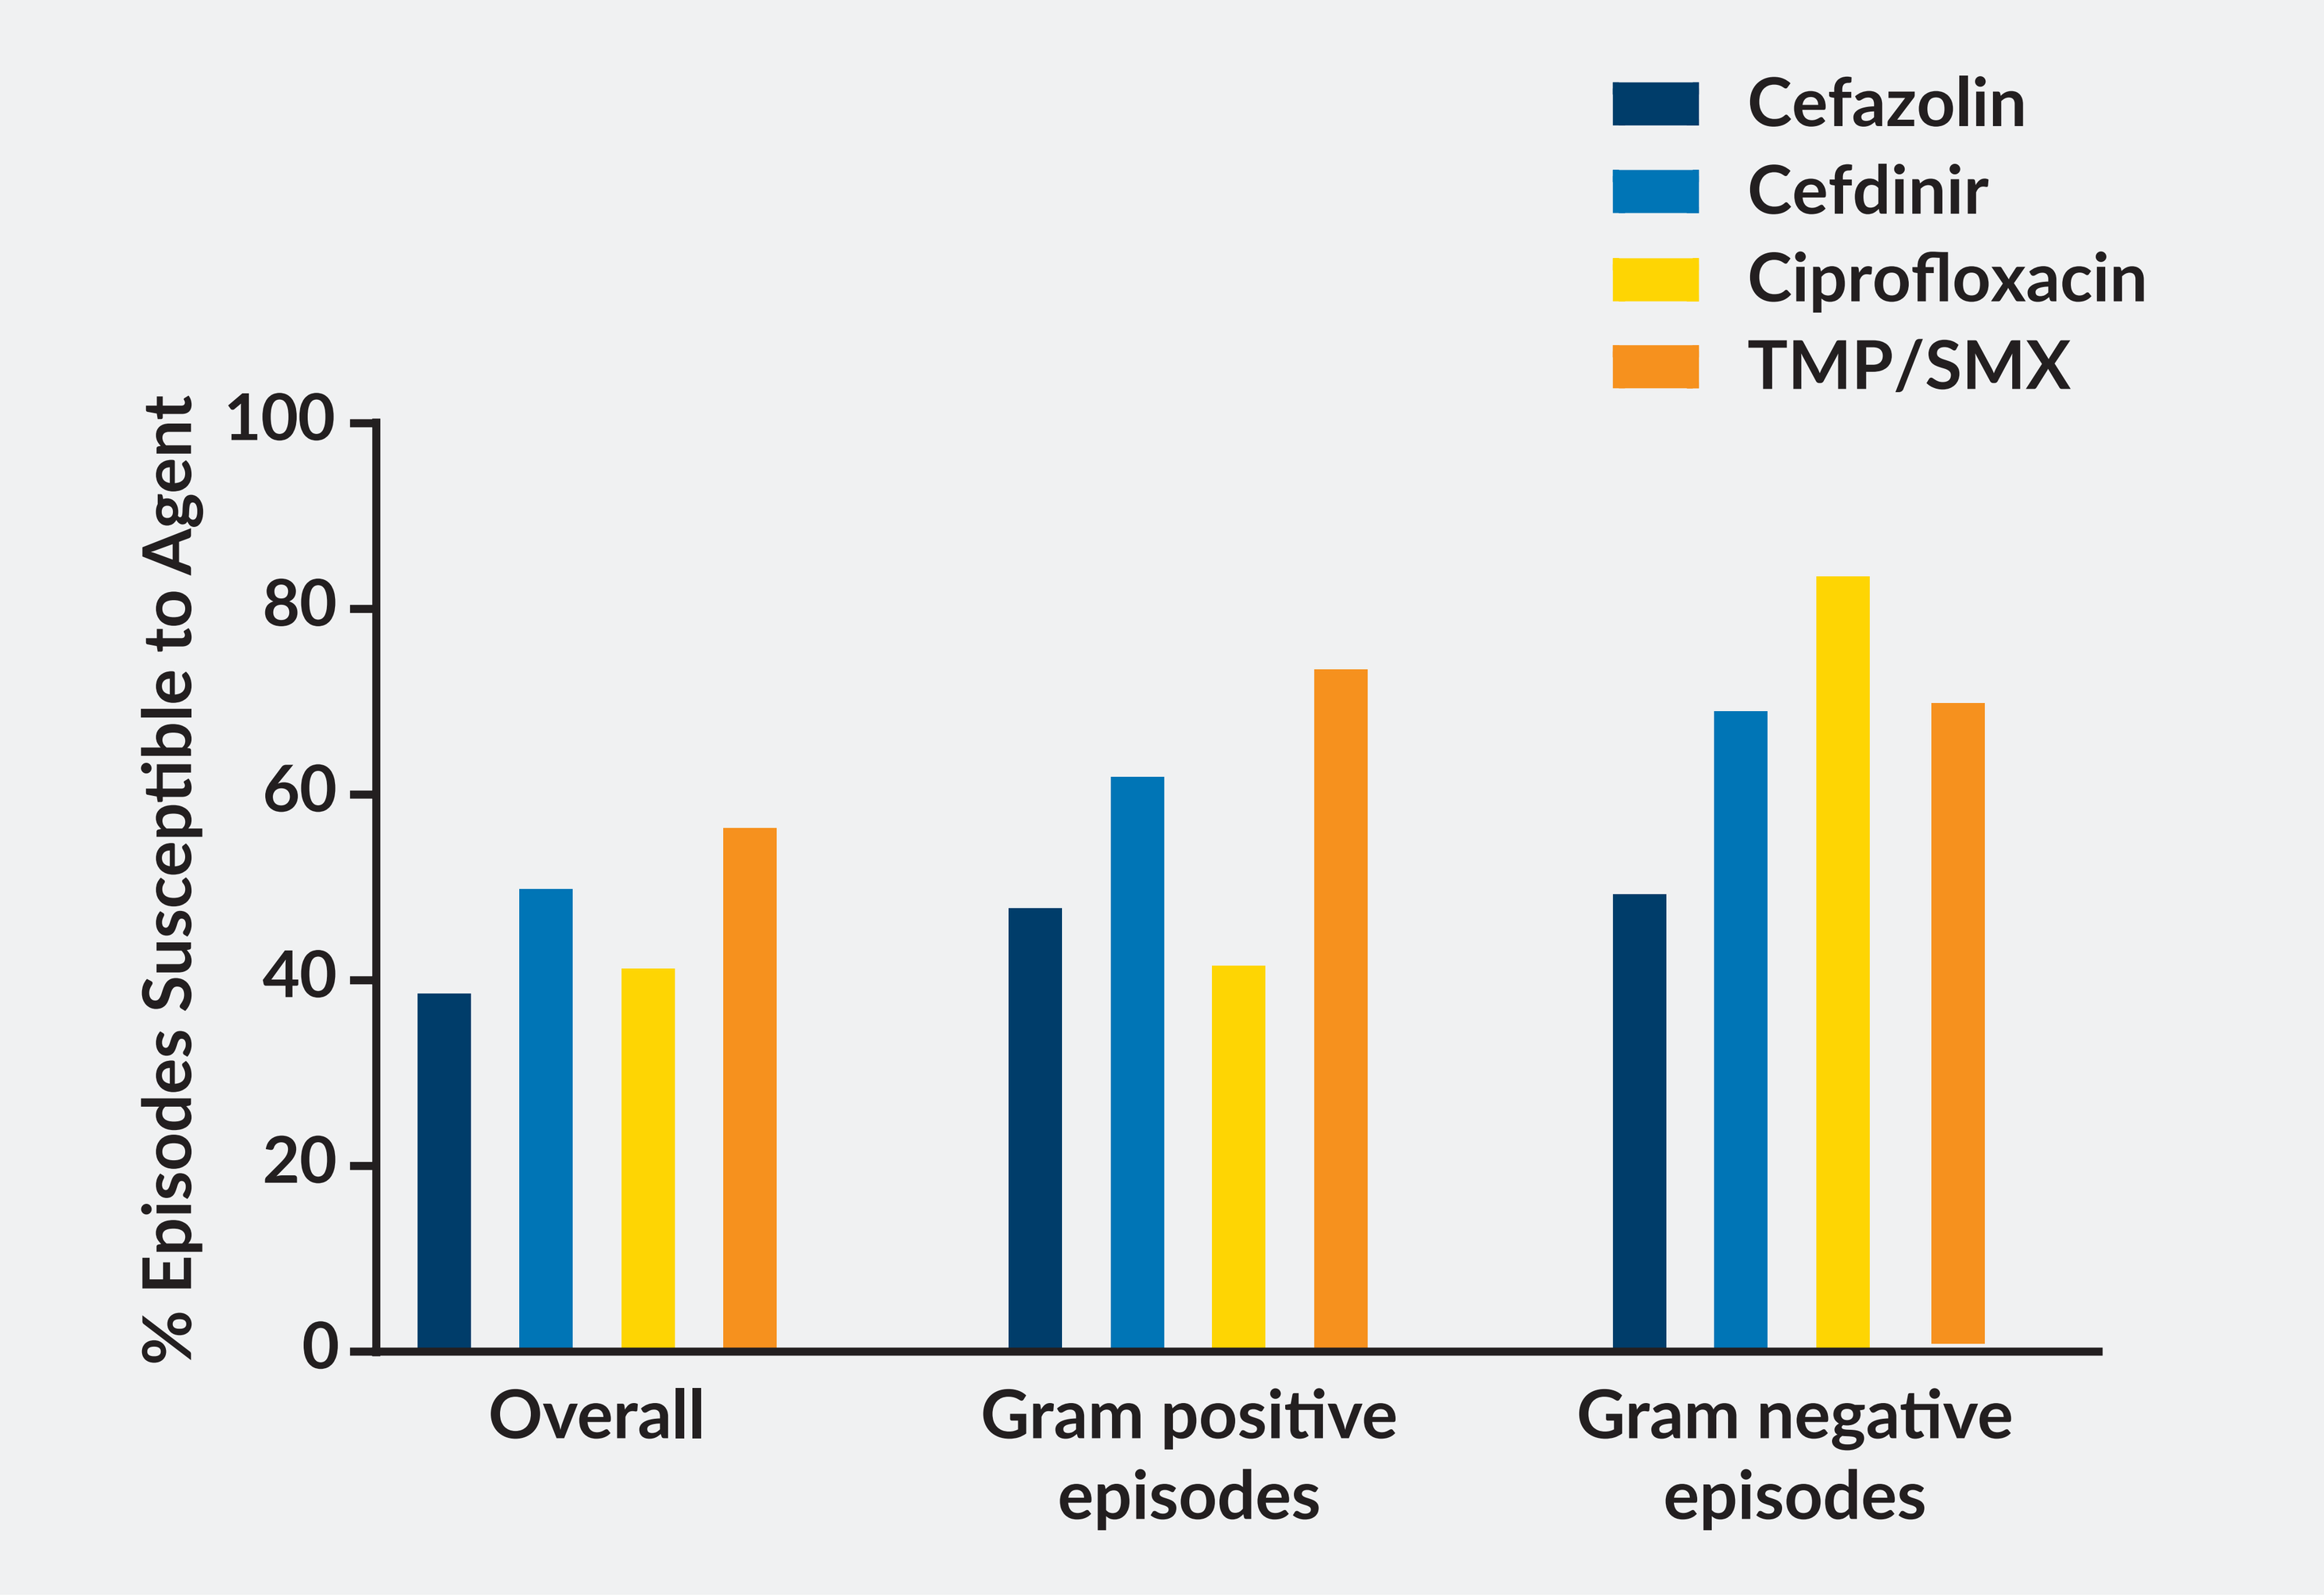
Cefazolin
Cefdinir
Ciprofloxacin
TMP/SMX
% Episodes Susceptible to Agent
0
20
40
60
80
100
Overall
Gram positive episodes
Gram negative episodes

Abbreviation: TMP/SMX, trimethoprim/sulfamethoxazole

- A total of 4534 cases of peritonitis were identified at the LDO during 2016.
- Causative organisms were identified for all peritonitis episodes (Figure 1): the most common were coagulase-negative *Staphylococcus*, accounting for 41.4% of infections, and gram-negative rods, accounting for 21.2% of infections.
- Assuming this distribution of pathogens, susceptibility of PD infections to common oral antibiotic agents was predicted. Predicted susceptibilities are shown in Figure 2, likelihood of ineffective therapy values are shown in Table 1.
 - For peritonitis infections overall, TMP/SMX was predicted to be the most active agent (57.2% of episodes susceptible), followed by cefdinir (50.6%), ciprofloxacin (42%), and cefazolin (39.4%).
 - Among gram-positive infections alone, 74.3% would be expected to be susceptible to TMP/SMX, and 62.7% to cefdinir.
 - Among gram-negative infections, ciprofloxacin was predicted to be the most active agent (84.3% of episodes susceptible).

Table 1: Likelihood of Ineffective Therapy

	Likelihood of Ineffective Therapy[a]			
	Cefazolin	Cefdinir	Ciprofloxacin	TMP/SMX
Overall	2.5	2.0	2.4	1.7
Gram positive infections	1.9	2.7	1.7	3.9
Gram negative infections	2.0	3.3	6.4	3.3

[a] A likelihood of ineffective therapy value of 2 means that 1 out of 2 patients with infection caused by the organism would be treated inadequately with the agent.

Green shading indicates most effective agents

Abbreviation: TMP/SMX, trimethoprim/sulfamethoxazole

Conclusions

- Early initiation of treatment for peritonitis is critical to avoid PD failure.
 - Designing a strategy for early therapy that can be administered at home, until a patient can receive IP treatment, can potentially improve outcomes.
- Based on the current analyses, TMP/SMX and cefdinir are predicted to be the most active oral agents for the treatment of suspected gram-positive infections.
- Ciprofloxacin should be used only in the case of suspected high-risk, gram-negative infections.

References

1. Muthucumarana K, Howson P, Crawford D, Burrows S, Swaminathan R, Irish A. The Relationship Between Presentation and the Time of Initial Administration of Antibiotics With Outcomes of Peritonitis in Peritoneal Dialysis Patients: The PROMPT Study. Kidney Int Rep. 2016;1(2):65-72.
2. Mehlhorn AJ, Brown DA. Safety concerns with fluoroquinolones. Ann Pharmacother. 2007;41(11):1859-66.
3. De Sarro A, De Sarro G. Adverse reactions to fluoroquinolones. an overview on mechanistic aspects. Curr Med Chem. 2001;8(4):371-84.
4. United States Food and Drug Administration Safety Communication. July 26, 2016. https://www.fda.gov/Drugs/DrugSafety/ucm500143.htm

Acknowledgments

We extend our sincere appreciation to the teammates in more than 2,000 DaVita clinics who work every day to take care of patients and also to ensure the extensive data collection on which our work is based. We thank DaVita Clinical Research® (DCR®), and specifically acknowledge Abigail E. Hunt, PhD, of DCR for editorial contributions in preparing this poster.

Correspondence: martin.schreiber@davita.com

Poster available at www.davitaclinicalresearch.com

American Society of Nephrology Kidney Week, October 23 - 28, 2018; San Diego, CA

© 2018 DaVita Inc. All rights reserved. Proprietary. May not be copied, reprinted, or distributed without the permission of DaVita Inc.