Correspondence between Causes of Recurrent Hospitalizations among Hemodialysis Patients

DaVita Clinical Research

Scott Sibbel, PhD, MPH; Adam G. Walker, PhD; Francesca Tentori, MD, MS; Steven M. Brunelli, MD, MSCE

DaVita Clinical Research, Minneapolis, MN, USA

Introduction

- Despite targeted efforts, hospitalizations represent a major burden for end-stage kidney disease (ESKD) patients.
- On average, ESKD patients experience 1.7 hospital admissions per year and 30.7% will be readmitted within 30 days of discharge.[1]
- In order to create programs to reduce hospitalizations, there is a need to identify factors that drive hospitalization risk within this population.

Objective

To evaluate the relationship between cause of initial hospitalization and cause of readmission among ESKD patients receiving in-center hemodialysis (ICHD)

Methods

Data Sources and Study Patients

- Data for this study were derived from the electronic health records of a large dialysis organization (LDO) and Medicare claims contained in the United States Renal Data System (USRDS).
- Patients eligible for inclusion in this study were those who during 2014:
 - Were Medicare Part A beneficiaries
 - Were receiving three times weekly ICHD at LDO facilities
 - Had a qualifying index hospitalization ≥ 1 day in length

Figure 1. Study Design

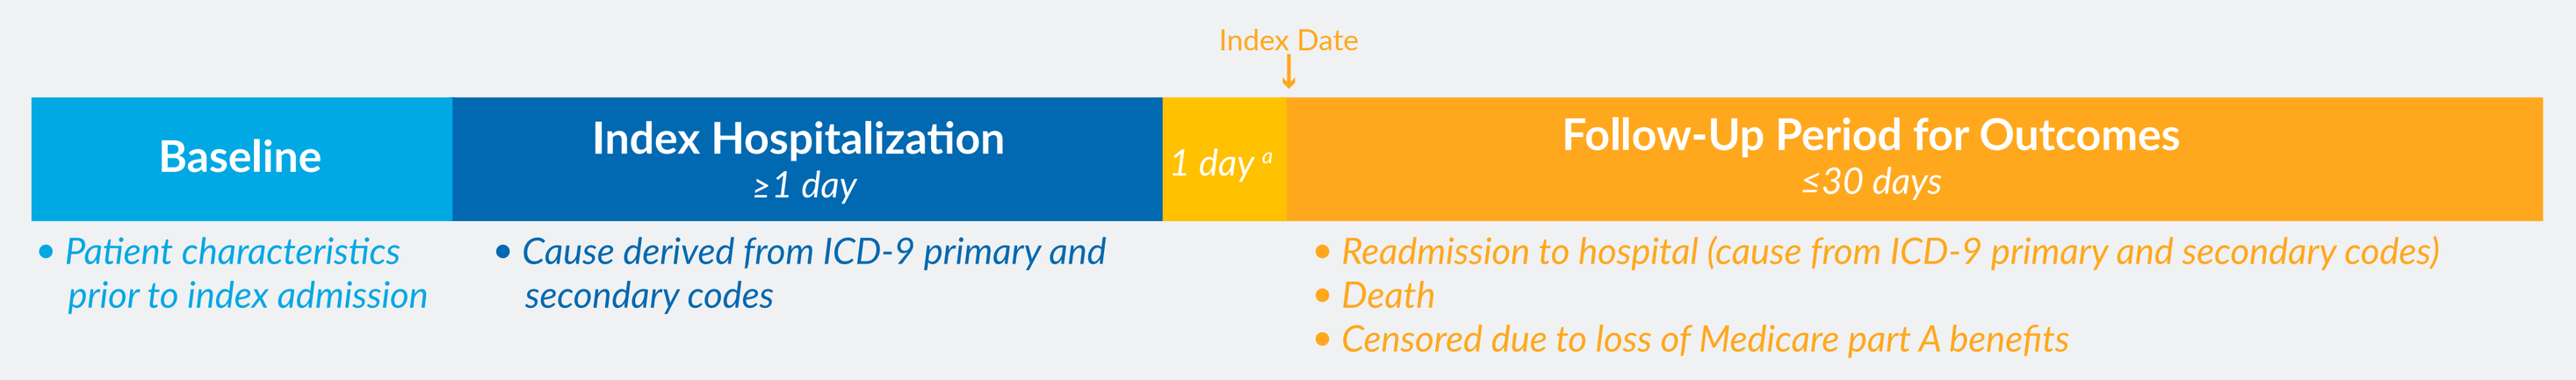

Index Date
Baseline
Index Hospitalization
≥1 day
1 day [a]
Follow-Up Period for Outcomes
≤30 days
• Patient characteristics prior to index admission
• Cause derived from ICD-9 primary and secondary codes
• Readmission to hospital (cause from ICD-9 primary and secondary codes)
• Death
• Censored due to loss of Medicare part A benefits

[a] Hospital admissions that occurred on the same day as index hospitalization discharge were considered an extension of the index hospitalization and not a readmission.

Outcomes

- The study index date was set 1 calendar day after discharge for index hospitalization and patients were followed forward in time for up to 30 days for the following outcomes:
 - Readmission to the hospital
 - Death
 - Censoring due to loss of Medicare part A benefits
- Causes of index hospitalizations and readmissions were derived from primary and secondary ICD-9 codes and were categorized using the Agency for Healthcare Research and Quality (AHRQ) level 1 clinical classification system (CCS).
- The relationship between causes of index hospitalization and readmission were determined through cross-tabulation of the percent difference between observed (O) and expected (E) readmissions accounting for background admission rate (O-E/E*100).

Results

Table 1. Baseline Patient Characteristics

		Not Readmitted/Died N = 69,007	Readmitted/Died N = 65,918	Overall N = 134,925
Age, years	mean ± SD	62.7 ± 14.5	61.6 ± 15.5	62.4 ± 14.9
Female, n (%)		42,361 (48.4)	23,557 (49.7)	65,918 (48.9)
Vintage, months	mean ± SD	53.4 ± 48.8	51.1 ± 47.7	52.6 ± 48.4
Body mass index, kg/m²	mean ± SD	27.8 ± 7.6	26.9 ± 7.4	27.5 ± 7.5
Race, n (%) index admissions				
White		35,147 (40.1)	19,721 (41.6)	54,868 (40.7)
Black		33,911 (38.7)	18,677 (39.4)	52,588 (39.0)
Hispanic		12,672 (14.5)	63,26 (13.4)	18,998 (14.1)
Other/unknown		5830 (6.7)	2641 (5.6)	8471 (6.3)
Etiology, n (%) index admissions				
Diabetes		42,216 (48.2)	22,721 (48.0)	64,937 (48.1)
Hypertension		25,320 (28.9)	13,408 (28.3)	38,728 (28.7)
Other		20,024 (22.9)	11,236 (23.7)	31,260 (23.2)
Vascular access type, n (%) index admissions				
Arteriovenous fistula		50,457 (57.6)	24,656 (52.1)	75,113 (55.7)
Arteriovenous graft		18,543 (21.2)	10,276 (21.7)	28,819 (21.4)
Central venous catheter		18,558 (21.2)	12,433 (26.3)	30,991 (23.0)
Charlson Comorbidity Index	mean ± SD	6.0 ± 2.0	6.0 ± 2.1	6.0 ± 2.1
Cerebrovascular disease, n (%) index admissions		2342 (2.7)	1346 (2.8)	3688 (2.7)
Congestive heart failure, n (%) index admissions		19,155 (21.9)	11,368 (24.0)	30,523 (22.6)
Secondary insurance, n (%) index admissions				
Medicare		274 (0.3)	116 (0.2)	390 (0.3)
Medicaid		41,247 (47.1)	24,233 (51.2)	65,480 (48.5)
Commercial		34,088 (38.9)	16,613 (35.1)	50,701 (37.6)
Government		887 (1.0)	423 (0.9)	1310 (1.0)
No insurance		797 (0.9)	361 (0.8)	1158 (0.9)
Unknown/missing		10,267 (11.7)	5619 (11.9)	15,886 (11.8)
Index discharge day, n (%) index admissions				
Sunday		7464 (8.5)	3659 (7.7)	11,123 (8.2)
Monday		10,924 (12.5)	5862 (12.4)	16,786 (12.4)
Tuesday		13,663 (15.6)	7665 (16.2)	21,328 (15.8)
Wednesday		14,137 (16.2)	7749 (16.4)	21,886 (16.2)
Thursday		14,652 (16.7)	7817 (16.5)	22,469 (16.7)
Friday		15,672 (17.9)	8701 (18.4)	24,373 (18.1)
Saturday		11,048 (12.6)	5912 (12.5)	16,960 (12.6)

Based on 61,987 unique ICHD patients eligible during the study period. Data are presented at the level of the index hospital admission.
Abbreviations: SD, standard deviation

Patient Characteristics and Hospitalization Patterns

- Baseline characteristics and demographics among 61,987 patients eligible for inclusion in this analysis are presented in Table 1.
 - There were no appreciable differences between patients that were not readmitted and those that were readmitted.
- Figure 2 illustrates patterns of index admissions and readmissions among ICHD patients at the LDO during the study period.
 - There were 134,925 qualifying index admission resulting in an overall hospitalization rate of 1.67 admissions per patient-year.
 - Readmission within 30 days post discharge occurred in 31.5% of patients.
 - The composite outcome of readmission or death occurred within 35.1% of patients within 30 days of discharge.

Figure 2. Index Admissions and Readmission among ICHD Patients

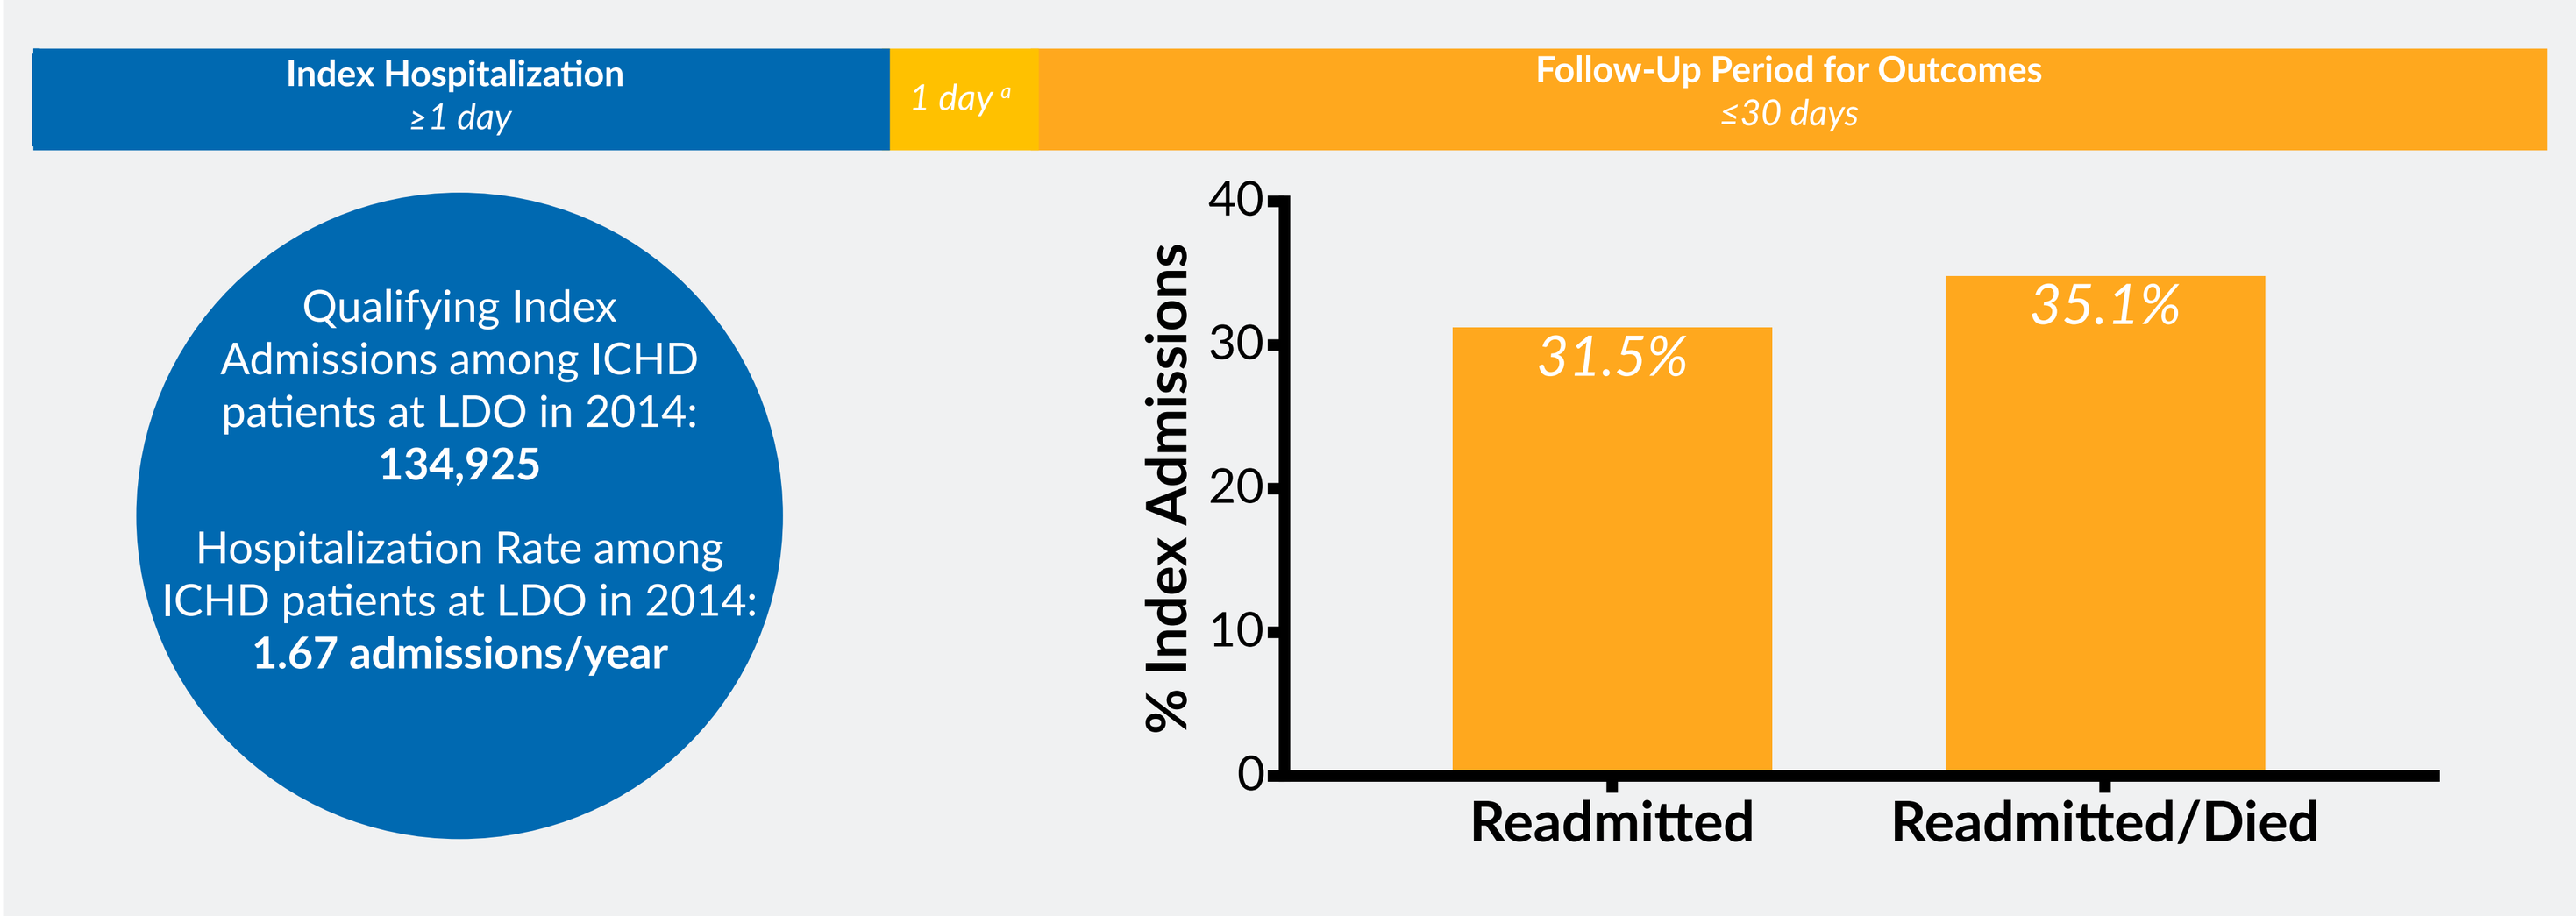

Index Hospitalization
≥1 day
1 day [a]
Follow-Up Period for Outcomes
≤30 days
Qualifying Index Admissions among ICHD patients at LDO in 2014:
134,925
Hospitalization Rate among ICHD patients at LDO in 2014:
1.67 admissions/year
% Index Admissions
40
30
20
10
0
31.5%
35.1%
Readmitted
Readmitted/Died

[a] Hospital admissions that occurred on the same day as index hospitalization discharge were considered an extension of the index hospitalization and not a readmission.
[b] National hospitalization rate among hemodialysis patients in 2014 was 1.74 admissions per year [1]

Figure 3. Causes of Index Admissions and Readmission among ICHD Patients

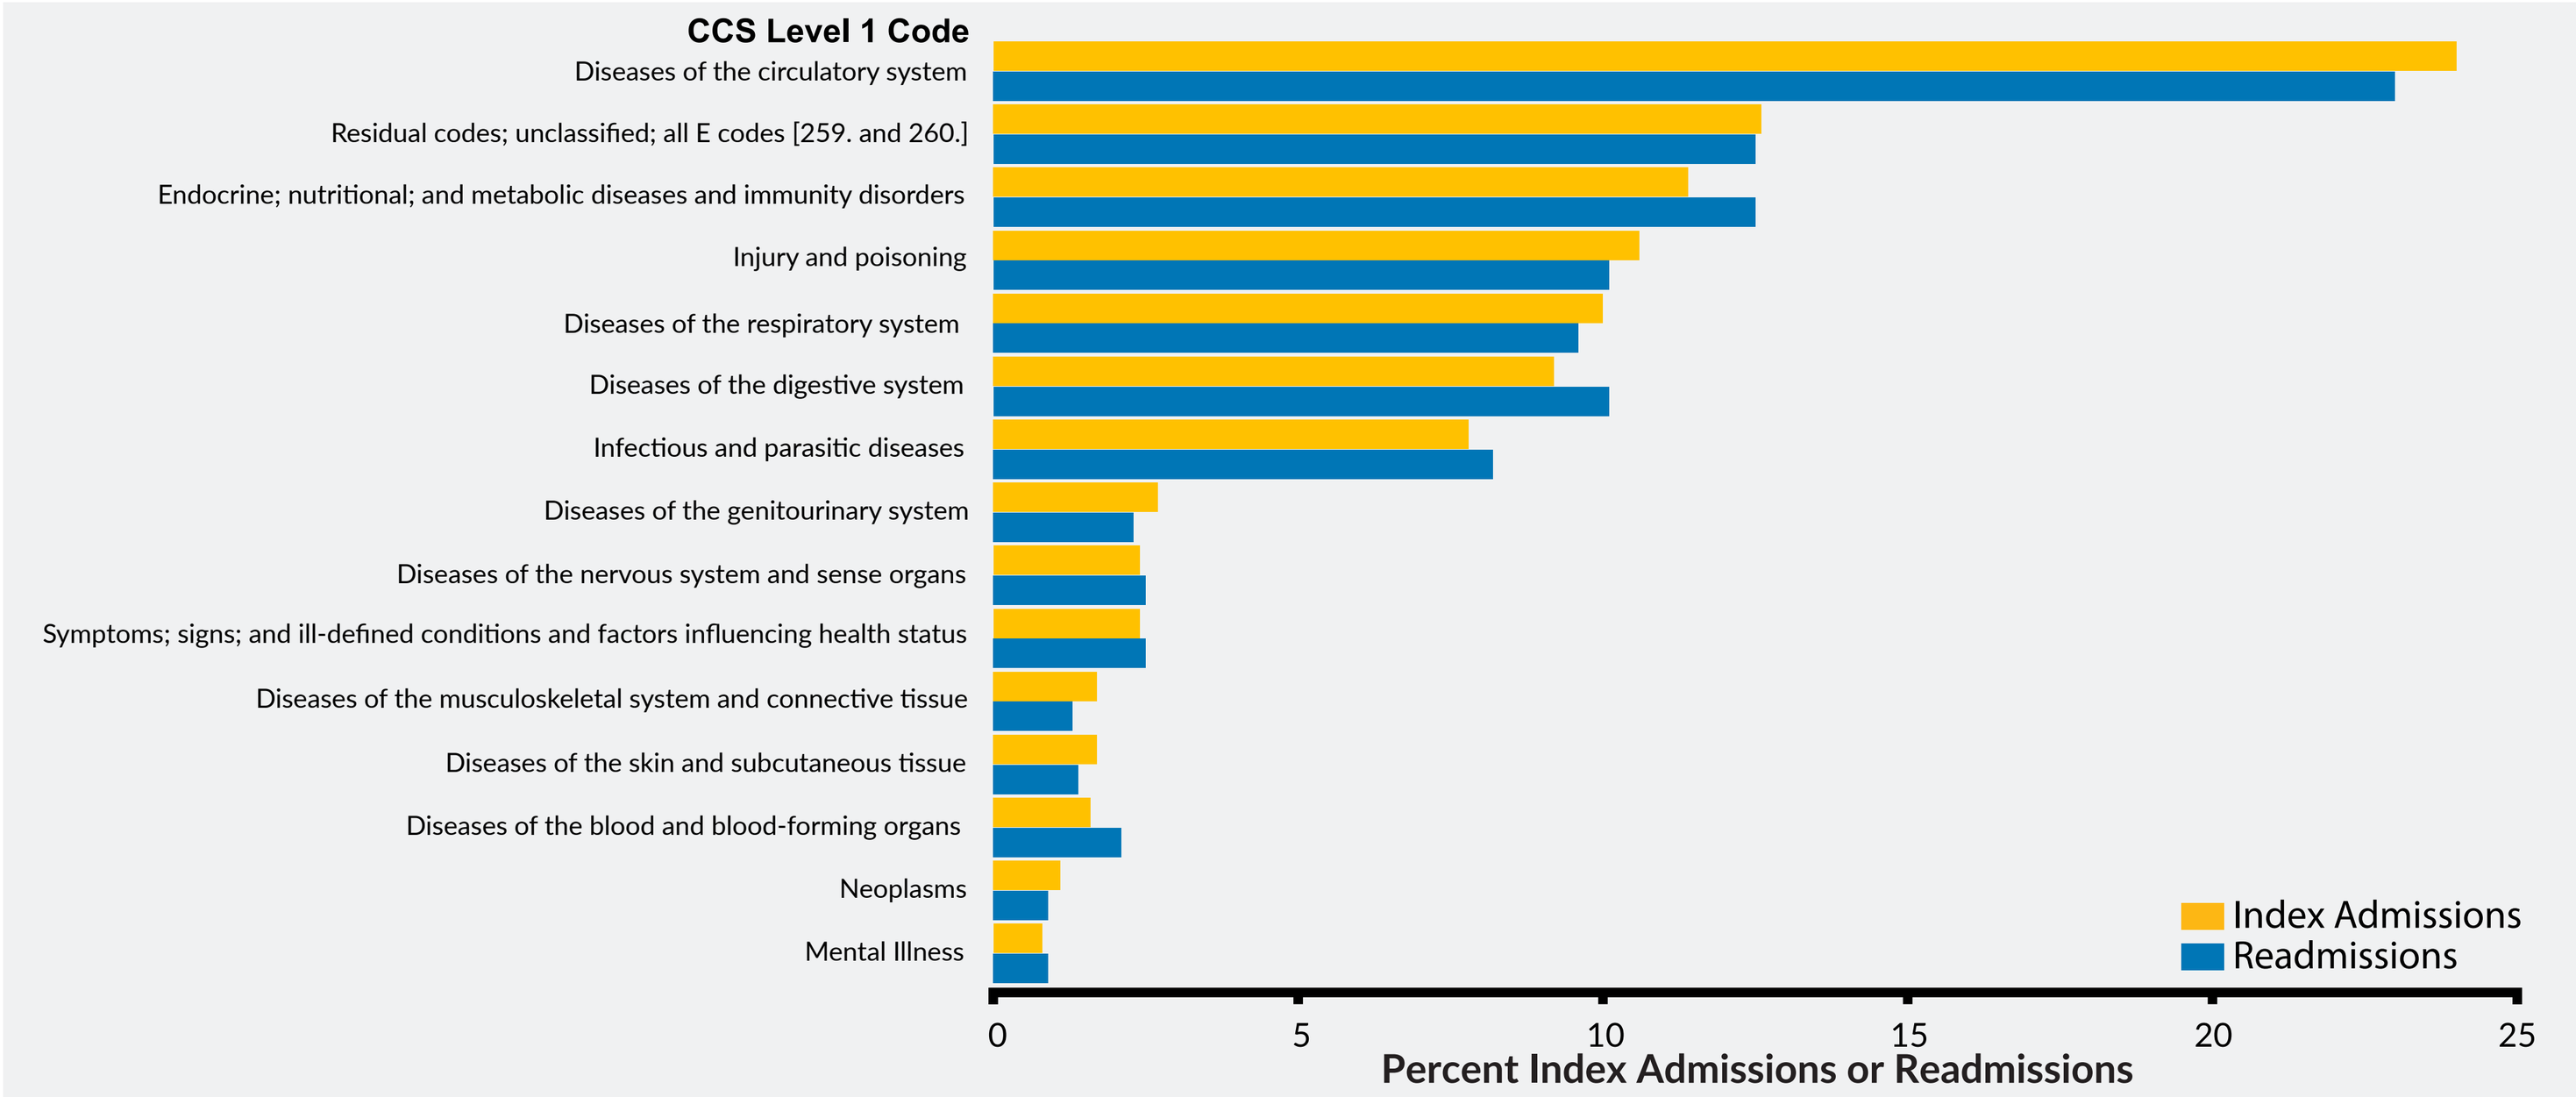

CCS Level 1 Code
Diseases of the circulatory system
Residual codes; unclassified; all E codes [259. and 260.]
Endocrine; nutritional; and metabolic diseases and immunity disorders
Injury and poisoning
Diseases of the respiratory system
Diseases of the digestive system
Infectious and parasitic diseases
Diseases of the genitourinary system
Diseases of the nervous system and sense organs
Symptoms; signs; and ill-defined conditions and factors influencing health status
Diseases of the musculoskeletal system and connective tissue
Diseases of the skin and subcutaneous tissue
Diseases of the blood and blood-forming organs
Neoplasms
Mental Illness
Index Admissions
Readmissions
0
5
10
15
20
25
Percent Index Admissions or Readmissions

Data not shown for: Congential anomalies; Complications of pregnancy, childbirth, and the puerperium; and certain conditions originating in the perinatal period. Together, these categories represented 0.13% and 0.06% of index admissions and readmissions, respectively.

Causes of Index Admissions and Readmissions

- CCS level 1 causes of index admissions and readmissions are shown in Figure 3.
 - The most common causes for index admissions and readmissions were related to diseases of the circulatory system.
- The heat map in Figure 4 illustrates the relationship between CCS level 1 causes of index admissions and causes of readmissions
 - After accounting for background rate of index admissions, the most common cause for the majority of readmissions fell into the same CCS category as the preceding index admission.

Figure 4. Relationship between Causes of Index Hospitalization and Readmission

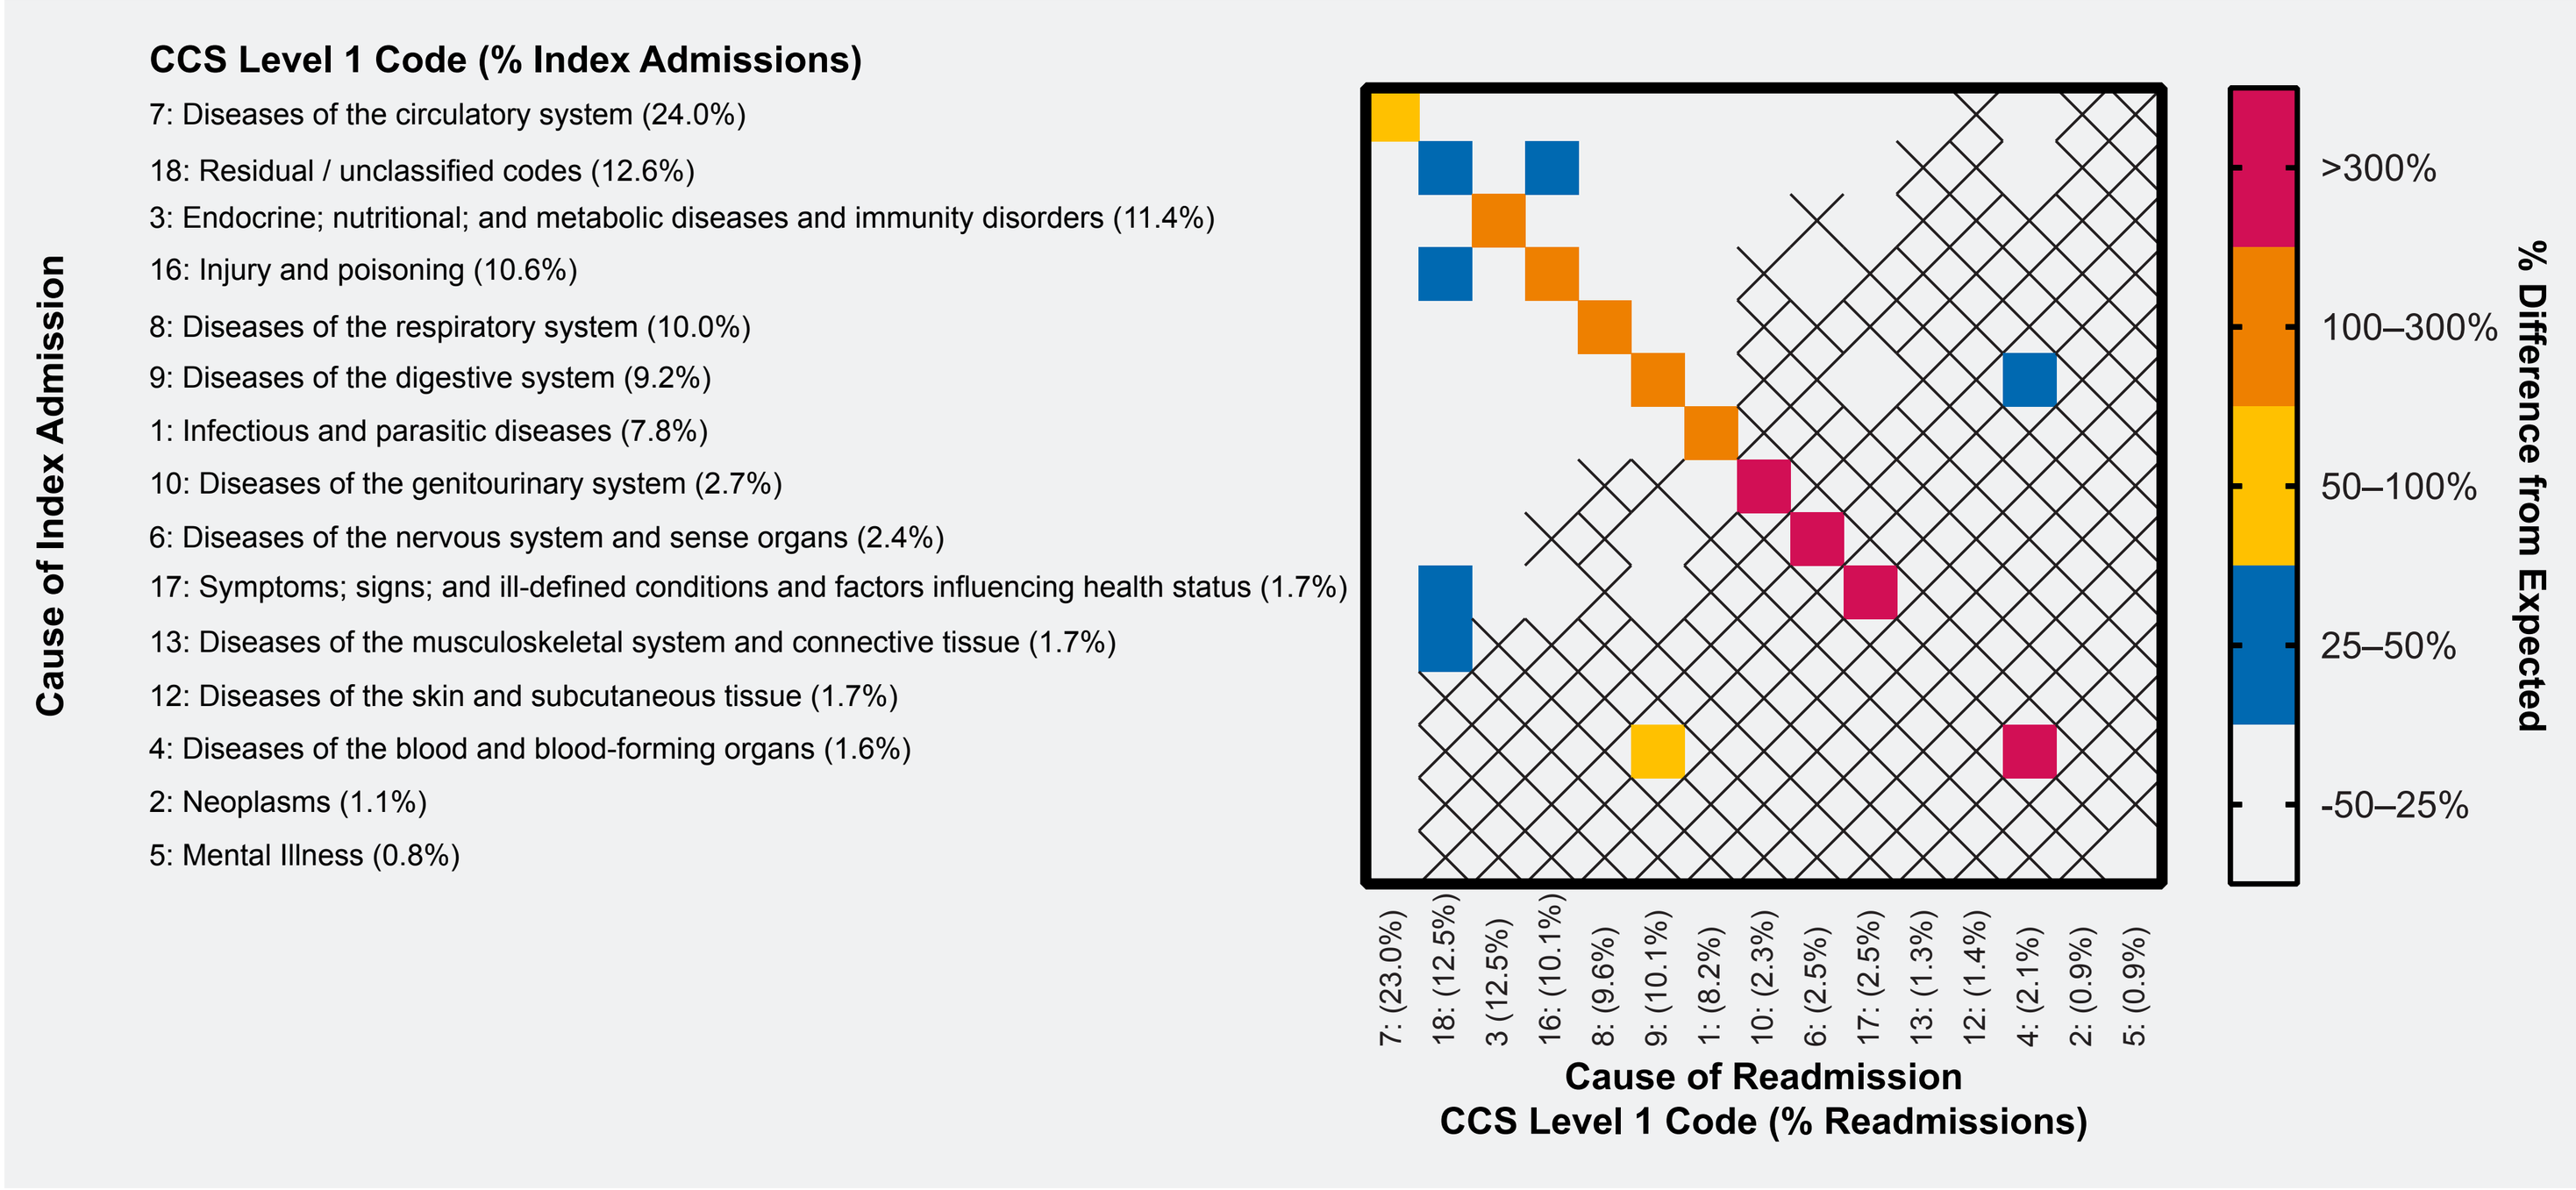

CCS Level 1 Code (% Index Admissions)
7: Diseases of the circulatory system (24.0%)
18: Residual / unclassified codes (12.6%)
3: Endocrine; nutritional; and metabolic diseases and immunity disorders (11.4%)
16: Injury and poisoning (10.6%)
8: Diseases of the respiratory system (10.0%)
9: Diseases of the digestive system (9.2%)
1: Infectious and parasitic diseases (7.8%)
10: Diseases of the genitourinary system (2.7%)
6: Diseases of the nervous system and sense organs (2.4%)
17: Symptoms; signs; and ill-defined conditions and factors influencing health status (1.7%)
13: Diseases of the musculoskeletal system and connective tissue (1.7%)
12: Diseases of the skin and subcutaneous tissue (1.7%)
4: Diseases of the blood and blood-forming organs (1.6%)
2: Neoplasms (1.1%)
5: Mental Illness (0.8%)
Cause of Index Admission
7: (23.0%)
18: (12.5%)
3: (12.5%)
16: (10.1%)
8: (9.6%)
9: (10.1%)
1: (8.2%)
10: (2.3%)
6: (2.5%)
17: (2.5%)
13: (1.3%)
12: (1.4%)
4: (2.1%)
2: (0.9%)
5: (0.9%)
Cause of Readmission
CCS Level 1 Code (% Readmissions)
>300%
100–300%
50–100%
25–50%
-50–25%
% Difference from Expected

Cells with less than 100 cases were excluded and are marked by an X.

Conclusions

- We found that the cause of readmission was closely related to the cause of prior hospitalization in a large sample of US dialysis patients.
- Despite potential misclassification of readmission cause, these results raise the possibility that the clinical issue was not fully addressed during the index admission.
- Further studies are needed to identify clinical practices that may help reduce readmission rates.

References

1. United States Renal Data System. 2016 USRDS annual data report: Epidemiology of kidney disease in the United States. National Institutes of Health, National Institute of Diabetes and Digestive and Kidney Diseases, Bethesda, MD, 2016.

Acknowledgments

We extend our sincere appreciation to the teammates in more than 2,000 DaVita clinics who work every day to take care of patients and also to ensure the extensive data collection on which our work is based. Some of the data used in the analyses described here were supplied by the United States Renal Data System (USRDS). The interpretation and reporting of these data are the responsibility of the authors and in no way should be seen as an official policy or interpretation of the US government.

Correspondence: scott.sibbel@davita.com

Poster available at www.davitaclinicalresearch.com

American Society of Nephrology Kidney Week, October 23 - 28, 2018; San Diego, CA

© 2018 DaVita Inc. All rights reserved. Proprietary. May not be copied, reprinted, or distributed without the permission of DaVita Inc.