Midodrine in the Context of Intradialytic Hypotension: Association with Outcomes

Steven M. Brunelli, MD, MSCE[1,2]; Dena E. Cohen, PhD[1,2]; Gilbert Marlowe[1,2]; David Van Wyck, MD[2]

[1]DaVita Clinical Research, Minneapolis, MN, USA; [2]DaVita Institute for Patient Safety, Denver, CO, USA

Introduction

- Intradialytic hypotension (IDH) is a frequent complication of hemodialysis, and is associated with significant morbidity and mortality.[1]
- Off-label use of the alpha-1 adrenergic receptor agonist midodrine to reduce the frequency and severity of IDH is common.
- Small-scale clinical trial data support this practice;[2] however, limited data exist with regard to real-world efficacy.

Objective

To evaluate the real-world effectiveness of midodrine for management of IDH by determining whether use of midodrine is associated with better clinical and hemodynamic outcomes among patients treated with in-center hemodialysis (ICHD)

Methods

Patients

- This retrospective, observational study used data derived from deidentified patient electronic health records.
- Included patients were adults with dialysis vintage ≥90 days receiving thrice-weekly ICHD between 01 July 2015 and 30 September 2016. Veteran's Affairs beneficiaries, and patients for whom data were not available for 30 or more days following dialysis initiation were excluded.

Exposure, Matching, and Cohort Construction

- Exposure status was assigned based on an order for midodrine in the electronic health record.
 - For exposed patients, the date of the first midodrine order during the study period was defined as the index date.
- For each midodrine patient, eligible controls were those who, as of the start of the corresponding month, had similar values for dialysis vintage (within ± 6 months), mean monthly pre-dialysis systolic blood pressure (SBP; within ± 5 mmHg), mean monthly nadir SBP (within ± 5 mmHg) and percentage of treatments impacted by IDH (within ± 5% treatments), defined as nadir SBP < 90mmHg.[1]
 - Each midodrine patient was matched to up to 2 eligible controls.
- Although midrodrine patients and controls were well-matched at the start of the index month, differences emerged by the index date due to changes in clinical status that occurred in the time between the start of the month and the index date. To minimize this effect, the analytic cohort was limited to matched groups whose index date fell within the first 10 days of the month.

Outcomes and Statistical Analysis

- Patients were followed forward in time from index until the earliest of study end (30 September 2016) or censoring for death, transplant, or loss to follow-up.
- All analyses followed intention-to-treat principles.
- Death, all-cause hospitalization, and cardiovascular hospitalization were expressed as rates (events per patient-year) and compared using Poisson models.
- Percent of treatments affected by IDH and other hemodynamic outcomes were expressed as mean values during each month of follow-up and compared using linear mixed models.
- All models were adjusted for baseline values of age, sex, race, etiology of end-stage renal disease (ESRD), pre-dialysis SBP, nadir SBP, percent of treatments affected by IDH, UF volume, and target weight; time-updated values for dialysis vintage, vascular access type, albumin, creatinine, hemoglobin, and Kt/V; and time-updated presence of a diagnosis of coronary artery disease, congestive heart failure, cerebrovascular disease, diabetes, and peripheral vascular disease.
- All analyses were performed using Stata version 10.0MP (College Station, TX).

Results

Baseline Characteristics and Matching

- During the month prior to index, patients with a midodrine order had shorter dialysis vintage, lower pre-dialysis SBP and nadir SBP, and a higher percent of treatments affected by IDH compared to those without an order. These characteristics were well-balanced after matching (Table 1, Figure 1).
- At baseline, midodrine patients in the analytic cohort were, on average, older, more likely to be white and less likely to be black, were more likely to have congestive heart failure, and had lower serum albumin than controls (Table 2).

Table 1: Characteristics of Unmatched and Analytic Cohorts

		Unmatched		Analytic [c,d]	
		Control [a] N=887 735	Midodrine [b] N=3201	Control [b] N=2037	Midodrine [b] N=1046
Vintage, mo	mean ± SD	30.7 ± 21.8	27.6 ± 22.4*	27.1 ± 21.4	27.2 ± 21.7
Pre-dialysis SBP, mmHg	mean ± SD	149 ± 20	128 ± 20*	128 ± 21	127 ± 20
Nadir SBP, mm	mean ± SD	112 ± 18	91 ± 14*	91 ± 13	90 ± 13*
IDH, % treatments		18.1 ± 22.8	50.0 ± 31.3*	49.8 ± 30.6	50.8 ± 30.6*

[a] N represents patient-months, not unique patients; [b] N represents unique patients; [c] Values presented are as of index date

[d] Includes only midodrine patients (and matched controls) whose index dates were within 10 days of the start of the index month

*Significantly different than control, $p < 0.05$

Abbreviations: IDH, intradialytic hypotension; SBP, systolic blood pressure; SD, standard deviation

Figure 1: Balance in Unmatched and Analytic Cohorts

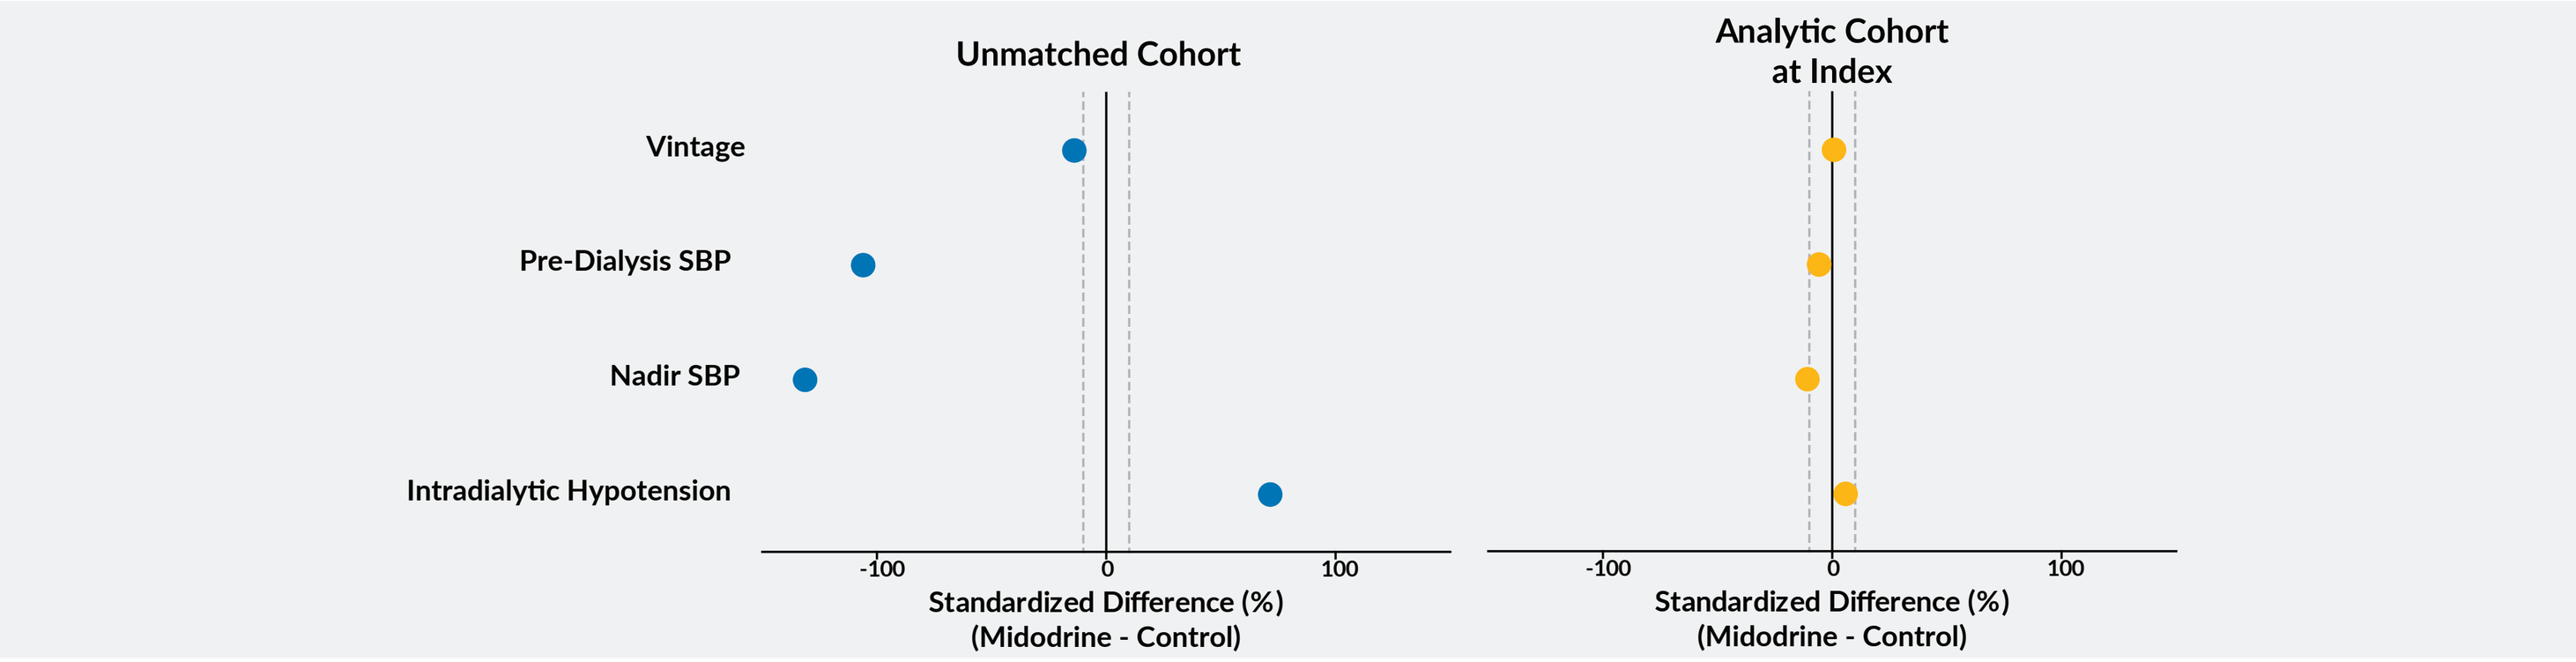
Unmatched Cohort
Analytic Cohort at Index
Vintage
Pre-Dialysis SBP
Nadir SBP
Intradialytic Hypotension
-100
0
100
Standardized Difference (%) (Midodrine - Control)

Data in unmatched cohort represents individual patient-months. Data in analytic cohort represents values as of index date date among matched clusters in which index date was ≤ 10 days from the end of the month in which patients were matched.

Abbreviations: SBP, systolic blood pressure.

Hemodynamic and Clinical Outcomes

- Over follow-up time, midodrine use was associated with a tendency toward lower pre-dialysis systolic blood pressure and lower nadir blood pressure than non-use (Figure 2).
 - Midodrine use was also associated with a greater fall in blood pressure during dialysis
 - Ultrafiltration volumes were comparable over follow-up time between the two groups.
- The percent of treatments affected by IDH tended to be higher in the midodrine group throughout follow-up.
- After adjusting for imbalanced patient characteristics, midodrine use was associated with higher rates of death, hospitalization, and cardiovascular hospitalization than non-use (Figure 3).

Table 2: Baseline Characteristics of the Analytic Cohort

		Control N=2037	Midodrine N=1046	Standardized Difference (%)	P-value
Age, years	mean ± SD	66.9 ± 14.2	69.0 ± 12.3	15.5	< 0.001
Gender, female, n (%)		922 (45.3)	478 (45.7)	0.9	0.81
Race, n (%)					< 0.001
White		961 (47.2)	588 (56.2)	18.1	
Black		920 (45.2)	374 (35.8)	-19.2	
Other/unknown/missing		156 (7.7)	84 (8.0)	1.4	
Vascular access, n (%)					0.03
Arteriovenous fistula		1329 (65.2)	633 (60.5)	-9.8	
Arteriovenous graft		330 (16.2)	189 (18.1)	5.0	
Central venous catheter		378 (18.6)	224 (21.4)	7.1	
Dialysis vintage, months					0.82
	mean ± SD	27.1 ± 21.4	27.3 ± 21.7		
	median [p25, p75]	20 [9, 41]	21 [9, 41]	0.8	
Target weight, kg	mean ± SD	82.6 ± 24.0	83.9 ± 23.8	5.8	
Etiology of ESRD, n (%)					0.02
Diabetes		881 (43.3)	505 (48.3)	10.1	
Hypertension		584 (28.7)	288 (27.5)	-2.5	
Other		572 (28.1)	253 (24.2)	-8.9	
Diabetes, n (%)		1445 (70.9)	771 (73.7)	6.2	0.11
Congestive heart failure, n (%)		274 (13.5)	185 (17.7)	11.7	0.002
Coronary artery disease, n (%)		219 (10.8)	129 (12.3)	4.9	0.19
Cerebrovascular disease, n (%)		19 (0.9)	13 (1.2)	3.0	0.42
Peripheral vascular disease, n (%)		82 (4.0)	45 (4.3)	1.4	0.71
Albumin, g/dL	mean ± SD	3.7 ± 0.4	3.6 ± 0.5	-22.8	< 0.001
Creatinine, mg/L	mean ± SD	7.8 ± 2.9	7.5 ± 2.8	-9.1	0.02
Kt/V,	mean ± SD	1.6 ± 0.3	1.5 ± 0.3	-10.1	0.009
Pre-dialysis SPB, mmHg	mean ± SD	128 ± 21	127 ± 20	-5.7	0.14
Nadir SPB, mmHg	mean ± SD	91 ± 13	90 ± 13	-10.8	0.005
Interdialytic hypotension, % of tx		49.8 ± 30.6	52.8 ± 3.05	9.7	0.01
Hemoglobin, g/dL	mean ± SD	10.8 ± 1.2	10.8 ± 1.3	-5.1	0.17
UF volume, L	mean ± SD	2.1 ± 1.0	2.0 ± 1.0	-3.0	0.42
Antihypertensive medications, n					< 0.001
	mean ± SD	1.8 ± 1.5	1.4 ± 1.3	-28.7	

Abbreviations: ESRD, end-stage renal disease; SD, standard deviation; SPB, systolic blood pressure; Tx, treatment; UF, ultrafiltration

Figure 2: Hemodynamic Outcomes

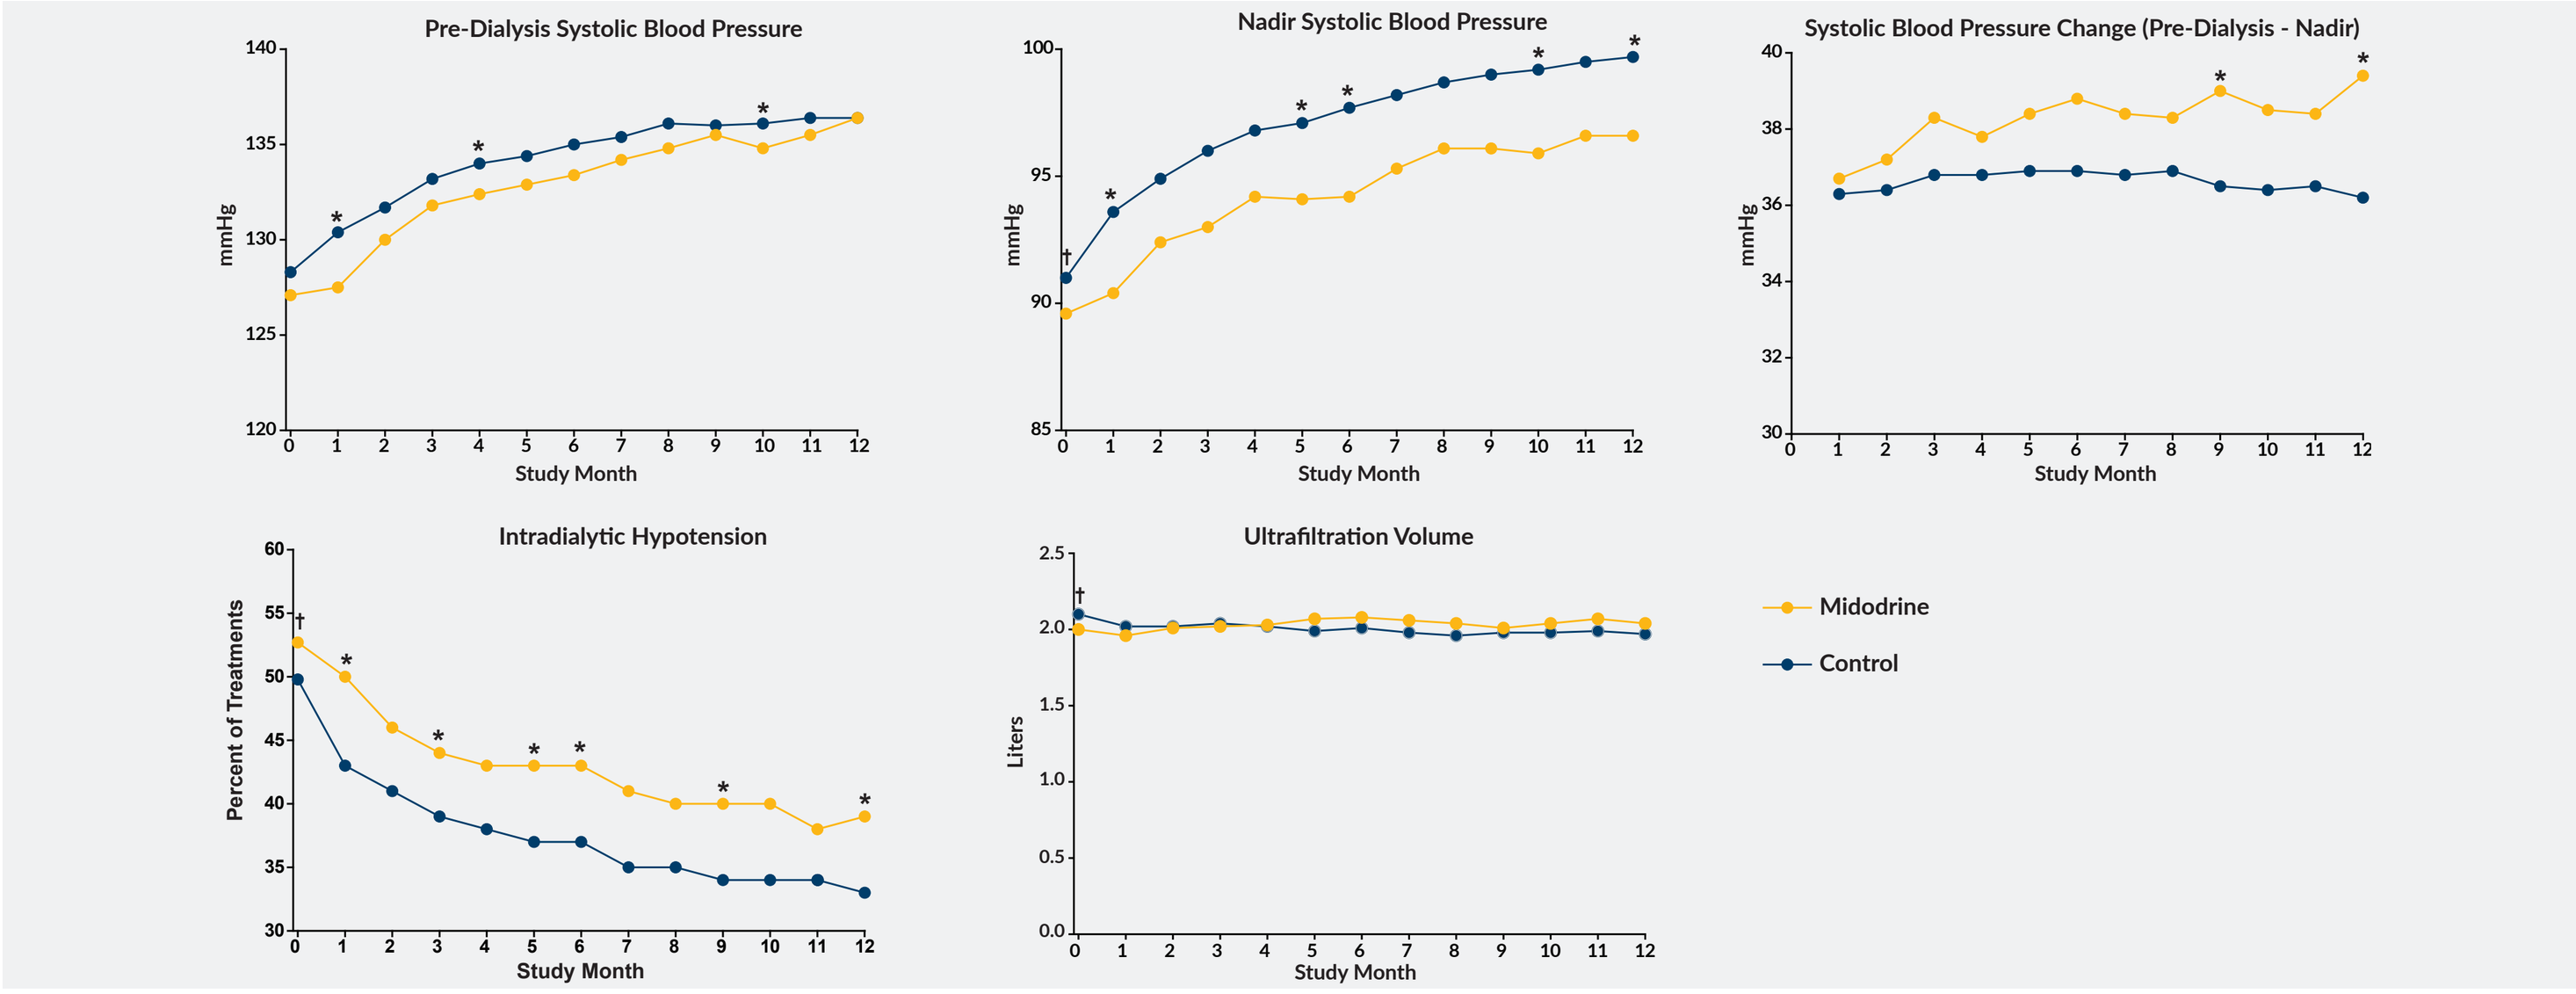
Pre-Dialysis Systolic Blood Pressure
Nadir Systolic Blood Pressure
Systolic Blood Pressure Change (Pre-Dialysis - Nadir)
Intradialytic Hypotension
Ultrafiltration Volume
mmHg
Percent of Treatments
Liters
Study Month
Midodrine
Control

Crude mean monthly values for the indicated parameter among the midodrine (gold) and control (blue) groups are indicated. Month 0 corresponds to the index month; months 1-12 represent follow-up time. † statistically significant difference between midodrine and control at index based on t-test, $p < 0.05$. * statistically significant difference between midodrine and control based on adjusted mean difference, $p < 0.05$.

Figure 3: Clinical Outcomes

	Control	Midodrine
Time at risk, pt-years	1755	811
Death		
Events, n	341	275
Crude rate, per pt-year	0.19	0.34
ARD (95% CI)	0 (ref)	0.04 (0.02, 0.07)
All-Cause Hospitalization		
Events, n	3072	1897
Crude rate, per pt-year	1.75	2.34
ARD (95% CI)	0 (ref)	0.42 (0.26, 0.58)
Cardiovascular Hospitalization		
Events, n	468	306
Crude rate, per pt-year	0.27	0.38
ARD (95% CI)	0 (ref)	0.08 (0.03, 0.13)

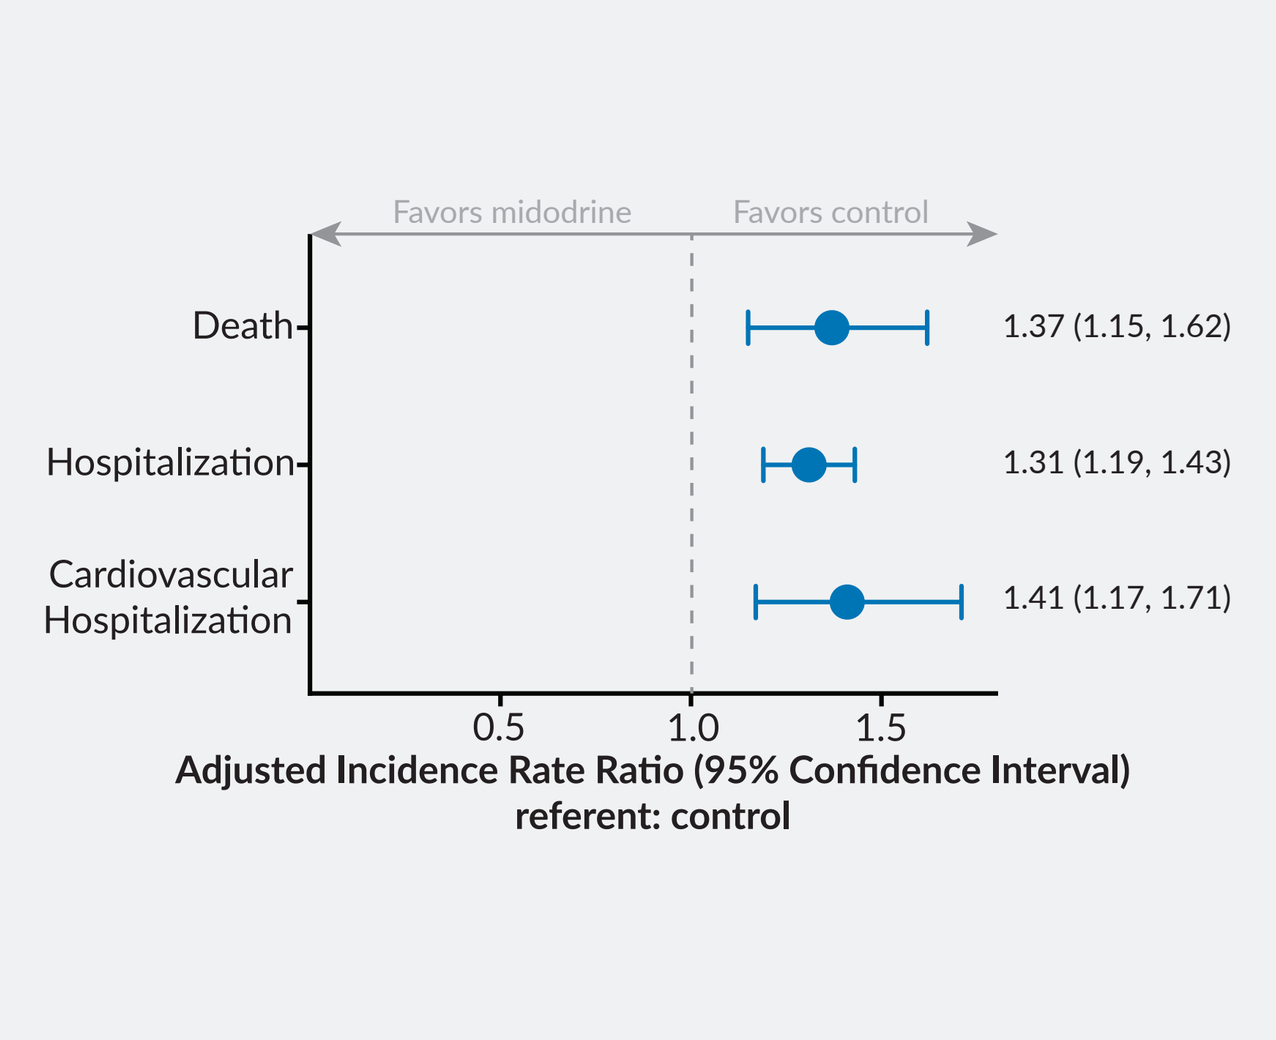
Favors midodrine
Favors control
Death
Hospitalization
Cardiovascular Hospitalization
1.37 (1.15, 1.62)
1.31 (1.19, 1.43)
1.41 (1.17, 1.71)
0.5
1.0
1.5
Adjusted Incidence Rate Ratio (95% Confidence Interval)
referent: control

Abbreviations: ARD, adjusted rate difference; CI, confidence interval; pt-year, patient-year

Conclusions

Although residual confounding almost certainly influenced the results, the data presented here are not consistent with a potent protective effect of midodrine with respect to any outcome examined.

References

1. Flythe JE, Xue H, Lynch KE, et al. Association of mortality risk with various definitions of intradialytic hypotension. J Am Soc Nephrol. Mar 2015;26(3):724-734
2. Prakash S, Garg AX, Heidenheim AP, et al. Midodrine appears to be safe and effective for dialysis-induced hypotension: a systematic review. Nephrol Dial Transplant. Oct 2004;19(10):2553-2558

Acknowledgments

This publication is the result of a research project conducted by the DaVita Institute for Patient Safety, Inc. a federally listed Patient Safety Organization, and is published with its permission. The data discussed herein has been certified as non-identifiable pursuant to 42 C.F.R. 3.212(a)(1). DaVita Clinical Research is a contractor to the DaVita Institute for Patient Safety, Inc.

Correspondence: steven.brunelli@davita.com

Poster available at www.davitaclinicalresearch.com

American Society of Nephrology Kidney Week, October 25 - 28, 2018; San Diego, CA

© 2018 DaVita Inc. All rights reserved. Proprietary. May not be copied, reprinted, or distributed without the permission of DaVita Inc.