Identification of Factors that Are Associated with Risk of Modality Failure Among Patients Treated with Peritoneal Dialysis

Steven M. Brunelli, MD, MSCE [1,2]; Dena E. Cohen, PhD [1,2]; Kathryn S. Gray, MS [1,2]; Michelle Cassin, RN [3]; Bram Van Hout, MSc [3]; Julie Rodriguez, MS [3]; David Van Wyck, MD [2]; Martin Schreiber, MD [3]

[1]DaVita Clinical Research, Minneapolis, MN, USA; [2]DaVita Institute for Patient Safety, Denver, CO, USA; [3]DaVita Kidney Care, Denver, CO, USA

Introduction

- Among patients with end-stage renal disease (ESRD), treatment with peritoneal dialysis (PD) is associated with better quality of life and clinical outcomes compared to in-center hemodialysis (ICHD).
- However, some patients who initiate PD may later fail the modality and switch to ICHD.
 - Clinical and non-clinical factors may contribute to modality failure.
- Targeted interventions that address such factors may enable retention of patients on PD.

Objective

To identify factors that are associated with greater risk of PD modality failure

Methods

Study Data, Timeframe and Patients

- All study data were derived from deidentified patient electronic health records.
- Included patients were adults (≥ 18 years old) who were either prevalent on PD or initiated PD between 01 January 2014 and 31 December 2016. Veterans Administration beneficiaries were excluded.
- The index event was defined as the first of at least 3 consecutive non-training PD treatments during the study period, indicating commencement of PD therapy.
- Follow-up time began on the first day of the first full calendar month following index and continued until the first of study end (31 July 2017), censoring (transplant, death, loss to follow-up), modality change (transition from PD to home hemodialysis), or modality failure (transition from PD to ICHD).

Statistical Analysis

- Analysis was conducted at the level of the patient-month.
 - Variables were considered as of index, as of the month prior to the month under consideration, or the difference between two months prior minus one month prior, as appropriate.
- This was a time-to-event analysis. Cox regression models were each tested on a bivariable basis against outcome, followed by joint modeling where multiple potential risk factors were entered into the model simultaneously.
- All models were adjusted for age, sex, race, history of congestive heart failure or amputation, number of months of historic data availability prior to index date, and Charlson comorbidity index, and used robust variance estimators to account for multiple observations made in individual patients over time.
- Interactions were tested using likelihood ratio testing.

Results

Patient Case Mix

- Patient case mix as of index date, and the association between these attributes and modality failure, are summarized in Table 1.
- No associations were observed between etiology of ESRD, diabetes, or coronary artery disease and modality failure (data not shown); these attributes were not considered further.
- Significant associations were observed between age, sex, race, number of months of historic data availability prior to index date, Charlson comorbidity index, congestive heart failure, and amputation.
 - All subsequent analyses were adjusted for these factors.

Table 1: Patient Case Mix and Associations with Modality Failure

		Patients on PD N = 303,126 patient-months	Adjusted Hazard Ratio (95% CI)
Age, years	mean ± SD	58.0 ± 15.3	0.91 (0.88, 0.94) [a]
Sex			
Male, n (%)		172,293 (57.8)	(ref)
Female, n (%)		130,833 (43.2)	0.93 (0.88, 0.99)
Race, n (%)			
White		157,766 (52.1)	(ref)
Black		116,746 (38.5)	0.96 (0.90, 1.02)
Other/unknown/missing		28,614 (9.4)	0.73 (0.65, 0.82)
Historic data availability prior to index, months			Significant Association [b]
	mean ± SD	13.5 ± 8.9	
	median [p25, p75]	12 [6, 19]	
Etiology of ESRD, n (%)			N.S.
Diabetes		110,154 (36.3)	
Hypertension		77,987 (25.7)	
Other/unknown/missing		114,985 (37.9)	
Charlson Comorbidity Index, n (%)			
2		43,657 (14.4)	0.72 (0.62, 0.82)
3		36,310 (12.0)	0.82 (0.72, 0.92)
4		59,631 (19.7)	0.99 (0.90, 1.10)
5		62,922 (20.8)	(ref)
6		57,694 (19.0)	1.02 (0.92, 1.13)
7		30,675 (10.1)	1.11 (0.98, 1.26)
8+		12,273 (4.0)	1.18 (0.99, 1.42)
Diabetes, n (%)		182,829 (60.3)	N.S.
Congestive heart failure, n (%)		6359 (2.1)	0.79 (0.63, 0.99)
Coronary artery disease, n (%)		29,934 (9.9)	N.S.
Amputation, n (%)		7903 (2.6)	1.23 (1.05, 1.46)

[a] Per 10 years greater

[b] Months of historic data availability prior to index had an inverse u-shaped association with modality failure with the peak hazard ratio occurring at approximately 20 months.

Abbreviations: CI, confidence interval; ESRD, end-stage renal disease; N.S., not significant; ref, referent; SD, standard deviation

Identification of Factors Associated with PD Modality Failure

- Candidate factors were grouped into broad thematic categories; factors within each thematic category were assessed to determine which factors within each category were associated with modality failure.
- Among fluid-related factors examined, body weight, body weight change, serum albumin, change in serum albumin, and Kt/V displayed significant associations with PD modality failure (Figure 1).
 - Drain volume, loop diuretic use, icodextrin use, serum sodium, change in serum sodium, fluid-related hospital admission, and membrane transport characteristics did not display significant associations with modality failure.
 - No interactions were detected between albumin and change in albumin or between Kt/V and treatment type. Membrane transport characteristics did not interact with drain volume, icodextrin, use of loop diuretics, weight, serum sodium, or serum albumin. Fluid-related hospitalization did not interact with either use of loop diuretics or icodextrin.

Figure 1: Association of Fluid-Related Factors with PD Modality Failure

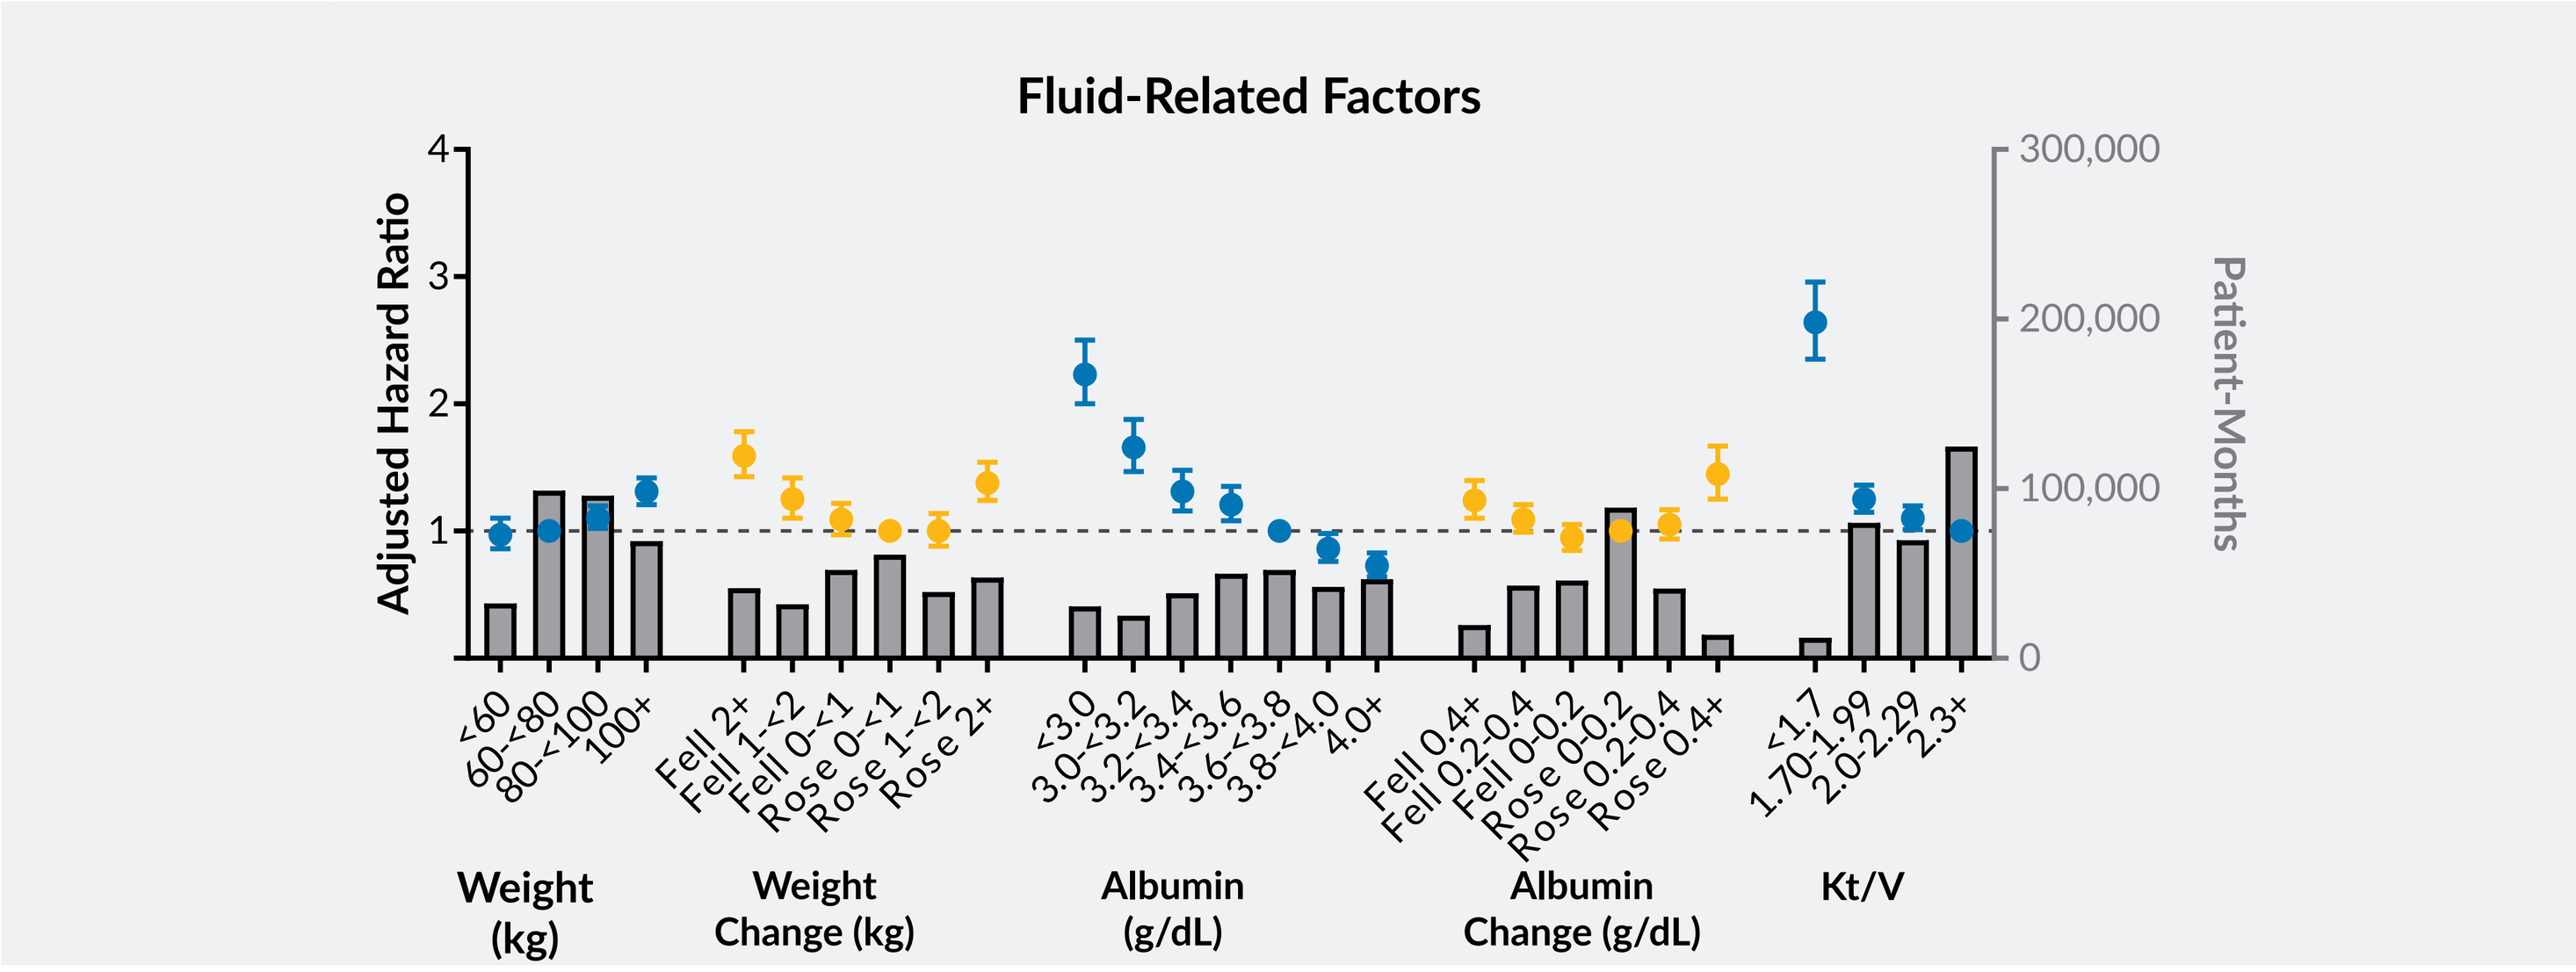
Fluid-Related Factors
Adjusted Hazard Ratio
Patient-Months
<60
60-<80
80-<100
100+
Fell 2+
Fell 1-<2
Fell 0-<1
Rose 0-<1
Rose 1-<2
Rose 2+
<3.0
3.0-<3.2
3.2-<3.4
3.4-<3.6
3.6-<3.8
3.8-<4.0
4.0+
Fell 0.4+
Fell 0.2-0.4
Fell 0-0.2
Rose 0-0.2
Rose 0.2-0.4
Rose 0.4+
<1.7
1.70-1.99
2.0-2.29
2.3+
Weight (kg)
Weight Change (kg)
Albumin (g/dL)
Albumin Change (g/dL)
Kt/V

Adjusted for age, sex, race, vintage, Charlson comorbidity index, congestive heart failure, amputation, and, for each factor, all of the following candidate factors except itself: drain volume, diuretic use, icodextrin use, weight, weight change, serum sodium, change in serum sodium, albumin, change in albumin, Kt/V, fluid-related hospital admission, membrane transport characteristics. Change in weight or albumin was considered as the difference between the measured value two months prior to the patient-month being considered minus the value in the month prior to the month being considered.

Figure 2: Association of Burden-Related Factors with PD Modality Failure

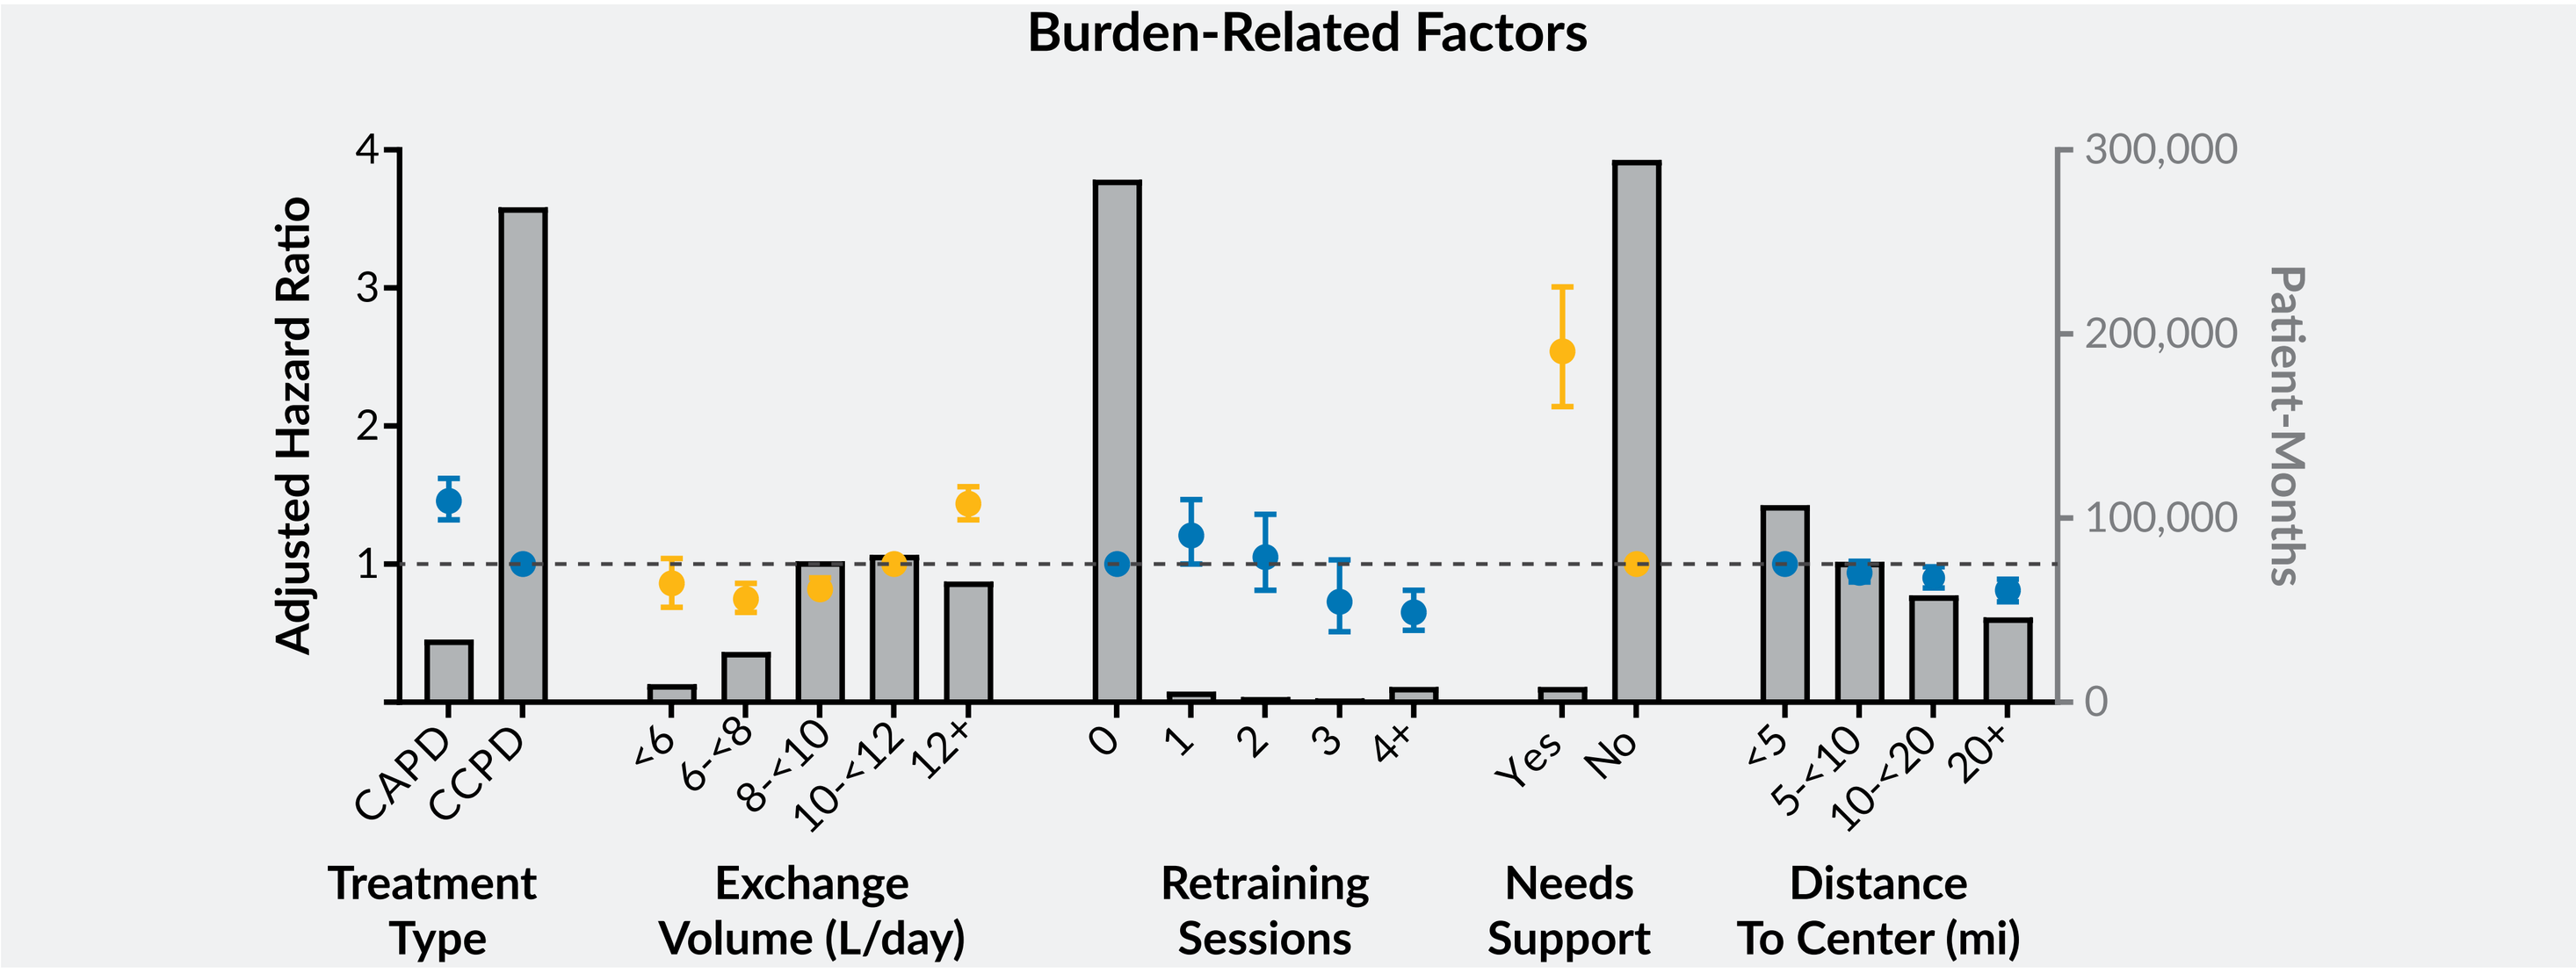
Burden-Related Factors
Adjusted Hazard Ratio
Patient-Months
CAPD
CCPD
<6
6-<8
8-<10
10-<12
12+
0
1
2
3
4+
Yes
No
<5
5-<10
10-<20
20+
Treatment Type
Exchange Volume (L/day)
Retraining Sessions
Needs Support
Distance To Center (mi)

Adjusted for age, sex, race, vintage, Charlson comorbidity index, congestive heart failure, amputation, and, for each factor, all of the following candidate factors except itself: treatment type (continuous ambulatory vs. continuous cycling), exchange volume, retraining sessions, needs support (an indication that the patient needs support to perform treatment), and distance to center (where patient receives treatment).

- All of the burden-related factors examined displayed associations with PD modality failure. These were: treatment type (continuous ambulatory vs continuous cycling PD), daily exchange volume, receipt of retraining sessions, an indication of the need for support during treatment, and the distance to the treatment center (Figure 2).
 - No interactions were observed between treatment type and exchange volume, treatment type and re-training, coronary artery disease and need for support, or retraining and need for support. Although an interaction was detected between treatment type and membrane transport status, risk of modality failure did not differ significantly across treatment type X membrane transport categories (not shown).
- Among peritonitis-related factors examined, hospital admission, performance of a peritoneal culture, detection of gram-negative rods in the peritoneal fluid, and prescription of antibiotics were significantly associated with PD modality failure (Figure 3A).
 - Hospital admission for peritonitis, white blood cell count, and change in white blood cell count were not associated with PD modality failure.
- Both of the facility-related factors examined, namely the number of ICHD chairs available at the facility, and the number of patients treated, were associated with PD modality failure (Figure 3B).

Figure 3: Association of Peritonitis- and Facility-Related Factors with PD Modality Failure

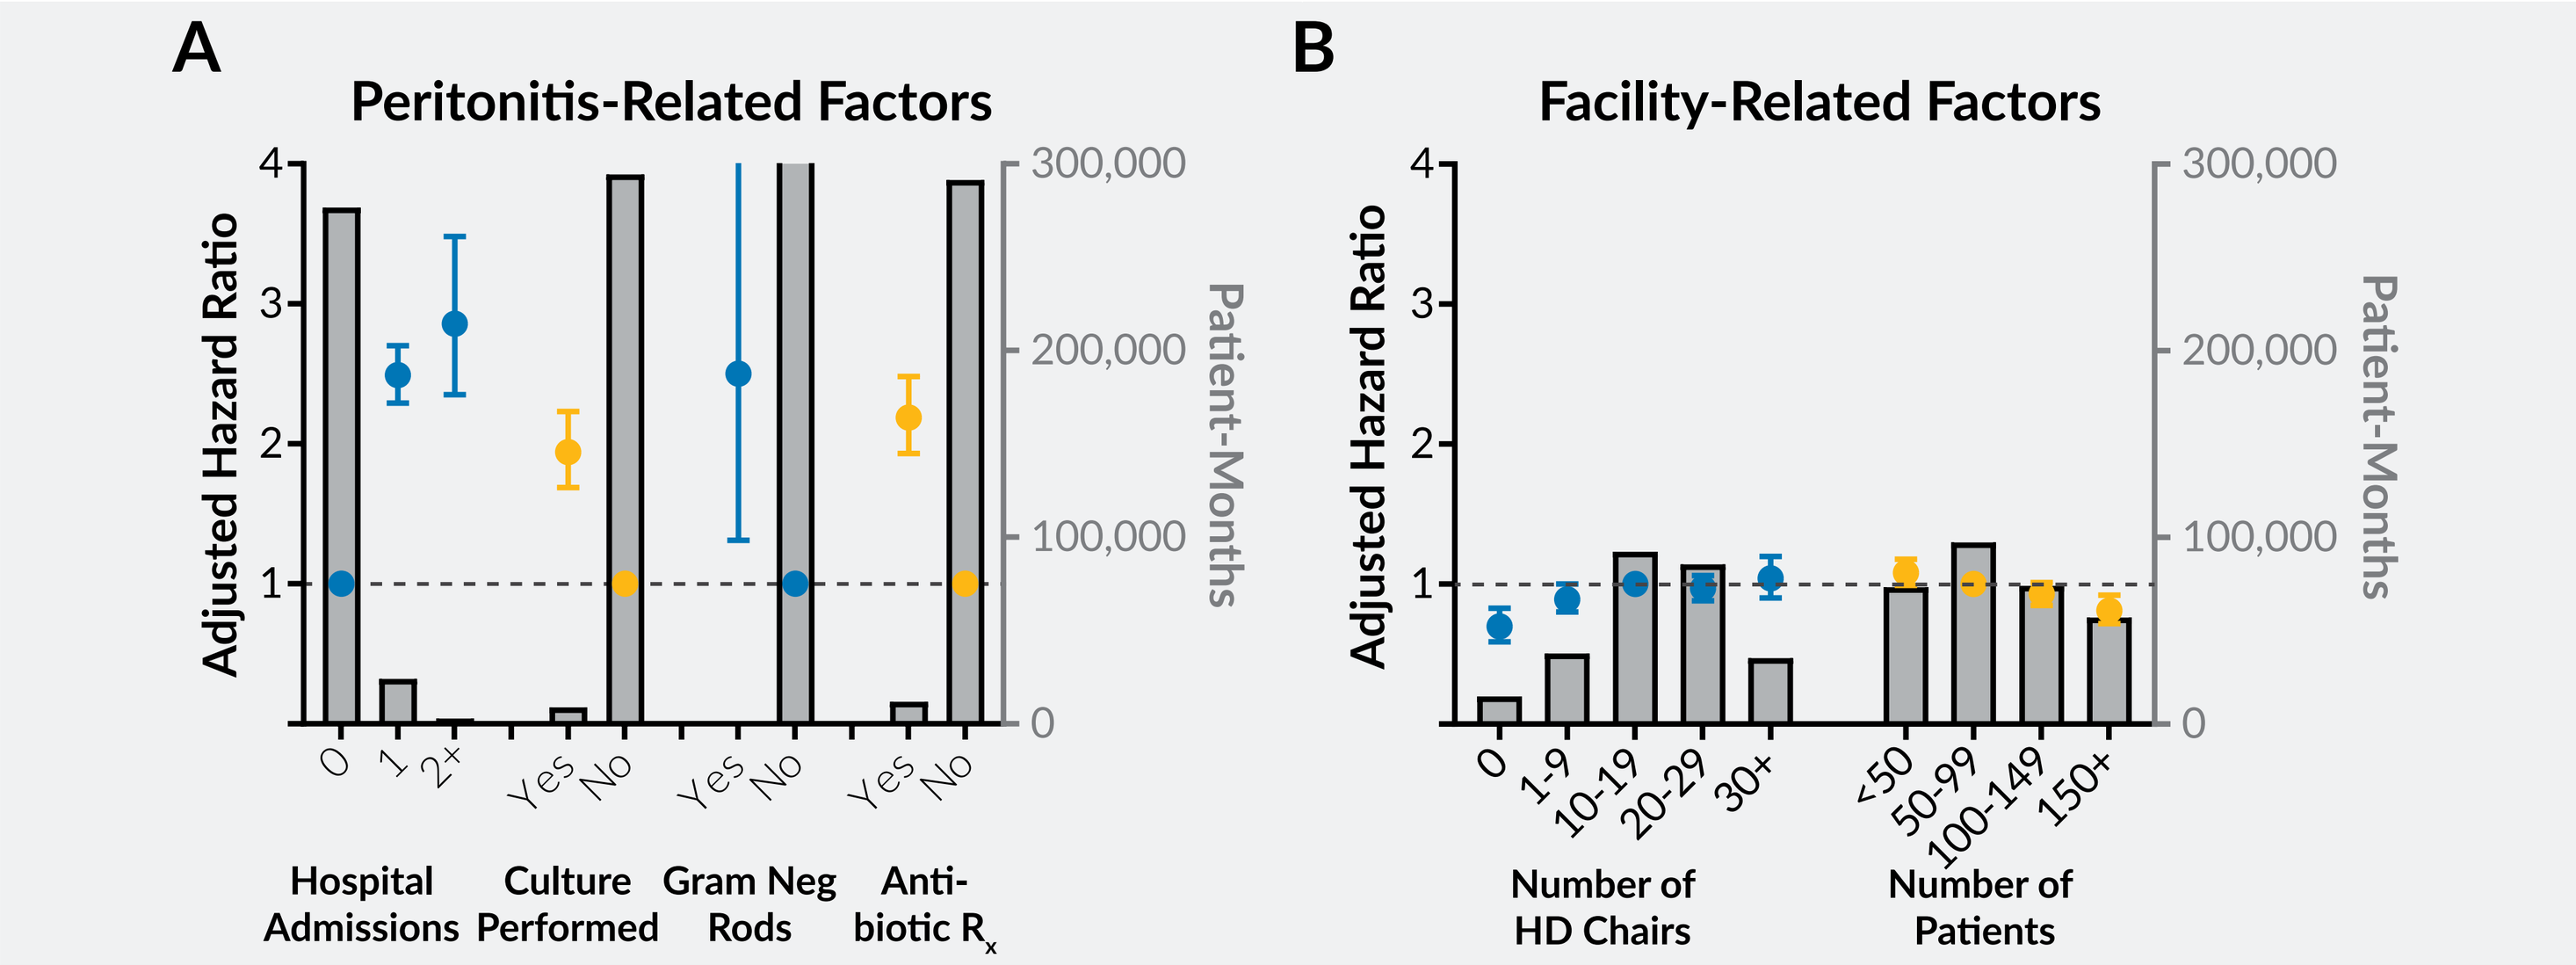
A
Peritonitis-Related Factors
Adjusted Hazard Ratio
Patient-Months
0
1
2+
Yes
No
Hospital Admissions
Culture Performed
Gram Neg Rods
Anti-biotic R_x
B
Facility-Related Factors
0
1-9
10-19
20-29
30+
<50
50-99
100-149
150+
Number of HD Chairs
Number of Patients

Adjusted for age, sex, race, vintage, Charlson comorbidity index, congestive heart failure, amputation, and, for each factor, all of the following candidate factors except itself: A) hospital admission, peritonitis-related hospital admission, culture of gram-negative rods, culture performed, antibiotic prescription, white blood cell count, change in white blood cell count. B) number of HD chairs, number of patients.
Abbreviations: HD, hemodialysis, Neg, negative; R_x, prescription.

Conclusions

- The factors with the most prominent associations with PD modality failure were the need for support during treatment, low Kt/V, low albumin, hospital admission (all-cause and fluid-related), and the need for antibiotics and/or peritoneal fluid culture.
 - Each of these factors was associated with at least a doubling of risk of modality failure.

Acknowledgments

This publication is the result of a research project conducted by the DaVita Institute for Patient Safety, Inc. a federally listed Patient Safety Organization, and is published with its permission. The data discussed herein has been certified as non-identifiable pursuant to 42 C.F.R. 3.212(a)(1). DaVita Clinical Research is a contractor to the DaVita Institute for Patient Safety, Inc.

Correspondence: steven.brunelli@davita.com

Poster available at www.davitaclinicalresearch.com

American Society of Nephrology Kidney Week, October 23 - 28, 2018; San Diego, CA

© 2018 DaVita Inc. All rights reserved. Proprietary. May not be copied, reprinted, or distributed without the permission of DaVita Inc.